CT235 AACR 2021 | 10-15 APRIL 2021 | VIRTUAL MEETING

Trial in Progress: A Phase 2, multicenter study of autologous tumor infiltrating lymphocytes (TIL, LN-144/LN-145/LN-145-S1) in patients with solid tumors (IOV-COM-202)

Scott Gettinger, MD[1]; Harriet Kluger, MD[1]; Adam Schoenfeld, MD[2]; Allison Betof Warner, MD, PhD[2]; Kai He, MD, PhD[3]; Ammar Sukari, MD[4]; Sajeve Samuel Thomas, MD[5]; Bernard Doger de Speville Uribe, MD, PhD[6]; Sylvia Lee, MD[7]; Simon Haefliger, MD, PhD[8]; Zelanna Goldberg, MD, MAS[9]; Alex Cacovean, MD[9]; Rana Fiaz, MD[9]; Guang Cheng, PhD[9]; Madan Jagasia, MD, MS, MMHC[9]; Friedrich Graf Finckenstein, MD[9]; Maria Fardis, PhD, MBA[9]; Antonio Jimeno, MD, PhD[10]

[1]Yale Cancer Center, New Haven, CT, USA; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]James Cancer Center, The Ohio State University, Columbus, OH, USA; [4]Barbara Ann Karmanos Cancer Center, Wayne State University, Detroit, MI, USA; [5]University of Florida Health Cancer Center at Orlando Health, Orlando, FL, USA; [6]Hospital Universitario Fundacion Jimenez Diaz, Madrid, ES; [7]Seattle Cancer Care Alliance, University of Washington, Seattle, WA, USA; [8]Inselspital, Universitätsspital Bern, Bern, CH; [9]Iovance Biotherapeutics, Inc., San Carlos, CA, USA; [10]University of Colorado Comprehensive Cancer Center, Aurora, CO, USA.

IOVANCE BIOTHERAPEUTICS

ADVANCING IMMUNO-ONCOLOGY

999 Skyway Road, STE 150, San Carlos, CA 94070

For more information, please contact
Madan Jagasia, MD, MS, MMHC
madan.jagasia@iovance.com

Background

- **Adoptive cell transfer (ACT) using autologous tumor-infiltrating lymphocytes (TIL)** has been shown to be efficient for the treatment of advanced metastatic melanoma, and other solid tumors with high tumor mutational burden.[1,2]
- **Iovance TIL** cell therapy (LN-144 [lifileucel] and LN-145), has demonstrated efficacy and safety in clinical trials for several high unmet medical need patient populations, specifically unresectable and metastatic melanoma, in relapsed, refractory or persistent cervical cancer, and in head and neck squamous cell carcinoma (HNSCC).[3-5]
- Further, TIL cell therapy has shown evidence of efficacy in metastatic non-small cell lung cancer (mNSCLC) in a Phase 1 study in combination with nivolumab.[6]

IOV-COM-202

- **IOV-COM-202 (NCT03645928)** is a prospective, open-label, multi-cohort, non-randomized, multicenter Phase 2 study evaluating Iovance TIL cell therapy (LN-144, LN-145, LN-145-S1) in combination with immune checkpoint inhibitors (ICIs) and as a single therapy.

Iovance TIL Manufacturing

- **The one-time Iovance TIL cell therapy** requires procurement of a small 1.5 cm sample of tumor tissue, which is shipped to a central manufacturing facility where outside of the suppressive tumor microenvironment the TIL are reinvigorated and expanded to approximately 10^9–10^{11} cells (LN-144, LN-145, LN-145-S1). Manufacturing is a 16 day (Gen 3, Cohort 1C) or 22 day (Gen 2 or LN-145-S1) process.

Figure 1. Patient Journey and Central GMP Manufacturing

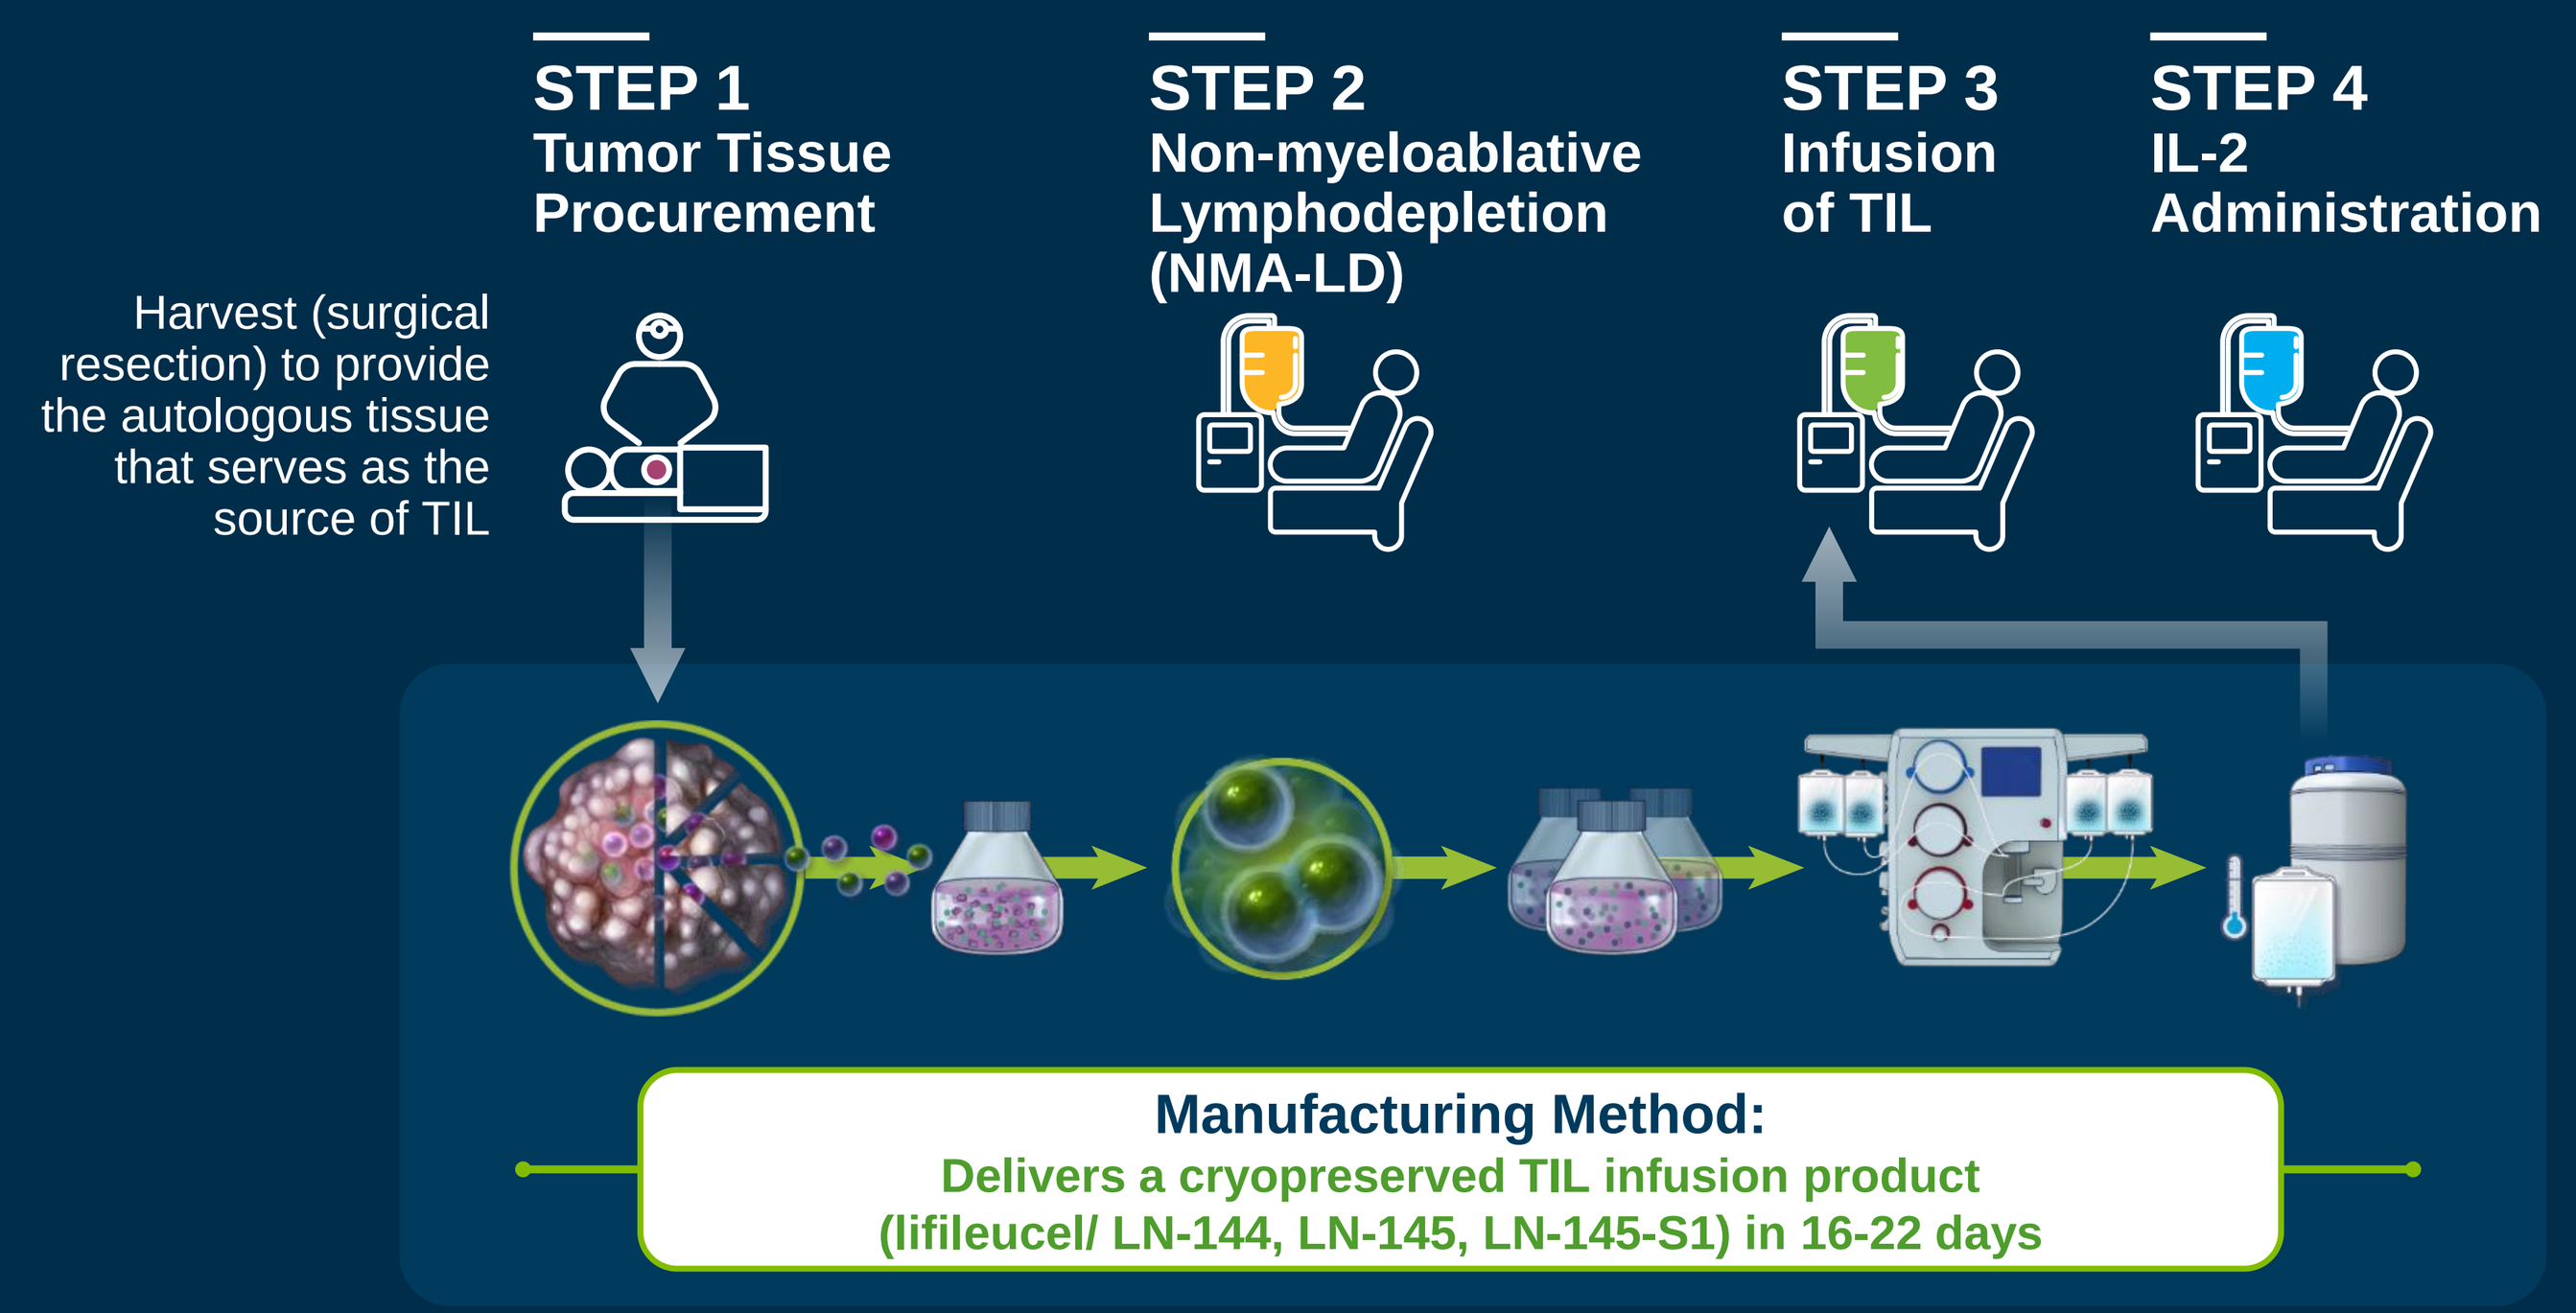

STEP 1
Tumor Tissue Procurement
STEP 2
Non-myeloablative Lymphodepletion (NMA-LD)
STEP 3
Infusion of TIL
STEP 4
IL-2 Administration
Harvest (surgical resection) to provide the autologous tissue that serves as the source of TIL
Manufacturing Method:
Delivers a cryopreserved TIL infusion product
(lifileucel/ LN-144, LN-145, LN-145-S1) in 16-22 days

Study Overview & Endpoints

- Approximately 50 clinical sites in the US, Canada, and Europe.
- Primary endpoint:
 - Efficacy and safety: Objective response rate (ORR) per RECIST 1.1 as assessed by investigator.
- Secondary endpoints:
 - Additional efficacy parameters.
- Exploratory endpoints:
 - Predictive and pharmacodynamic biomarkers of clinical response to TIL products.

© 2021, Iovance Biotherapeutics, Inc. All Rights Reserved.

Study Cohorts

Figure 2. Study Design

A Phase 2, multicenter study of autologous Tumor Infiltrating Lymphocytes in patients with solid tumors

→ **Cohort 1A: Melanoma**
PD-1/ PD-L1 Naïve
LN-144 + Pembrolizumab
N=12

→ **Cohort 1B: Melanoma**
≥1 prior systemic therapies
LN-145-S1
N up to 27

→ **Cohort 1C: Melanoma**
≥1 prior systemic therapies
LN-144 Gen 3
N up to 27

→ **Cohort 2A: Head and Neck**
PD-1/ PD-L1 Naïve
LN-145 + Pembrolizumab
N=19

→ **Cohort 3A: NSCLC**
PD-1/ PD-L1 Naïve
LN-145 + Pembrolizumab
N=12

→ **Cohort 3B: NSCLC**
≥1 prior systemic therapies
LN-145
N=12

→ **Cohort 3C: NSCLC**
1 prior systemic therapy
TIL + Ipilimumab / Nivolumab
N up to 26

- **LN 144/LN-145:** 22-day manufacturing
- **LN144 Gen 3:** 16-day manufacturing
- **LN-145-S1:** PD-1 selected TIL

IOV-COM-202 TIL Regimen

Figure 3. Patient Treatment Schema

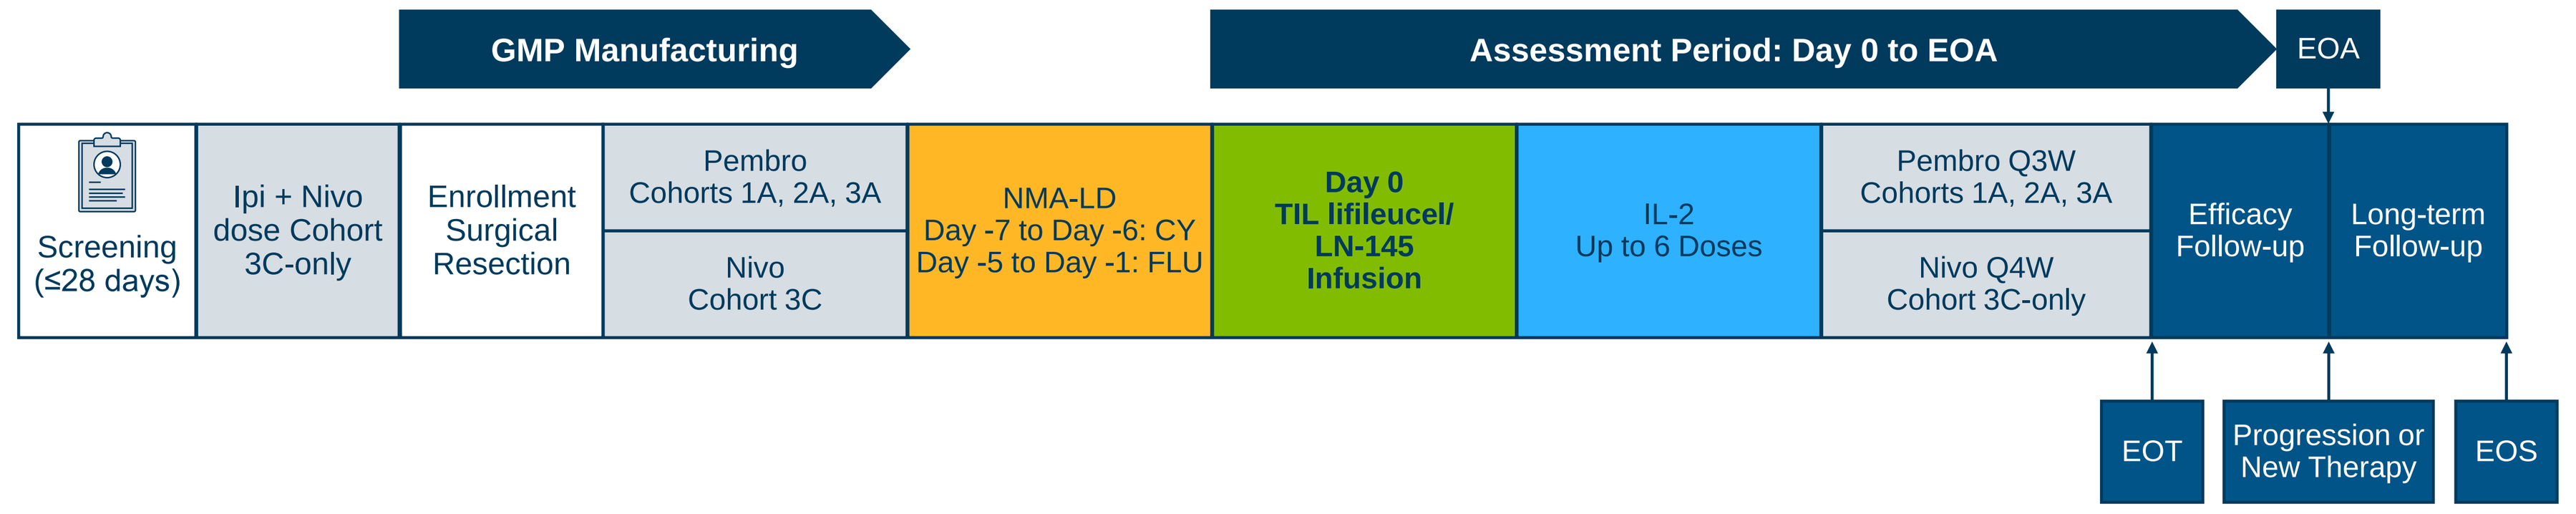

GMP Manufacturing
Assessment Period: Day 0 to EOA
EOA
Screening (≤28 days)
Ipi + Nivo dose Cohort 3C-only
Enrollment Surgical Resection
Pembro Cohorts 1A, 2A, 3A
Nivo Cohort 3C
NMA-LD Day -7 to Day -6: CY Day -5 to Day -1: FLU
Day 0 TIL lifileucel/ LN-145 Infusion
IL-2 Up to 6 Doses
Pembro Q3W Cohorts 1A, 2A, 3A
Nivo Q4W Cohort 3C-only
Efficacy Follow-up
Long-term Follow-up
EOT
Progression or New Therapy
EOS

References:
[1]Goff SL, et al. *JCO.* 2016;34(20):2389-97.
[2]Stevanović S, et al. *Clin Can Res.* 2019;25(5):1486-1493.
[3]Sarnaik A, et al. *JCO.* 2020;38 (suppl; abstr 10006).
[4]Jazaeri A, et al. *JCO.* 2019;37 (suppl; abstract 2538).
[5]Jimeno A, et al. *JITC.* 2020;8 (suppl; abstract A378.
[6]Creelan, B et al. *Can Res.* 2020;80:16 (suppl; abstract CT056).

Abbreviations: ACT, adoptive cell therapy; CY, cyclophosphamide; ECOG, Eastern Cooperative Oncology Group; EOA, end of assessment; EOT, end of treatment; EOS, end of study; FLU, fludarabine; FEV_1, forced expiratory volume in 1 second; FVC, forced vital capacity; GMP, good manufacturing practice; HNSCC, head and neck squamous cell carcinoma; ICI, immune checkpoint inhibitors; IL-2, interleukin-2; Ipi, ipilimumab; LVEF, left ventricular ejection fraction; mNSCLC, metastatic non-small cell lung cancer; Nivo, nivolumab; NMA-LD, nonmyeloablative lymphodepletion; Pembro, pembrolizumab; PD-1, programmed cell death protein 1; PD-L1, programmed death-ligand 1; RECIST, Response Evaluation Criteria in Solid Tumors; TIL, tumor infiltrating lymphocytes; VFC, forced vital capacity.

Key Inclusion & Exclusion Criteria

Inclusion Criteria – All Patients

- At least 1 resectable lesion.
- Must have remaining measurable disease as defined by RECIST 1.1 following tumor resection.
- ≥18 years of age at the time of consent. Enrollment of patients >70 years of age may be allowed after consultation with the Medical Monitor.
- ECOG performance status of 0 or 1, and an estimated life expectancy of ≥6 months.

Prior Therapy Criteria

Cohort	Indication	Count of prior lines of systemic therapy	Prior CPI required	Prior targeted therapy required
1A	Melanoma (Stage IIIC or IV)	Treatment naïve – 3 L*		[4]
1B	Melanoma (Stage IIIC or IV)	≥1 L	✓[2]	✓[5]
1C	Melanoma (Stage IIIC or IV)	≥1 L	✓[2]	✓[5]
2A	HNSCC (advanced, recurrent, or metastatic)	Treatment naïve – 3 L*		
3A	NSCLC (Stage III or IV)	Treatment naïve – 3 L*[1]		✓[6]
3B	NSCLC (Stage III or IV)	1 L – 3 L	✓[3]	✓[6]
3C	NSCLC (Stage III or IV)	1 L (CPI monotherapy)	✓	

[1]Or ≤4 lines if ≥2 TKIs.
[2] Must include PD-1 blocking antibody.
[3] Except for those patients with known oncogene drivers that are sensitive to targeted therapies
[4] May have received BRAFi/MEKi if *BRAF* mutation positive.
[5] Must-have received BRAFi/MEKi if *BRAF* mutation positive.
[6] Must have received targeted therapy if known oncogene driver mutations.
* Must be ICI naïve.

Exclusion Criteria – All Patients

- Received an organ allograft or prior cell transfer therapy that included a nonmyeloablative or myeloablative chemotherapy regimen within the past 20 years.
- Symptomatic and/or untreated brain metastases.
- Receiving systemic steroid therapy ≥10 mg/day of prednisone or another steroid equivalent.
- Receiving steroids as replacement therapy for adrenocortical insufficiency at ≤10 mg/day of prednisone or another steroid equivalent may be eligible.
- Active medical illness(es), which in the opinion of the Investigator, would pose increased risks for study participation.
- Any form of primary immunodeficiency.
- History of hypersensitivity to any component of the study drugs.
- Left ventricular ejection fraction (LVEF) >45% or New York Heart Association Class II or higher.
- Patients with respiratory dysfunction or history of smoking require pulmonary function testing and are excluded if not meeting either of forced expiratory volume in 1 second (FEV_1)/forced vital capacity (FVC) >0.7 or FEV1 >50%.

Cohort-specific Exclusion Criteria

- Cohort 1A, 2A, 3A, and 3C patients may not have a medical history of autoimmune disorders requiring treatment or active management.

Disclosures
•This study and poster are sponsored by Iovance Biotherapeutics, Inc. San Carlos, CA, USA.
•ZG, AC, BY, GC, FGF, MJ, and MF are employees or consultants of Iovance Biotherapeutics, Inc. and have stock options.