1015 AACR Annual Meeting 2022 | April 8–13 | New Orleans, LA, USA

Preclinical Activity and Manufacturing Feasibility of Genetically Modified *PDCD-1* Knockout (KO) Tumor-Infiltrating Lymphocyte (TIL) Cell Therapy

Arvind Natarajan,[1] Anand Veerapathran,[1] Adrian Wells,[1] Courtney Herman,[1] Viktoria Gontcharova,[1] Kenneth Onimus,[1] Marcus Machin,[1] Seth Wardell,[1] Jamie L. Blauvelt,[2] Madan Jagasia,[1] Rafael Cubas[1]

[1]Iovance Biotherapeutics, Inc., San Carlos, CA, USA; [2]Moffit Cancer Center, Tampa, FL

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from AACR Annual Meeting 2022, and the author of this poster

Corresponding Author:
Arvind Natarajan; arvind.natarajan@iovance.com

Background

- Adoptive cell therapy using autologous tumor-infiltrating lymphocytes (TIL; lifileucel, LN-145) has demonstrated encouraging efficacy and safety in both the post-immune checkpoint inhibitor (ICI) and ICI-naïve settings in patients with advanced solid tumors
 - One-time lifileucel TIL cell therapy achieved durable responses in the post-ICI setting in patients with advanced/unresectable melanoma,[1,2] with an objective response rate (ORR) of 36% and duration of response (DOR) not reached after 33.1 months of follow-up[2]
 - In ICI-naïve patients with advanced melanoma, early-line combination of lifileucel plus pembrolizumab resulted in a 60% ORR, with a 30% complete response rate[3]
- Although effective, anti–programmed cell death protein (PD)-1 ICI therapy is limited by poor penetration into the tumor, internalization, and endocytic clearance[4-8]; by contrast, TIL are actively transported into the tumor, can move independently based on chemoattraction, and are not cleared or internalized[9]
- Administration of anti–PD-1 antibodies is associated with immune-related adverse events (AEs) due to non-specific upregulation of immune pathways through the systemic activation and proliferation of T cells[10]
- IOV-4001, a *PDCD-1* knockout (KO) TIL cell therapy, may enhance the efficacy of TIL cell therapy and abrogate the need for systemic anti–PD-1 therapy, while avoiding short- and long-term systemic AEs associated with anti–PD-1/PD-L1 therapy
 - Transcription activator-like effector endonucleases (TALEN®) are hybrid molecules composed of a DNA-binding domain and the FokI nuclease. Combination of 2 TALEN® arms directed at the *PDCD-1* gene encoding PD-1 mediates DNA double-strand breaks, leading to gene disruption and PD-1 inactivation[11-13]
 - A process has been established for the generation of TALEN®-mediated *PDCD-1* KO TIL and their expansion to therapeutically relevant numbers with robust effector function and phenotypic markers indicative of functional TIL (TALEN® gene-editing technology is licensed from Cellectis)[14]
- Here, we describe IOV-4001 (1) in vivo preclinical activity, (2) clinical-scale manufacturing process development, and (3) phenotypic and functional characterization

Methods

- **Manufacturing**:
 - A 22-day clinical-scale IOV-4001 manufacturing process was established, including pre-rapid expansion protocol (pre-REP), activation, electroporation, resting, and REP for the generation of *PDCD-1* KO TIL (developed with TALEN® gene-editing technology in collaboration with Cellectis, Paris, France)
 - Development runs were generated in non-Good Manufacturing Practice (GMP) scale at Iovance Process Development (Tampa, FL)
 - Manufacturing runs were generated in GMP manufacturing scale at the contract manufacturing organization (CMO)

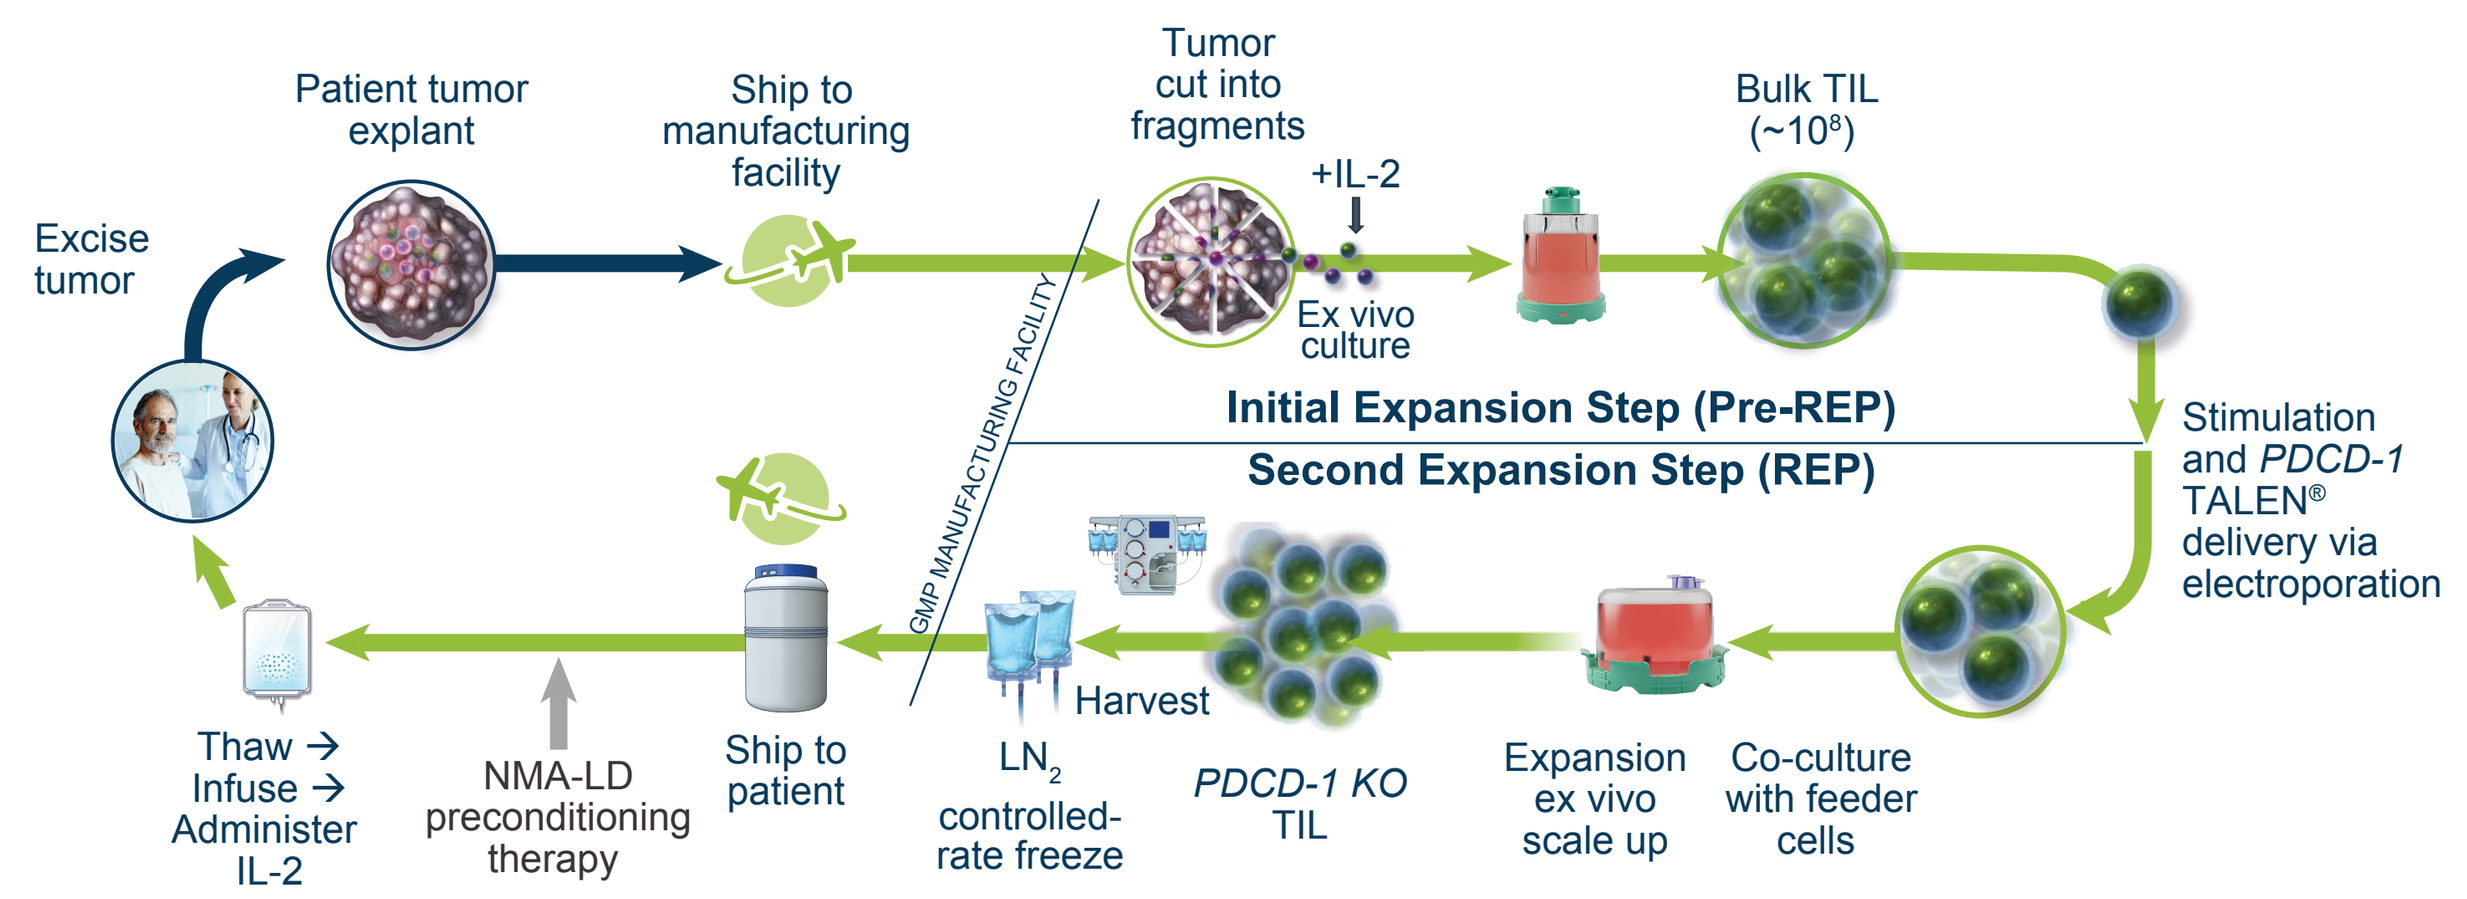
Excise tumor
Patient tumor explant
Ship to manufacturing facility
Tumor cut into fragments
+IL-2
Ex vivo culture
Bulk TIL (~10^8)
GMP MANUFACTURING FACILITY
Initial Expansion Step (Pre-REP)
Second Expansion Step (REP)
Stimulation and PDCD-1 TALEN® delivery via electroporation
Co-culture with feeder cells
Expansion ex vivo scale up
Harvest
PDCD-1 KO TIL
LN_2 controlled-rate freeze
Ship to patient
NMA-LD preconditioning therapy
Thaw → Infuse → Administer IL-2

- **In Vivo Antitumor Activity:**
 - Mice expressing human interleukin-2 (hIL-2) under the control of a cytomegalovirus promoter (hIL-2 NOG)[15] were engrafted with melanoma tumor cells and received adoptive transfer of either **(A)** autologous *PDCD-1* KO TIL, **(B)** mock TIL (TIL electroporated without TALEN®), **(C)** mock TIL + anti–PD-1 antibody, or **(D)** no adoptive transfer of TIL; (n=14 each)
 - Tumor size was measured twice per week for 39 days
- **Product Release:**
 - Final *PDCD-1* KO TIL product was characterized for:
 - Total viable cells (TVC) and purity (% viability), determined by acridine orange/4',6-diamidino-2-phenylindole (DAPI) counterstain using the NucleoCounter® NC-200™ (ChemoMetec, Lillerød, Denmark) automated cell counter
 - Identity ($CD45^+CD3^+$ phenotype), assayed by immunofluorescence staining and flow cytometry
 - Potency (interferon-γ [IFNγ] release), assayed by ELISA using the Quantikine® IFNγ ELISA kit (R&D Systems, Minneapolis, MN, USA)
 - *PDCD-1* KO efficiency was evaluated based on PD-1 expression of *PDCD-1* KO TIL compared to mock TIL by flow cytometry
- **Phenotype:**
 - Final harvested *PDCD-1* KO TIL products were assayed for extended phenotypic markers using 2 multicolor flow cytometry panels to characterize TIL purity, identity, memory subset, activation, and exhaustion status
- **Characterization:**
 - **IL-2 Assay:** To assess the safety of the *PDCD-1* KO TIL product, the in vitro proliferative capacity of the final product in the absence of IL-2 was assessed over a period of 28 days
 - **Karyotyping Assay:** Cytogenetic examinations were performed by NeoGenomics Laboratories (Fort Myers, FL, USA). Briefly, cryopreserved *PDCD-1* KO TIL samples were rested and activated to harvest the metaphase cells for G-banding cytogenetic analysis. Three replicates of mitotic cells were analyzed, fixed, and stained to perform G-banding
- **Statistical Analysis:**
 - Unpaired Student t-test was used to analyze differences in phenotype, and Wilcoxon rank-sum test was used to detect differences in mouse in vivo studies; $p < 0.05$ was considered statistically significant

© 2022, Iovance Biotherapeutics

Results

Figure 1. *PDCD-1* KO TIL Show Increased Antitumor Activity

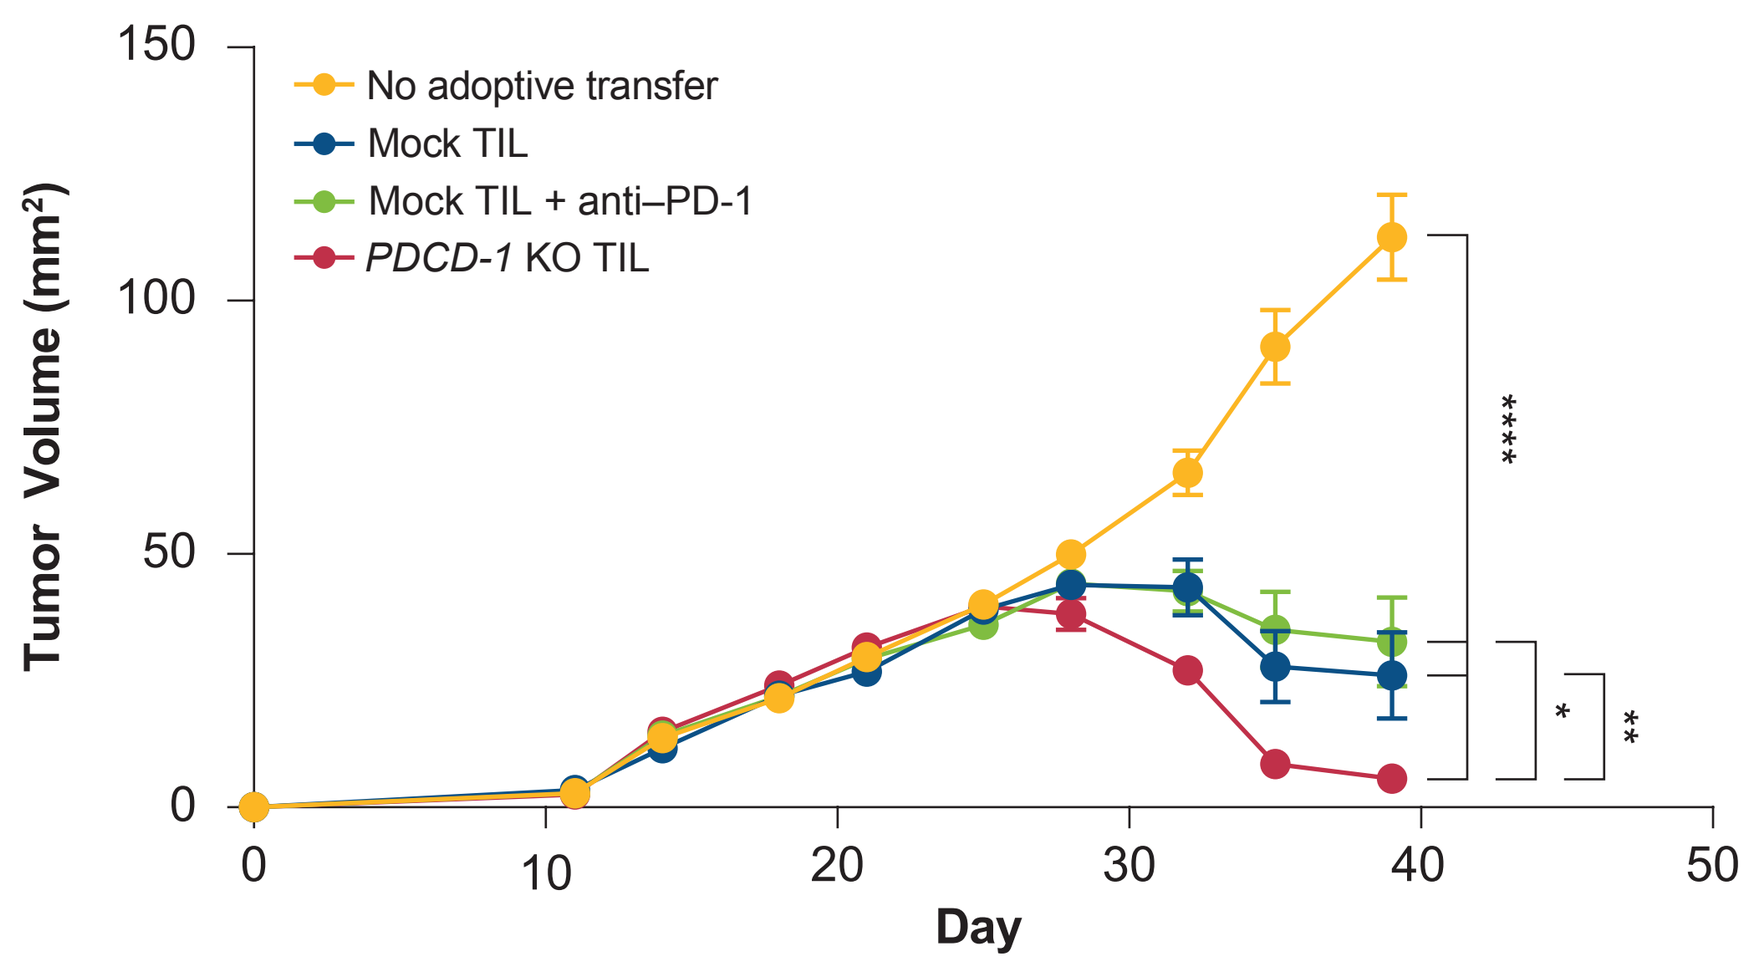
No adoptive transfer
Mock TIL
Mock TIL + anti–PD-1
PDCD-1 KO TIL
Tumor Volume (mm²)
150
100
50
0
0
10
20
30
40
50
Day

*
**

To assess the in vivo efficacy of *PDCD-1* KO TIL, hIL-2 NOG mice (n=14 per treatment group) engrafted with melanoma tumor cells were adoptively transferred with *PDCD-1* KO or mock TIL. Anti–PD-1 antibody treatment combined with mock TIL was included as a control for PD-1/PD-L1 blockade. Statistical significance is denoted by $^*p < 0.05$, $^{**}p < 0.01$, and $^{****}p < 0.0001$.

- Efficiency of *PDCD-1* KO for the autologous melanoma TIL used in the patient-derived xenograft (PDX) model was 75%, as assessed by flow cytometry
- Enhanced in vivo antitumor activity was observed in *PDCD-1* KO TIL-treated mice relative to mice treated with mock TIL alone or mock TIL + anti–PD-1 antibody

Figure 2. Viable Cell Dose, Purity, Identity, Potency, and *PDCD-1* KO Efficiency of TIL Product

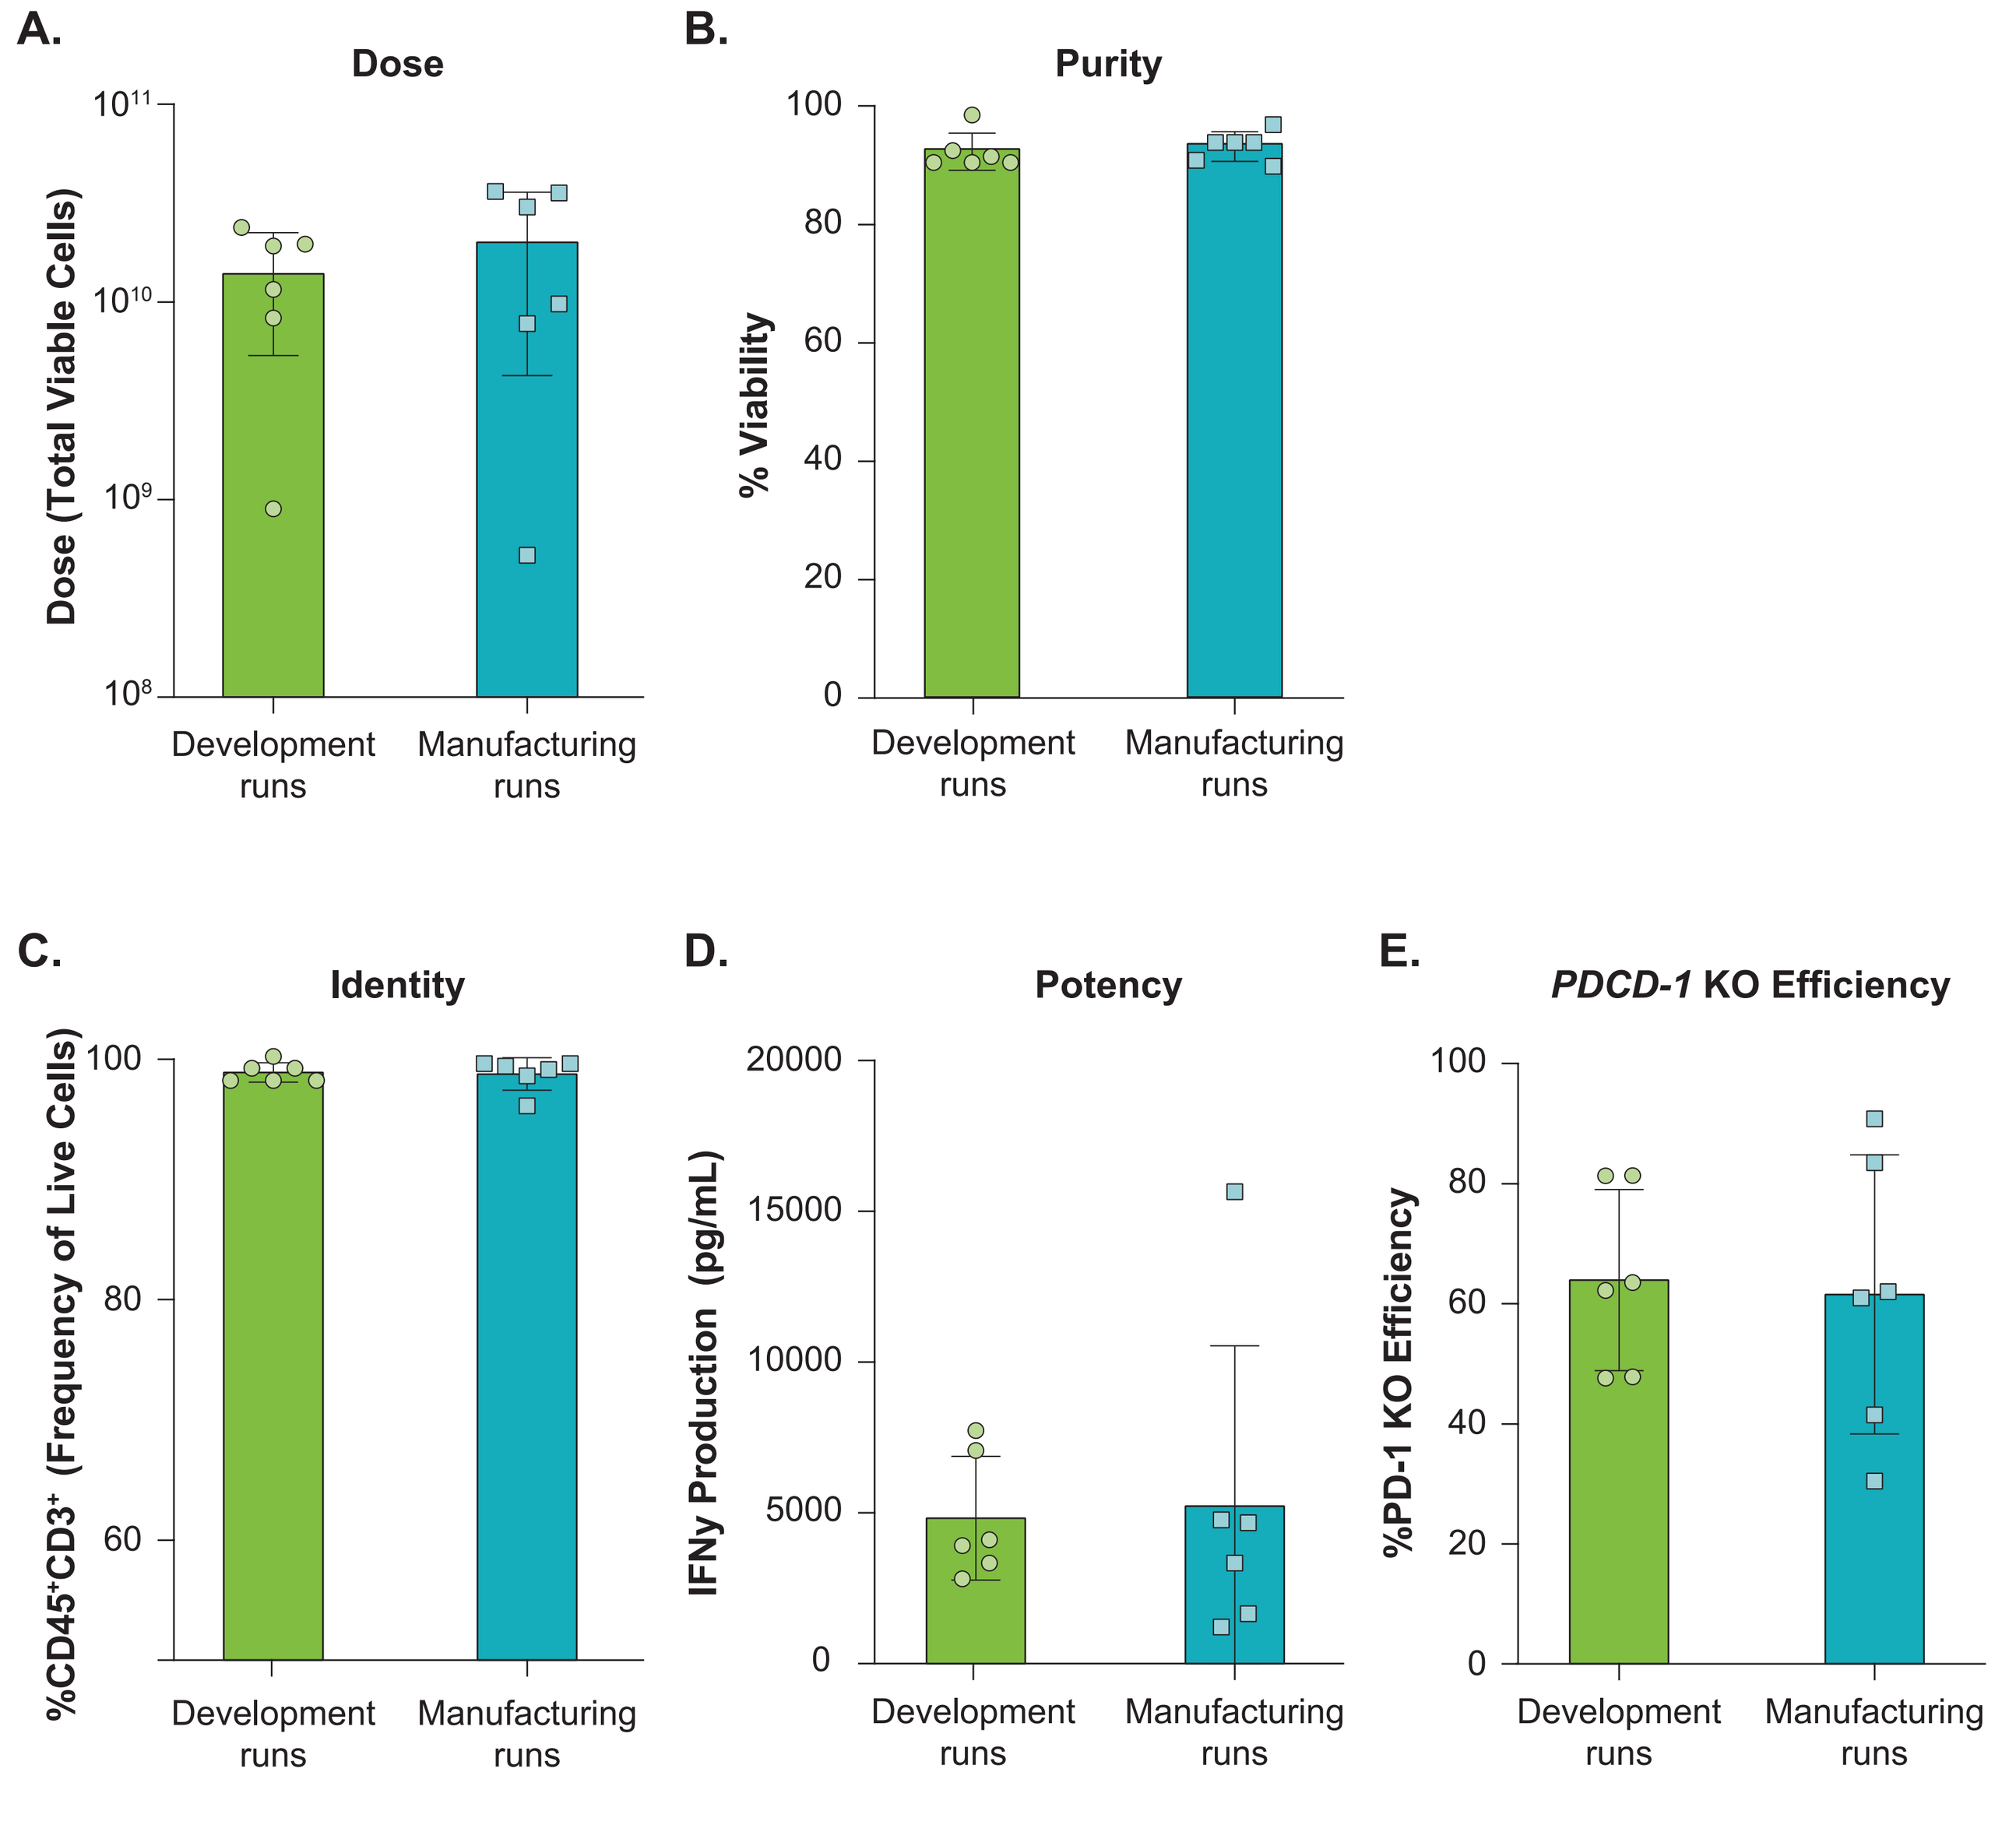
A.
Dose
Dose (Total Viable Cells)
10^{11}
10^{10}
10^9
10^8
B.
Purity
% Viability
C.
Identity
$\%CD45^+CD3^+$ (Frequency of Live Cells)
D.
Potency
IFNγ Production (pg/mL)
20000
15000
10000
5000
0
E.
PDCD-1 KO Efficiency
%PD-1 KO Efficiency
Development runs
Manufacturing runs

Values are displayed as mean ± SD.

- All *PDCD-1* KO TIL products met the release criteria for dose, purity, identity, potency, and *PDCD-1* KO efficiency. No statistically significant differences were observed between the development and manufacturing runs
 - Both development and manufacturing runs produced final *PDCD-1* KO TIL products of comparable dose **(A)** and viability **(B)**
 - The median (range) identity ($\%CD45^+CD3^+$) of the final TIL product in development and manufacturing runs was 98.5% (98%–100%) and 98.7% (96%–99%), respectively **(C)**
 - Median IFNγ release in development and manufacturing runs of the final *PDCD-1* KO TIL product was 4015 pg/mL and 4725 pg/mL, respectively **(D)**
 - Median (range) *PDCD-1* KO efficiency in development and manufacturing runs was 63% (48%–81%) and 62% (31%–91%), respectively **(E)**
 - *PDCD-1* KO TIL products were comparable to mock TIL in terms of growth, purity, identity, and potency
 - As shown in previous studies,[14] dose, purity, identity, and potency results were comparable between mock and *PDCD-1* KO in development runs (data not shown)

Figure 3. TIL Differentiation and Memory

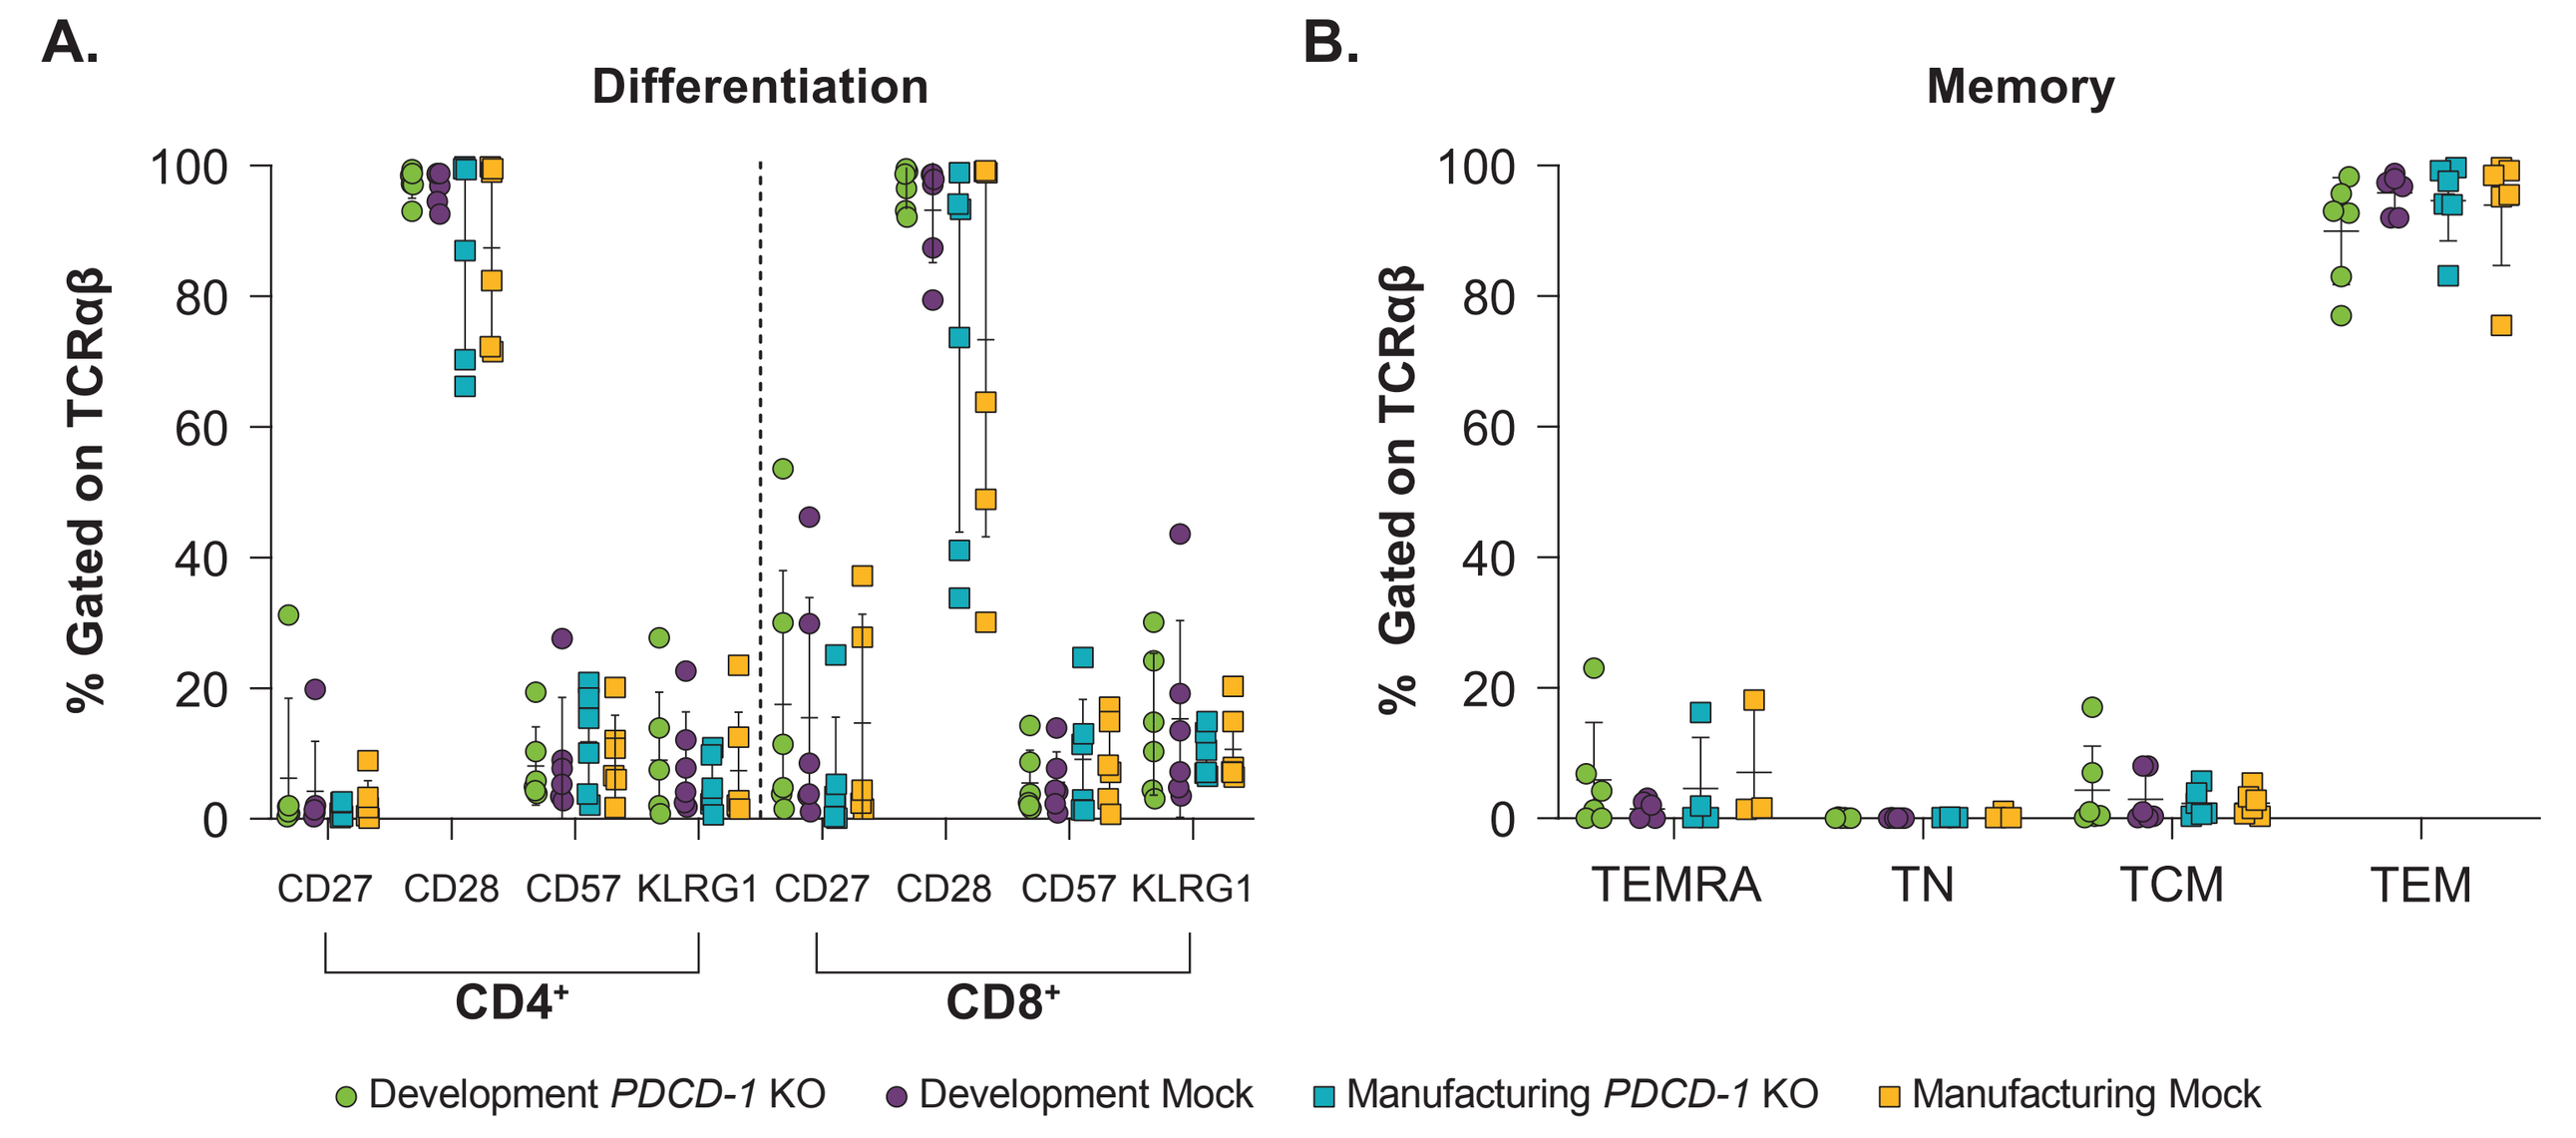
A.
Differentiation
% Gated on TCRαβ
CD27
CD28
CD57
KLRG1
CD4⁺
CD8⁺
B.
Memory
TEMRA
TN
TCM
TEM
Development PDCD-1 KO
Development Mock
Manufacturing PDCD-1 KO
Manufacturing Mock

- The CD28 marker was highly expressed in both *PDCD-1* KO and mock TIL in development and manufacturing samples, whereas other markers such as CD27, CD57, and KLRG1 were expressed at low levels **(A)**
- Naïve (TN), central memory (TCM), effector memory (TEM), and effector memory RA^+(TEMRA) T cell subsets were defined using CD45RA and CCR7 expression. A majority of the TIL lots displayed predominantly effector memory phenotype **(B)**
- No statistically significant differences in TIL differentiation markers or memory phenotype were observed between *PDCD-1* KO and mock TIL in the development and manufacturing runs

Figure 4. Expression of Activation- and Inhibitory-Related Markers on *PDCD-1* KO TIL

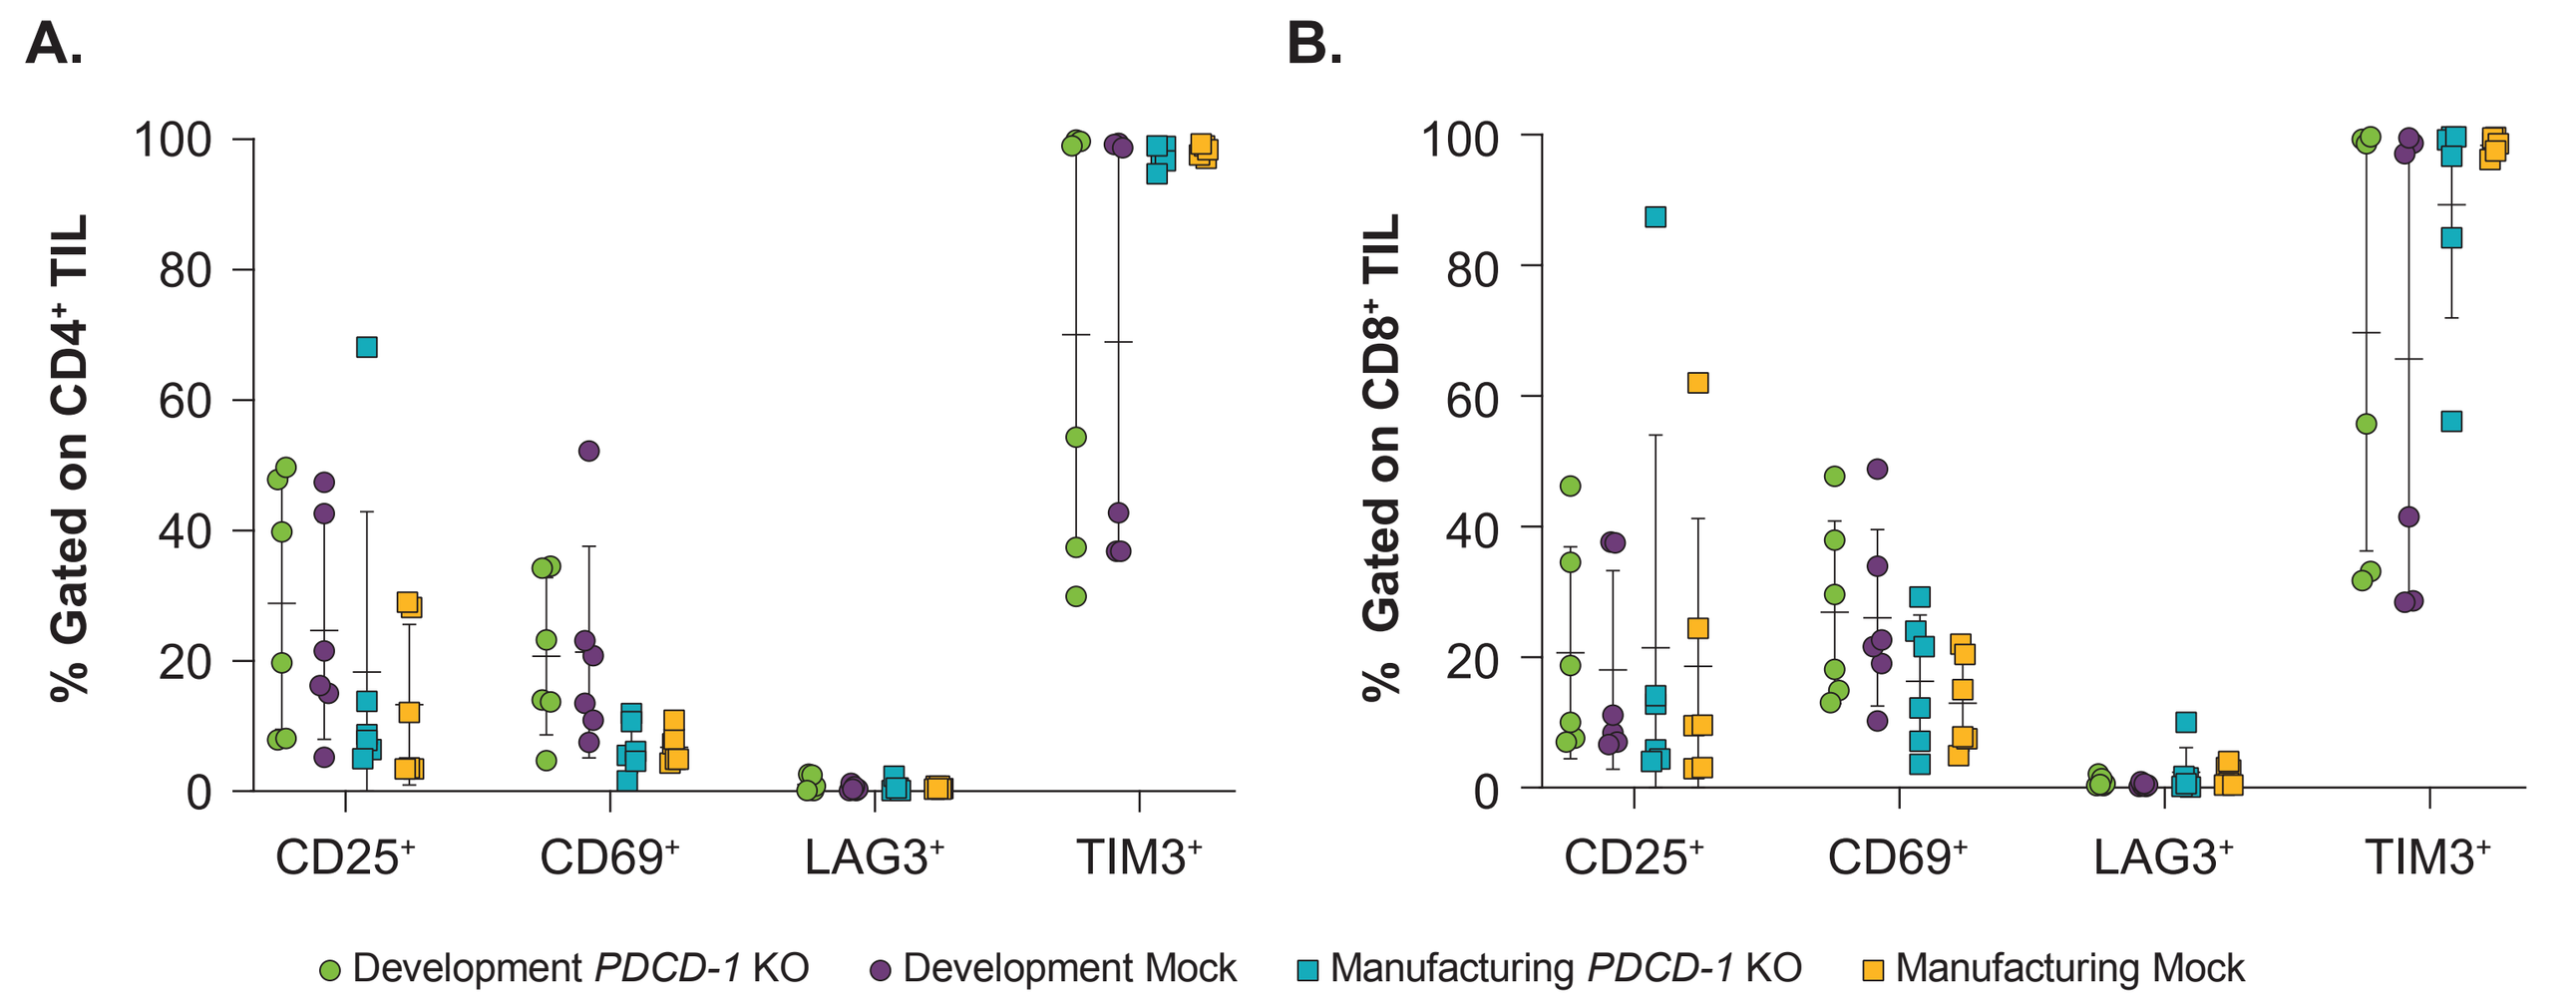
A.
% Gated on CD4⁺ TIL
B.
% Gated on CD8⁺ TIL
CD25⁺
CD69⁺
LAG3⁺
TIM3⁺
Development PDCD-1 KO
Development Mock
Manufacturing PDCD-1 KO
Manufacturing Mock

- Multicolor flow cytometry was used to characterize TIL activation and inhibitory receptor expression on $CD4^+$ **(A)** and $CD8^+$ TIL **(B)**
- No statistically significant differences in marker expression were observed between *PDCD-1* KO and mock TIL in the development and manufacturing runs

Figure 5. IL-2–Independent Proliferation Assay of *PDCD-1* KO TIL Products

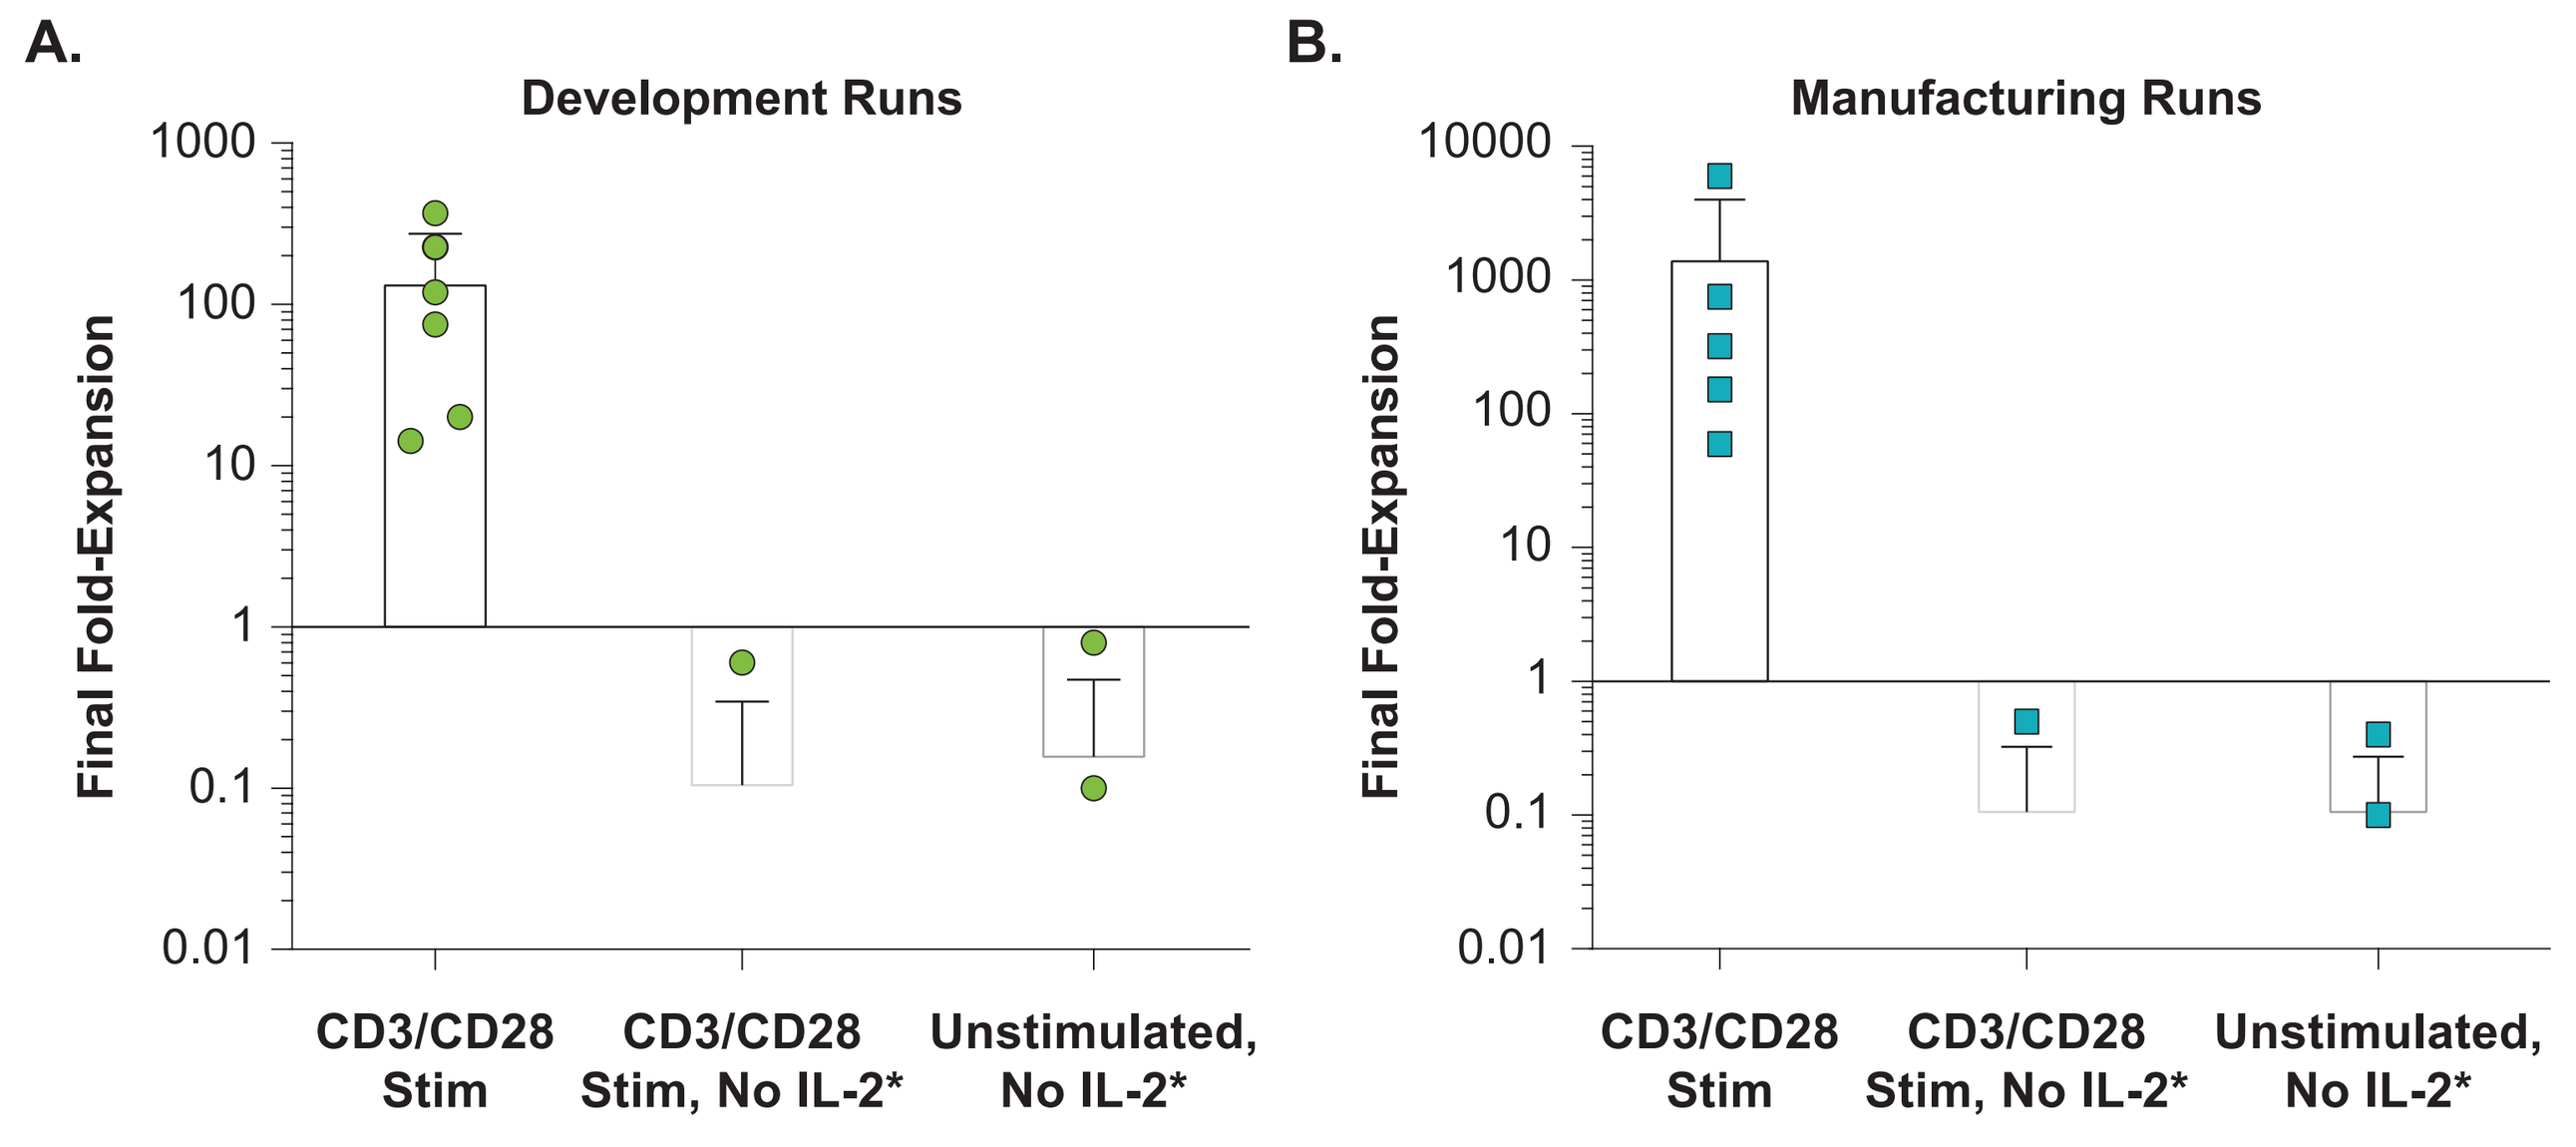
A.
Development Runs
B.
Manufacturing Runs
Final Fold-Expansion
CD3/CD28 Stim
CD3/CD28 Stim, No IL-2*
Unstimulated, No IL-2*

*Cell count data < lower limit of quantitation on the NC200 cell counter. Green circles and blue squares represent individual samples within the development and manufacturing runs, respectively, and values are displayed as mean + SD.

- Using an IL-2–independent proliferation assay, it was demonstrated that none of the *PDCD-1* KO TIL products underwent malignant transformation following TALEN®-mediated genome editing
 - None of the stimulated (anti-CD3/CD28) or unstimulated samples were proliferative in the absence of IL-2 **(A & B)**
 - All samples cultured in the presence of IL-2 showed proliferation **(A & B)**

Table 1. Summary of Karyotyping Results From *PDCD-1* KO TIL Products

	G-Banding Results	
Sample ID	**Mock**	***PDCD-1* KO**
1	Normal female: 46[a], XX[b], [20][c]	Normal female: 46[a], XX[b], [20][c]
2	Normal male: 46[a], XY[b], [20][c]	Normal male: 46[a], XY[b], [20][c]
3	Normal female: 46[a], XX[b], [20][c]	Normal female: 46[a], XX[b], [20][c]
4	Normal male: 46[a], XY[b], [20][c]	Normal male: 46[a], XY[b], [20][c]
5	Normal male: 46[a], XY[b], [20][c]	Normal male: 46[a], XY[b], [20][c]
6	Normal female: 46[a], XX[b], [20][c]	Normal female: 46[a], XX[b], [20][c]

[a]Number of chromosomes.
[b]Sex complement.
[c]Number of analyzed metaphase cells.

- No clonal chromosomal abnormalities were observed in G-banding analysis, indicating that there was no genotoxicity following TALEN®-mediated genome editing at *PDCD-1*
 - All samples analyzed by G-banding produced sufficient metaphases for a full study; normal G-banding patterns were observed

Conclusions

- The in vivo antitumor activity of *PDCD-1* KO TIL was superior to that of mock TIL (electroporated without TALEN®) in the presence or absence of anti–PD-1, suggesting that endogenous PD-1 inhibition may confer a functional advantage to TIL
- *PDCD-1* KO TIL clinical-scale manufacturing was feasible, and the TIL product quality attributes and phenotype were acceptable
 - TIL attributes in all development and manufacturing runs were comparable and met the product release criteria
 - None of the *PDCD-1* KO TIL products underwent malignant transformation following TALEN®-mediated genome editing as determined by IL-2–independent proliferation assay
 - No TALEN®-induced clonal chromosomal abnormalities were identified by G-banding
 - Importantly, lack of complete *PDCD-1* KO in the TIL product may spare other PD-1–dependent in vivo cellular functions
- Together, these data support clinical investigation of IOV-4001, an autologous *PDCD-1* KO TIL cell therapy, as a potential therapeutic option in patients with advanced solid tumors
 - A clinical study of IOV-4001 in patients with metastatic melanoma and advanced NSCLC is expected to begin in 2022

References

1. Sarnaik AA, Hamid A, Khushalani NI, et al. *J Clin Oncol*. 2021;39(24):2656-2666.
2. Larkin JMG, Sarnaik A, Chesney JA, et al. *J Clin Oncol*. 2021;39(suppl 15):Abstract 9505.
3. O'Malley D, Lee S, Psyrri A, et al. *J Immunother Cancer*. 2021;9(suppl 2):Abstract 492.
4. Thurber GM, Schmidt MM, Dane Wittrup K. *Adv Drug Deliv Rev*. 2008;60(12):1421-1434.
5. Less JR, Posner MC, Boucher Y, Borochovitz D, Wolmark N, Jain RK. *Cancer Res*. 1992;52(22):6371-6374.
6. Netti PA, Baxter LT, Boucher Y, Skalak R, Jain RK. *Cancer Res*. 1995:55(22):5451-5458.
7. Bordeau BM, Balthasar JP. *Cancer Biol Med*. 2021;18(3):649-664.
8. Saga T, Neumann RD, Heya T, et al. *Proc Natl Acad Sci U S A*. 1995;92(19):8999-9003.
9. Restifo NP, Dudley ME, Rosenberg SA. *Nat Rev Immunol*. 2018;12(4):269-281.
10. Postow MA, Sildow R, Hellmann MD. *N Engl J Med*. 2018;378:158-168.
11. Gautron AS, Juillerat A, Guyot V, et al. *Mol Ther Nucleic Acids*. 2017;9:312-321.
12. Menger L, Sledzinska A, Bergerhoff K, et al. *Cancer Res*. 2016;76:2087-2093.
13. Qasim W, Zhan H, Samarasinghe S, et al. *Sci Transl Med*. 2017;9(374):eaaj2013.
14. Ritthipichai K, Machin M, Lakshmipathi S, et al. Presented at the ESMO Virtual Congress; September 19–21, 2020. Abstract 1052P.
15. Jespersen H, Lindberg MF, Donia M et al. *Nat Commun*. 2017;8:707.

Abbreviations

CMO, contract manufacturing organization; DAPI, 4',6-diamidino-2-phenylindole; DOR, duration of response; GMP, Good Manufacturing Practice; hIL-2, human interleukin-2; ICI, immune checkpoint inhibitor; IFNγ, interferon-γ; IL-2, interleukin-2; LAG3, lymphocyte-activation gene 3; KO, knockout; LN_2, liquid nitrogen; ORR, objective response rate; PD-1, programmed cell death protein-1; PDX, patient-derived xenograft; REP, rapid expansion protocol; Stim, stimulated; TALEN®, transcription activator-like effector endonucleases; TCM, central memory T cells; TCR, T-cell receptor; TEM, effector memory T cells; TEMRA, effector memory RA^+ T cells; TIL, tumor-infiltrating lymphocytes; TIM3, T-cell immunoglobulin domain and mucin domain-3; TN, naïve T cells; TVC, total viable cells.

Acknowledgments

- The authors would like to thank the participating patients and their families for donation of material used in this study
- This study is sponsored by Iovance Biotherapeutics (San Carlos, CA, USA)
- Editorial support was provided by Second City Science (Chicago, IL, USA) and funded by Iovance Biotherapeutics

Disclosures

- AN, AV, AW, CH, VG, KO, MM, SW, MJ, and RC are employees of Iovance Biotherapeutics and have stock options
- JB has no financial disclosures