1052P ESMO VIRTUAL CONGRESS 2020 | 19-21 SEPTEMBER 2020

Genetic Modification of Iovance's TIL through TALEN®-mediated knockout of PD-1 as a strategy to empower TIL therapy for cancer

Krit Ritthipichai[1], Marcus Machin[1], Shwetha Lakshmipathi[1], Alexandre Juillerat[2], Laurent Poirot[3], Maria Fardis[1], Cécile Chartier[1]

[1]Iovance Biotherapeutics, Inc. San Carlos, CA, USA; [2]Cellectis, Inc, New York, NY, USA; [3]Cellectis S.A., Paris, France

IOVANCE BIOTHERAPEUTICS
ADVANCING IMMUNO-ONCOLOGY

For more information, please contact
Krit Ritthipichai, krit.ritthipichai@iovance.com

Introduction

Study Objectives

- To develop a robust process for the generation of TALEN®-mediated PD-1 knockout (KO) TIL.
- To phenotypically and functionally characterize PD-1 KO TIL and assess their therapeutic value.

Background

- Iovance's autologous tumor infiltrating lymphocytes (TIL) product has reported efficacy results in metastatic melanoma and cervical cancer patients that reflect an ORR of 36.4% and 44%, respectively.[1,2]
- One potential limitation of TIL therapy is T cell inactivation by the PD-1/PD-L1 pathway, providing rationale for combining TIL with anti-PD-1 antibodies.
- PD-1 pathway abrogation in TIL may enhance tumor cells killing while avoiding systemic adverse events mediated by anti-PD-1/PD-L1, thereby integrating two complementary benefits in a single treatment.
- Transcription activator-like effector endonucleases (TALEN®s) are hybrid molecules of a DNA binding domain and the FokI nuclease. Combination of two TALEN® arms directed at the *PDCD1* gene encoding PD-1 mediates DNA double strand break, leading to gene disruption and PD-1 knockout (KO).[3, 4, 5]
- TALEN®-edited CAR T cells were successfully and safely tested in the clinic.[5]
- In this study, we developed a robust process for the generation of TALEN®-mediated PD-1 KO TIL. Products obtained from several tumor histologies were characterized for their T-cell subset composition and the differentiation and activation status of those T cells, polyclonal distribution of KO, as well as their functionality.

Overview of TIL Therapy Process

1. The tumor is excised from the patient and transported to the GMP Manufacturing facility.
2. Upon arrival the tumor is fragmented and placed in media with IL-2 for a pre-Rapid Expansion Protocol (REP).
3. Pre-REP TIL are further propagated in a REP protocol in the presence of irradiated PBMCs, anti-CD3 antibody, and IL-2.
4. TIL products are assessed for phenotype and effector function.
5. Prior to infusion of expanded TIL, patients receive a non-myeloablative lymphodepletion regimen consisting of cyclophosphamide and fludarabine. Following infusion of TIL, patients receive a short duration (up to 6 doses) of high-dose IL-2 to support growth and engraftment of transferred TIL.

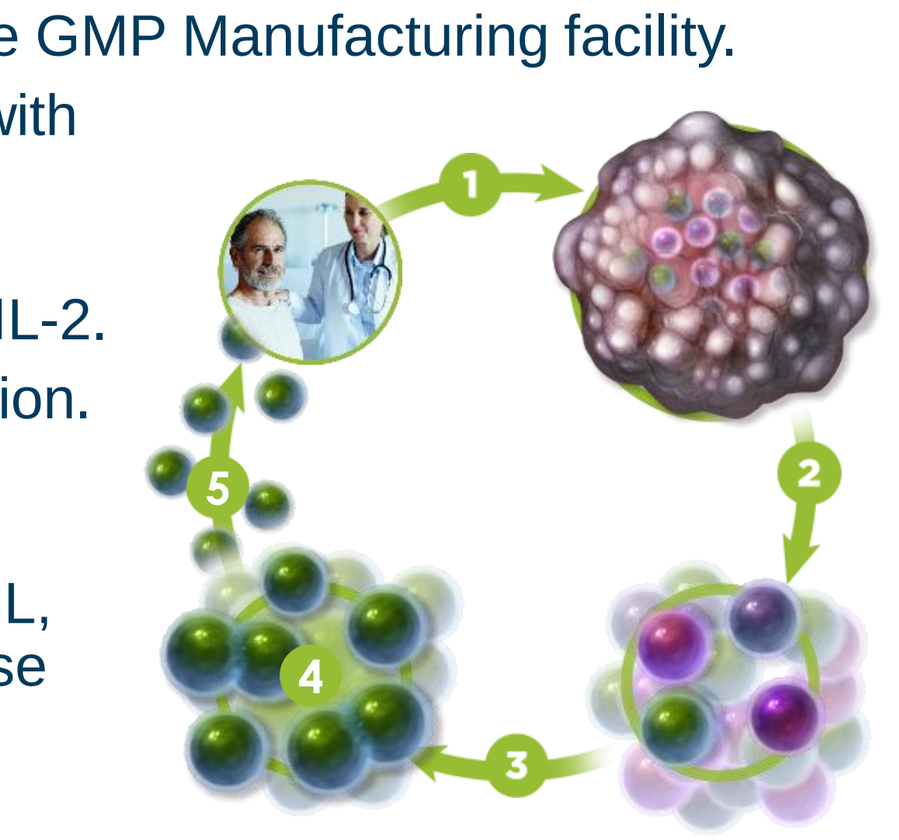
1
2
3
4
5

Experimental Design

- Tissue samples from melanoma and lung, breast, cervical, and ovarian cancers were used for this study.
- Several conditions of TIL electroporation with PD-1 TALEN® mRNAs were tested for optimization of KO efficiency during the TIL rapid expansion process used to manufacture Iovance's TIL (Gen 2).
- KO efficiency was assessed at the genomic and protein levels by NGS and flow cytometry, respectively.
- Impact of PD-1 KO on TIL count/viability, phenotype, and effector function was evaluated by flow cytometry and in cell-based assays including mixed lymphocyte reaction (MLR), redirected killing assay, and single-cell multiplexed cytokine profiling.
- Distribution of the KO across the T cell repertoire was monitored by flow cytometric T cell receptor (TCR)-V(β) repertoire analysis after separation of the cells by fluorescence-activated cell sorting (FACS).

PD-1 KO TIL Expansion Process

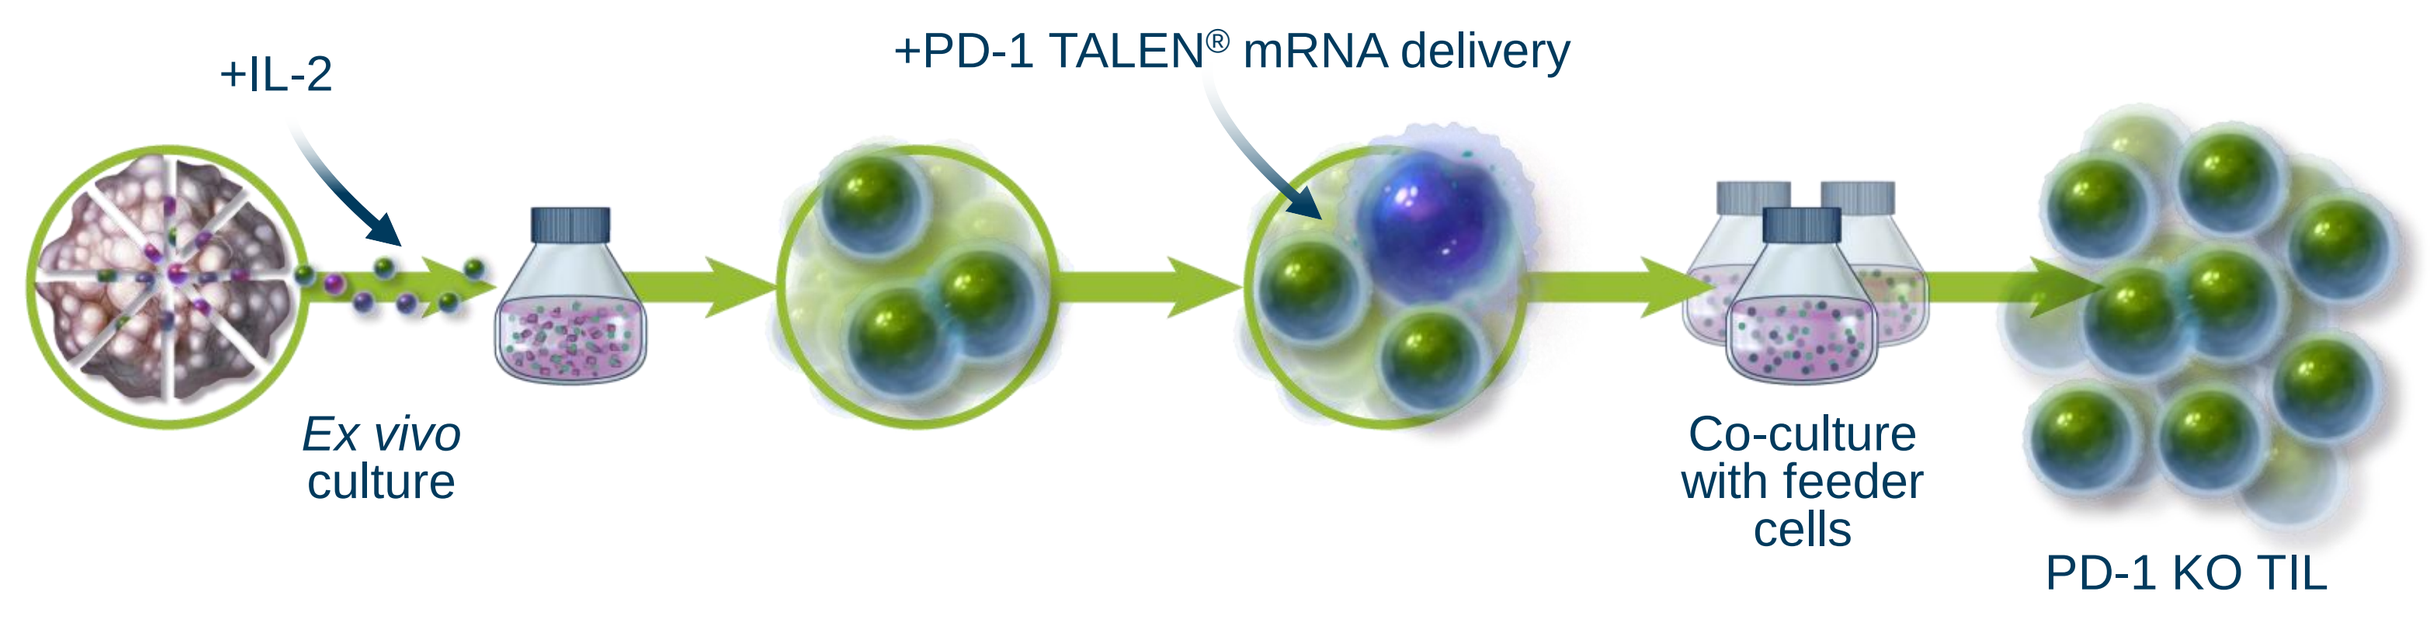
+IL-2
+PD-1 TALEN® mRNA delivery
Ex vivo culture
Co-culture with feeder cells
PD-1 KO TIL

© 2020, Iovance Biotherapeutics

Results

Figure 1. Addition of a PD-1 TALEN® electroporation step to Iovance's Generation 2 process mediated efficient PD-1 KO in TIL

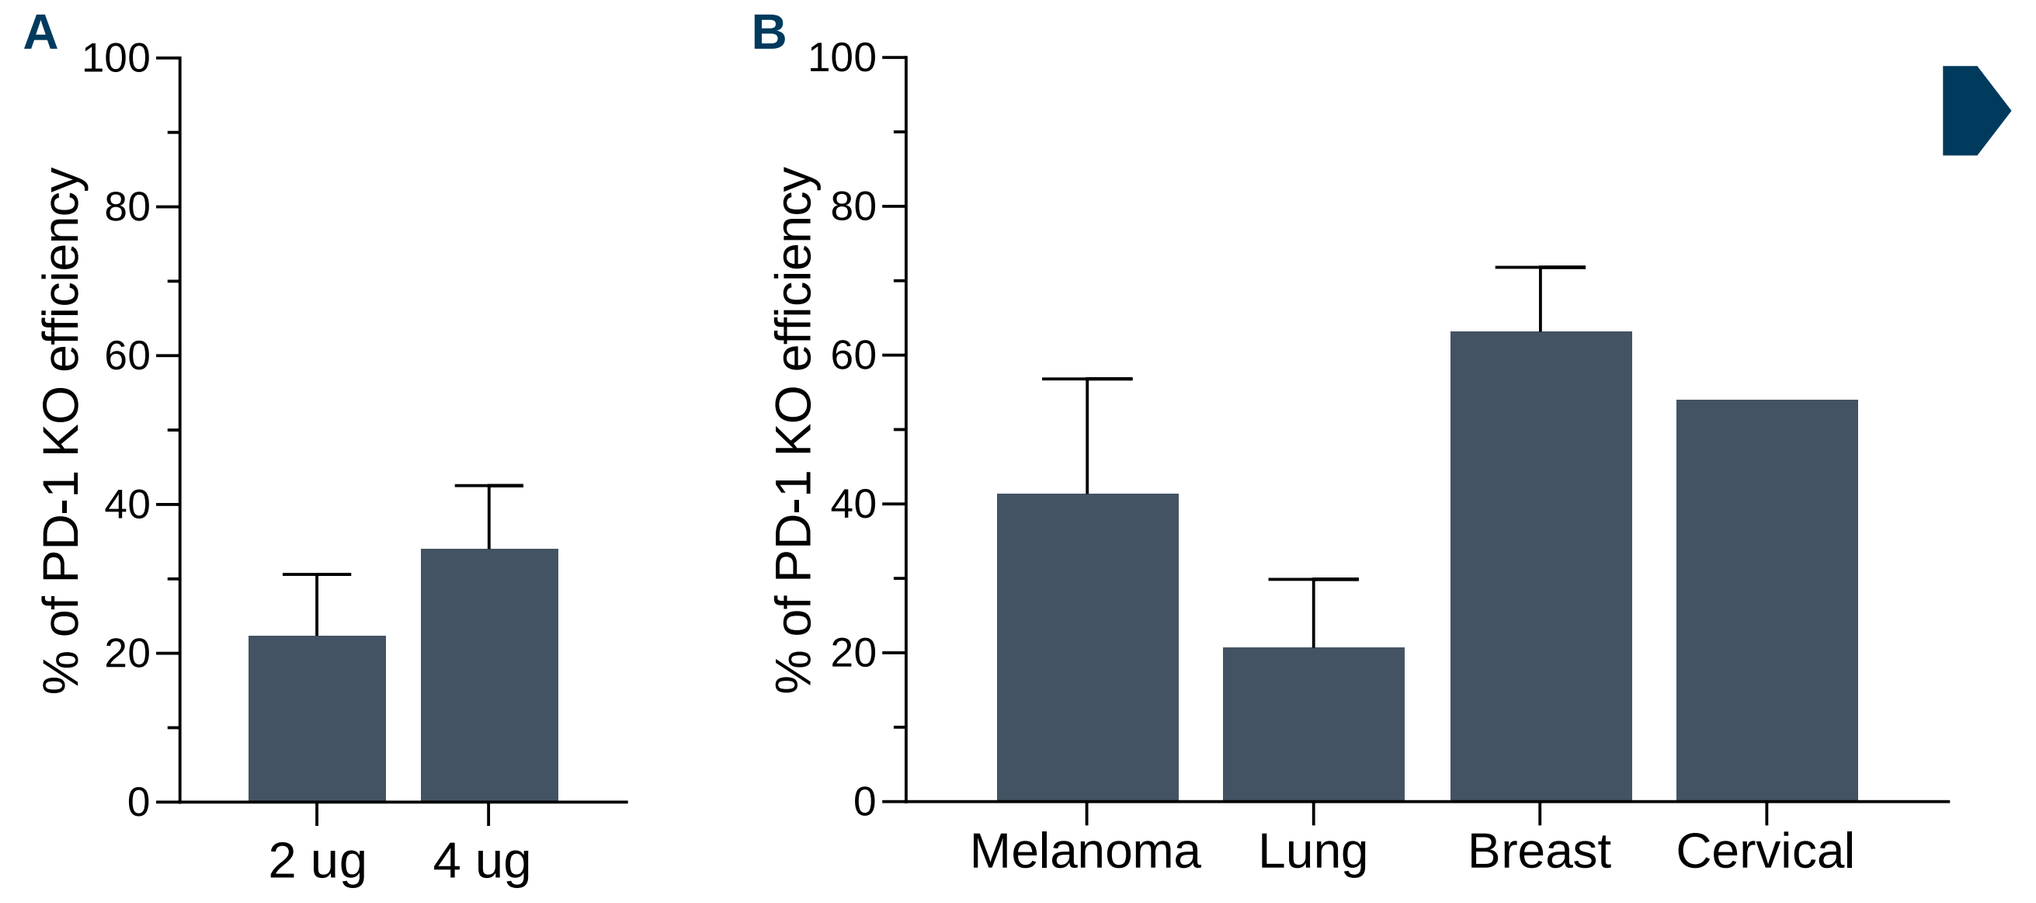
A
B
% of PD-1 KO efficiency
2 ug
4 ug
Melanoma
Lung
Breast
Cervical

The high electroporation efficiency led to up to 72% PD-1 KO efficiency in TIL.

Figure 1. PD-1 KO efficiency.
Electroporation efficiency was evaluated by GFP expression using flow cytometry one day after electroporation of 2 ug GFP mRNA in TIL from ovarian (n=1) and breast cancers (n=3) **(A)**. Shown is the average percentage of GFP-positive cells with standard error. PD-1 TALEN® mRNAs amounts of 2 and 4 ug were compared for KO efficiency in TIL from lung (n=3), breast (n=6), and ovarian cancer (n=1) **(A)**. PD-1 KO efficiency was assessed in TIL electroporated with 4 ug of PD-1 TALEN® mRNAs (or non-electroporation as control) from melanoma (n=3), lung (n=3), breast (n=3), and cervical cancer (n=1) **(B)**. Anti-CD3-induced PD-1 expression levels were measured post-TIL expansion, using flow cytometry, and PD-1 KO efficiency calculated by the formula (% PD-1 positive in non-electroporated TIL - % PD-1 positive in PD-1 KO TIL)/(% PD-1 positive in non-electroporated TIL) x100). Blue bars represent the mean KO efficiency calculated for each condition and are topped with standard errors.

Figure 2. PD-1 KO did not affect either TIL expansion or viability

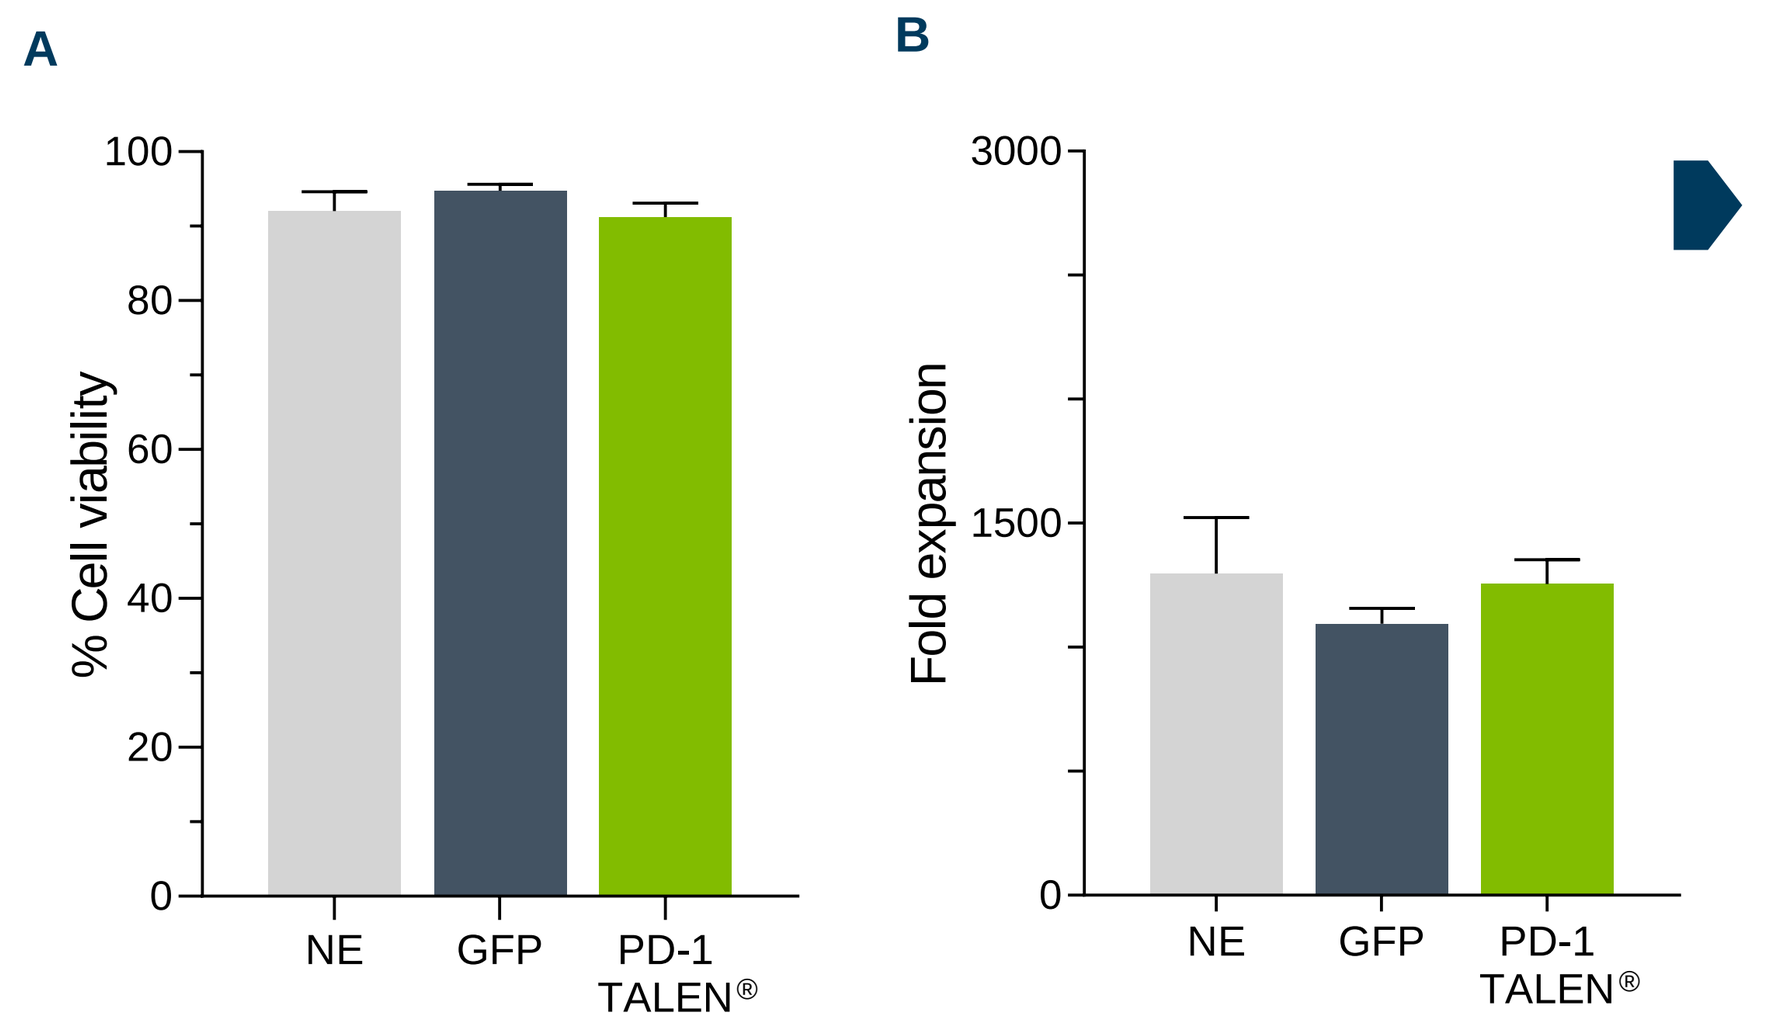
A
B
% Cell viability
Fold expansion
NE
GFP
PD-1 TALEN®

High PD-1 KO TIL viability and yield were comparable to those from unmodified TIL and thus adequate for therapeutic application.

Figure 2. TIL viability and fold expansion.
TIL from lung (n=1), ovarian (n=1), and breast cancers (n=4) were electroporated with 2 ug of PD-1 TALEN® (green) or GFP (blue) mRNAs or non-electroporated (grey). They were tested for cell viability and yield after the 11-day rapid expansion phase of Iovance Gen 2 process. Total number of viable cells was counted on Cellometer K2 and expressed as percentage of the total number of cells **(A)**. Fold expansion was calculated as the harvest cell count divided by the seeded cell count **(B)**. Mean values and standard errors are plotted for each condition.

Figure 3. PD-1 KO TIL exhibited comparable phenotype relative to control TIL

Figure 3. Phenotypic characterization of PD-1 KO TIL.
TIL from lung (n=1), ovarian (n=1), and breast cancers (n=4) were electroporated with 2 ug of PD-1 TALEN® (green) or GFP (blue) mRNAs or non-electroporated (grey). They were tested for the expression of T cell memory, differentiation, co-inhibition, and activation markers, using flow cytometry, after the 11-day rapid expansion of Iovance Gen 2 process. Three memory T-cell subsets including effector memory, central memory, and effector memory re-expressing CD45RA (T_{EMRA}) T cell subsets were defined as CCR7- CD45RA-, CCR7+ CD45RA-, and CCR7- CD45RA+, respectively **(A).** Differentiation markers included CD28, CD56, and KLRG-1 **(B).** Inhibitory markers included TIGIT and TIM-3 **(C).** Activation markers included CD69 and CD25 **(D)**. All results were expressed as percentages of CD3+ cells. Mean values and standard errors are shown.

PD-1 KO did not affect any of the phenotypic characteristics of unmodified TIL. Importantly, no compensatory mechanism such as the upregulation of other exhaustion markers was observed in PD-1 KO TIL.

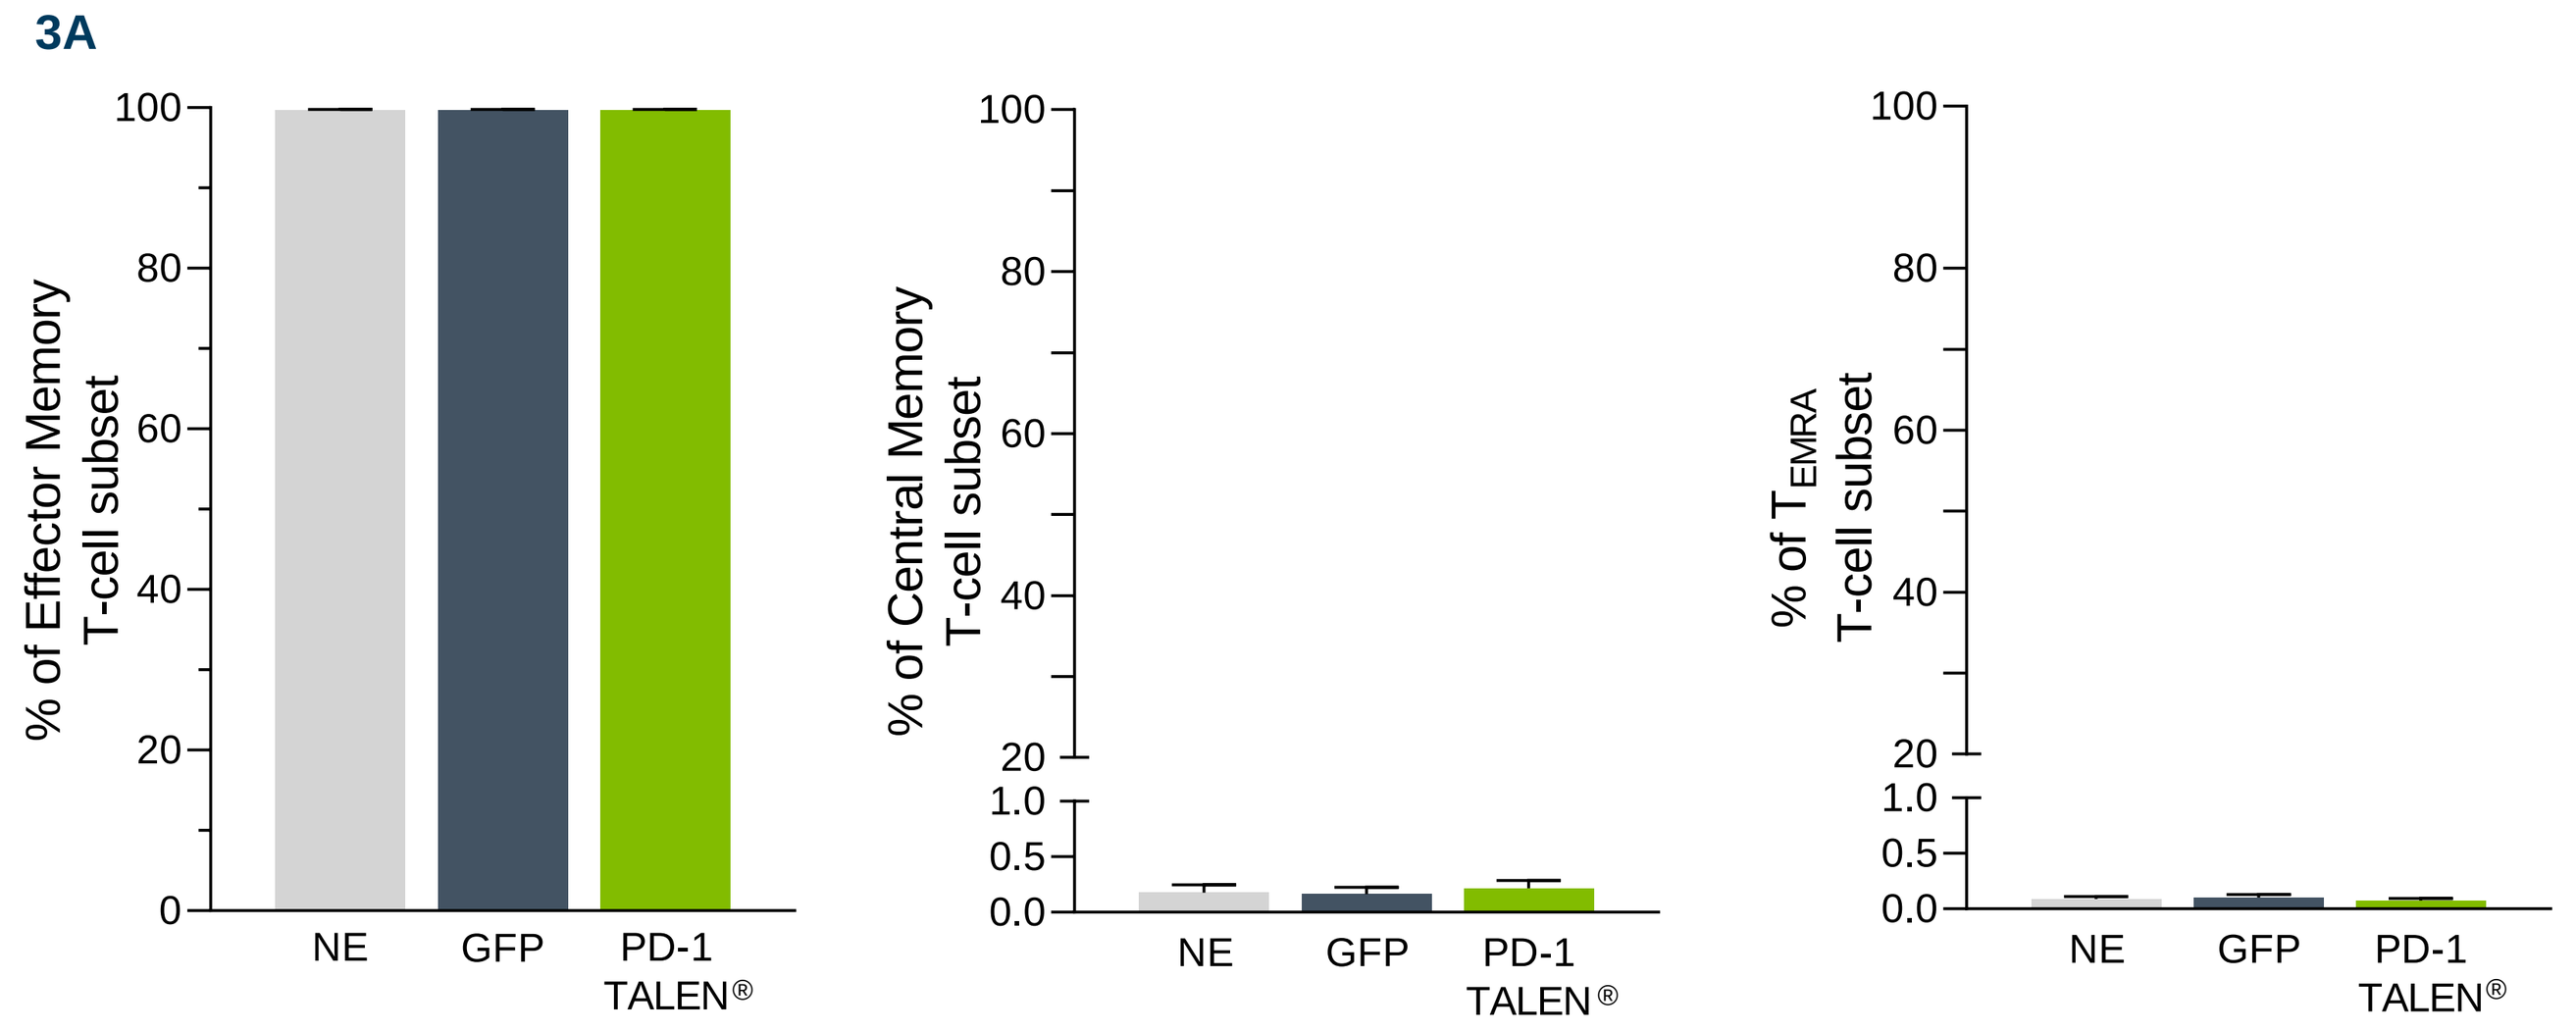
3A
% of Effector Memory T-cell subset
% of Central Memory T-cell subset
% of T_{EMRA} T-cell subset
NE
GFP
PD-1 TALEN®

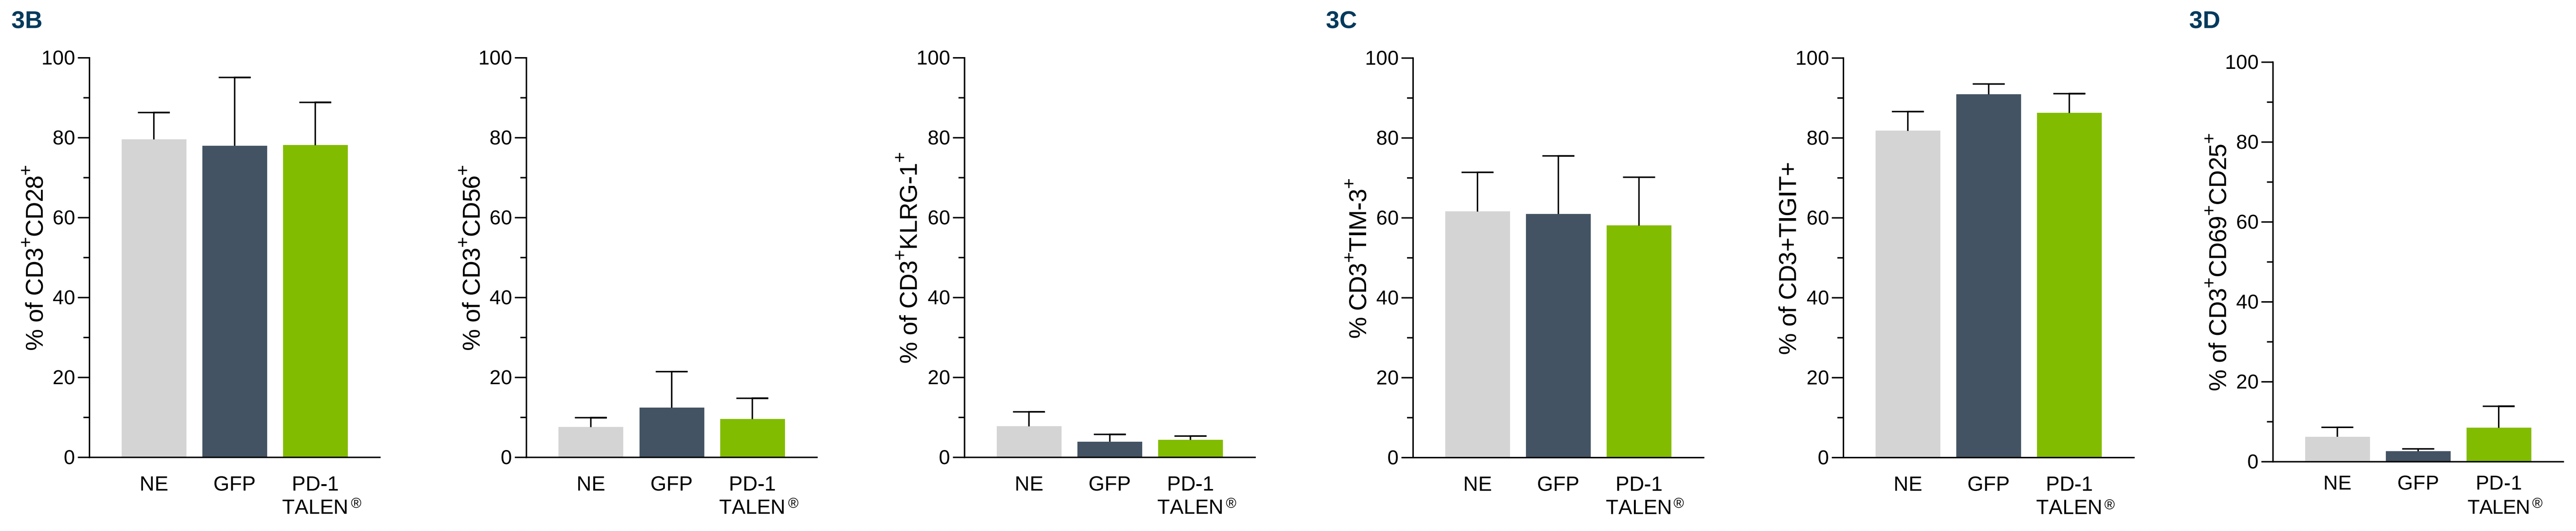
3B
% of CD3+CD28+
% of CD3+CD56+
% of CD3+KLRG-1+
3C
% CD3+TIM-3+
% of CD3+TIGIT+
3D
% of CD3+CD69+CD25+
NE
GFP
PD-1 TALEN®

Figure 4. PD-1 KO TIL were highly functional compared to control cells

Figure 4. Effector function of PD-1 KO TIL.
TIL from lung (n=1), ovarian (n=1), and breast (n=4) cancers were electroporated with 2 ug of PD-1 TALEN® (green) or GFP (blue) mRNAs or non-electroporated (grey). They were assessed for IFN-γ secretion and tumor killing capacity after rapid expansion. IFN-γ was detected by ELISA in the supernatants of TIL re-stimulated with anti-CD3 for 20 hours. Mean of IFN-γ concentrations (pg/ml) and standard errors are shown **(A)**. TIL were co-cultured with a recombinant PD-L1/TCR activator CHO cell line (BP Biosciences) in a 96-well electronic plate. Cell lysis was monitored by xCELLigence® Real-time cell analysis (RTCA) instrument for 60 hours and the data analyzed by RTCA pro software. Tumor killing capacity are shown as. Mean % of tumor cell lysis and standard errors are shown **(B)**.

PD-1 KO TIL demonstrated a robust level of IFN-γ production and killing capacity.

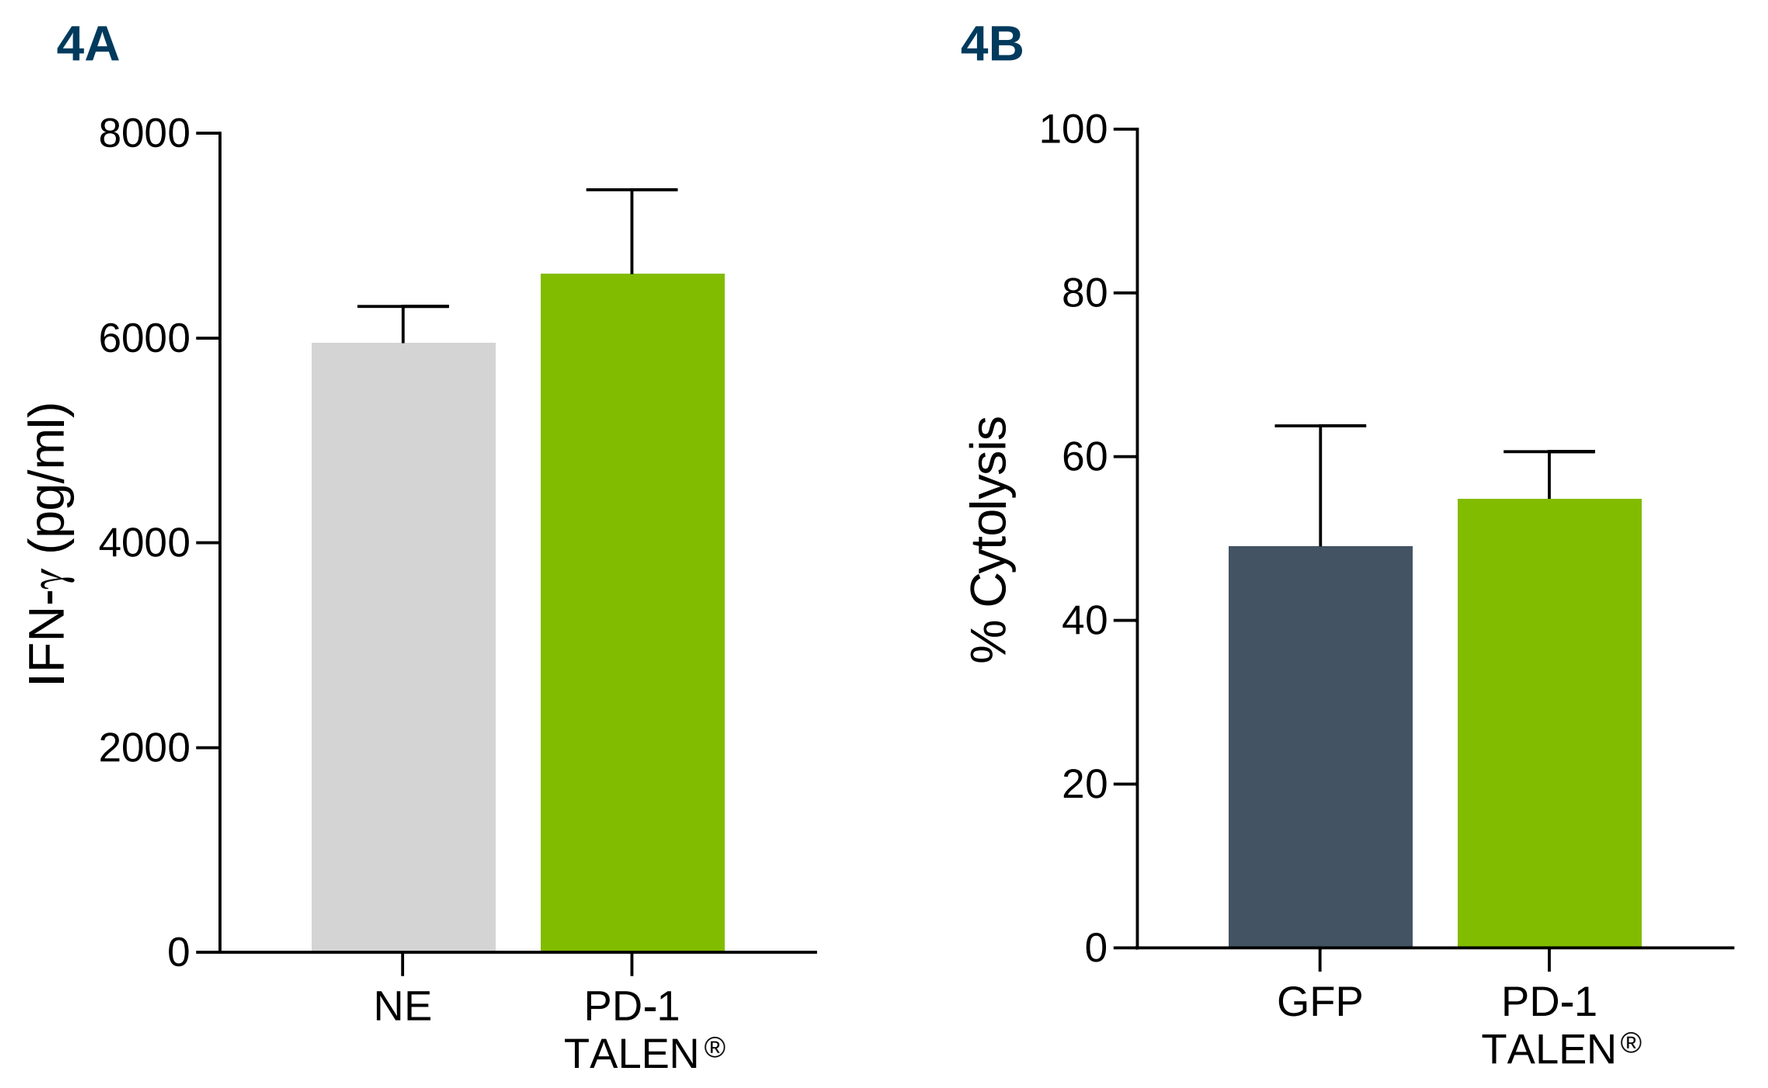
4A
IFN-γ (pg/ml)
NE
PD-1 TALEN®
4B
% Cytolysis
GFP
PD-1 TALEN®

References

1. Sarnaik A, *et al.* Safety and efficacy of cryopreserved autologous tumor infiltrating lymphocyte therapy (LN-144, lifileucel) in advanced metastatic melanoma patients who progressed on multiple prior therapies including anti-PD-1. *J Clin Oncol.* 2019;37:2518-2518.
2. Jazaeri A Sarnia, *et al.* Safety and efficacy of adoptive cell transfer using autologous tumor infiltrating lymphocytes (LN-145) for treatment of recurrent, metastatic, or persistent cervical carcinoma. *J Clin Oncol.* 2019;37:2538-2538.
3. Gautron, A. S., *et al.* 'Fine and Predictable Tuning of TALEN® Gene Editing Targeting for Improved T Cell Adoptive Immunotherapy', *Mol Ther Nucleic Acids.* 2017;9:312-21.
4. Menger, L., *et al.* 'TALEN®-Mediated Inactivation of PD-1 in Tumor-Reactive Lymphocytes Promotes Intratumoral T-cell Persistence and Rejection of Established Tumors', *Cancer Res.* 2016;76:2087-93.
5. Qasim, W., *et al.* 'Molecular remission of infant B-ALL after infusion of universal TALEN® gene-edited CAR T cells', *Sci Transl Med.* 2017;9(374).

Figure 5. Degree of reactivity of PD-1 KO TIL was slightly elevated relative to that of unmodified TIL

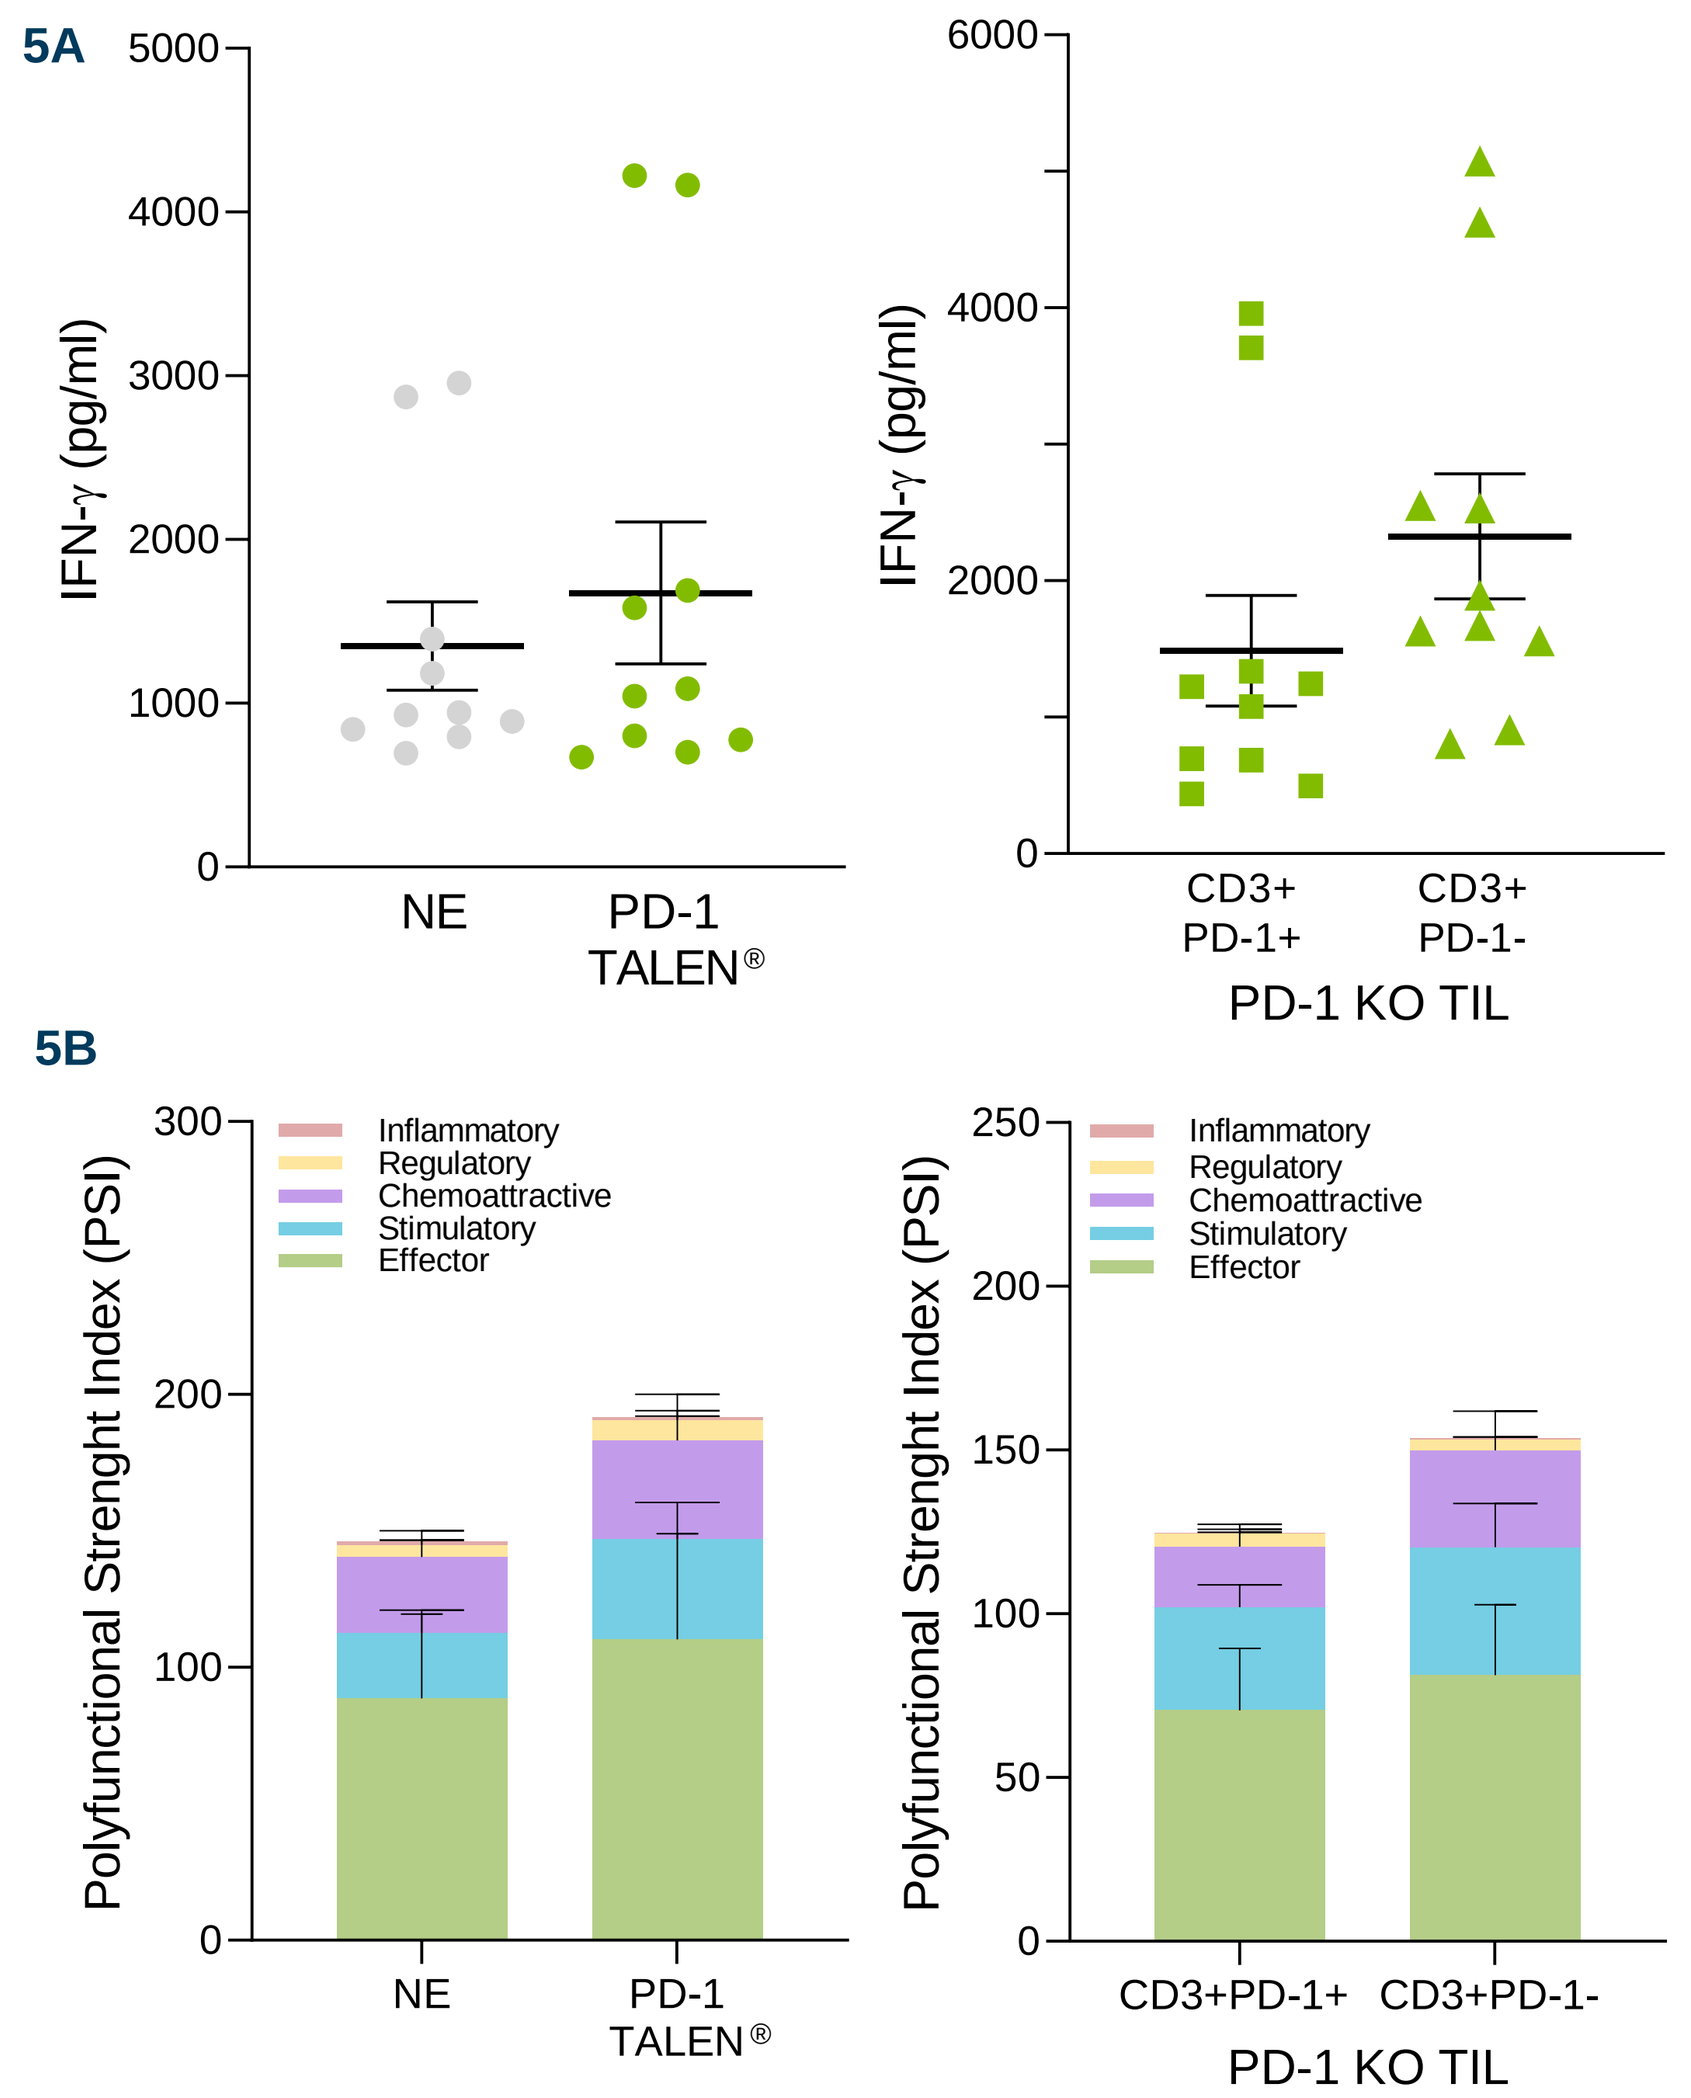
5A
IFN-γ (pg/ml)
NE
PD-1 TALEN®
CD3+ PD-1+
CD3+ PD-1-
PD-1 KO TIL
5B
Inflammatory
Regulatory
Chemoattractive
Stimulatory
Effector
Polyfunctional Strenght Index (PSI)
NE
PD-1 TALEN®
CD3+PD-1+
CD3+PD-1-
PD-1 KO TIL

Figure 5. PD-1 KO TIL reactivity.
TIL from breast cancer (n=3) and melanoma (n=1) were electroporated with 4 ug of TALEN® RNAs (green) or non-electroporated (gray). PD-1 positive (PD-1+) and PD-1 negative (PD-1-) cells were then sorted on a FACS instrument and bulk and sorted cells were tested for reactivity to Human leukocyte antigen (HLA)-mismatched mixed lymphocytes (3 days) and anti-CD3/CD28-coated beads (1 days). The supernatants of mixed lymphocyte reaction (MLR) were assessed for IFN-γ secretion using ELISA. Mean concentrations of IFN-γ (pg/ml) and standard errors are shown **(A)**. Single-cell polyfunctionality was assessed in TIL re-stimulated with anti-CD3/CD28 beads using the Isolight system. Polyfunctional strength index (PSI) was derived by multiplying the number of cytokines secreted per cell with the amount of each cytokine. Mean values and standard errors are shown **(B)**.

PD-1 KO may confer increased potency to TIL.

Figure 6. Clonal distribution was not altered by PDCD1 gene inactivation

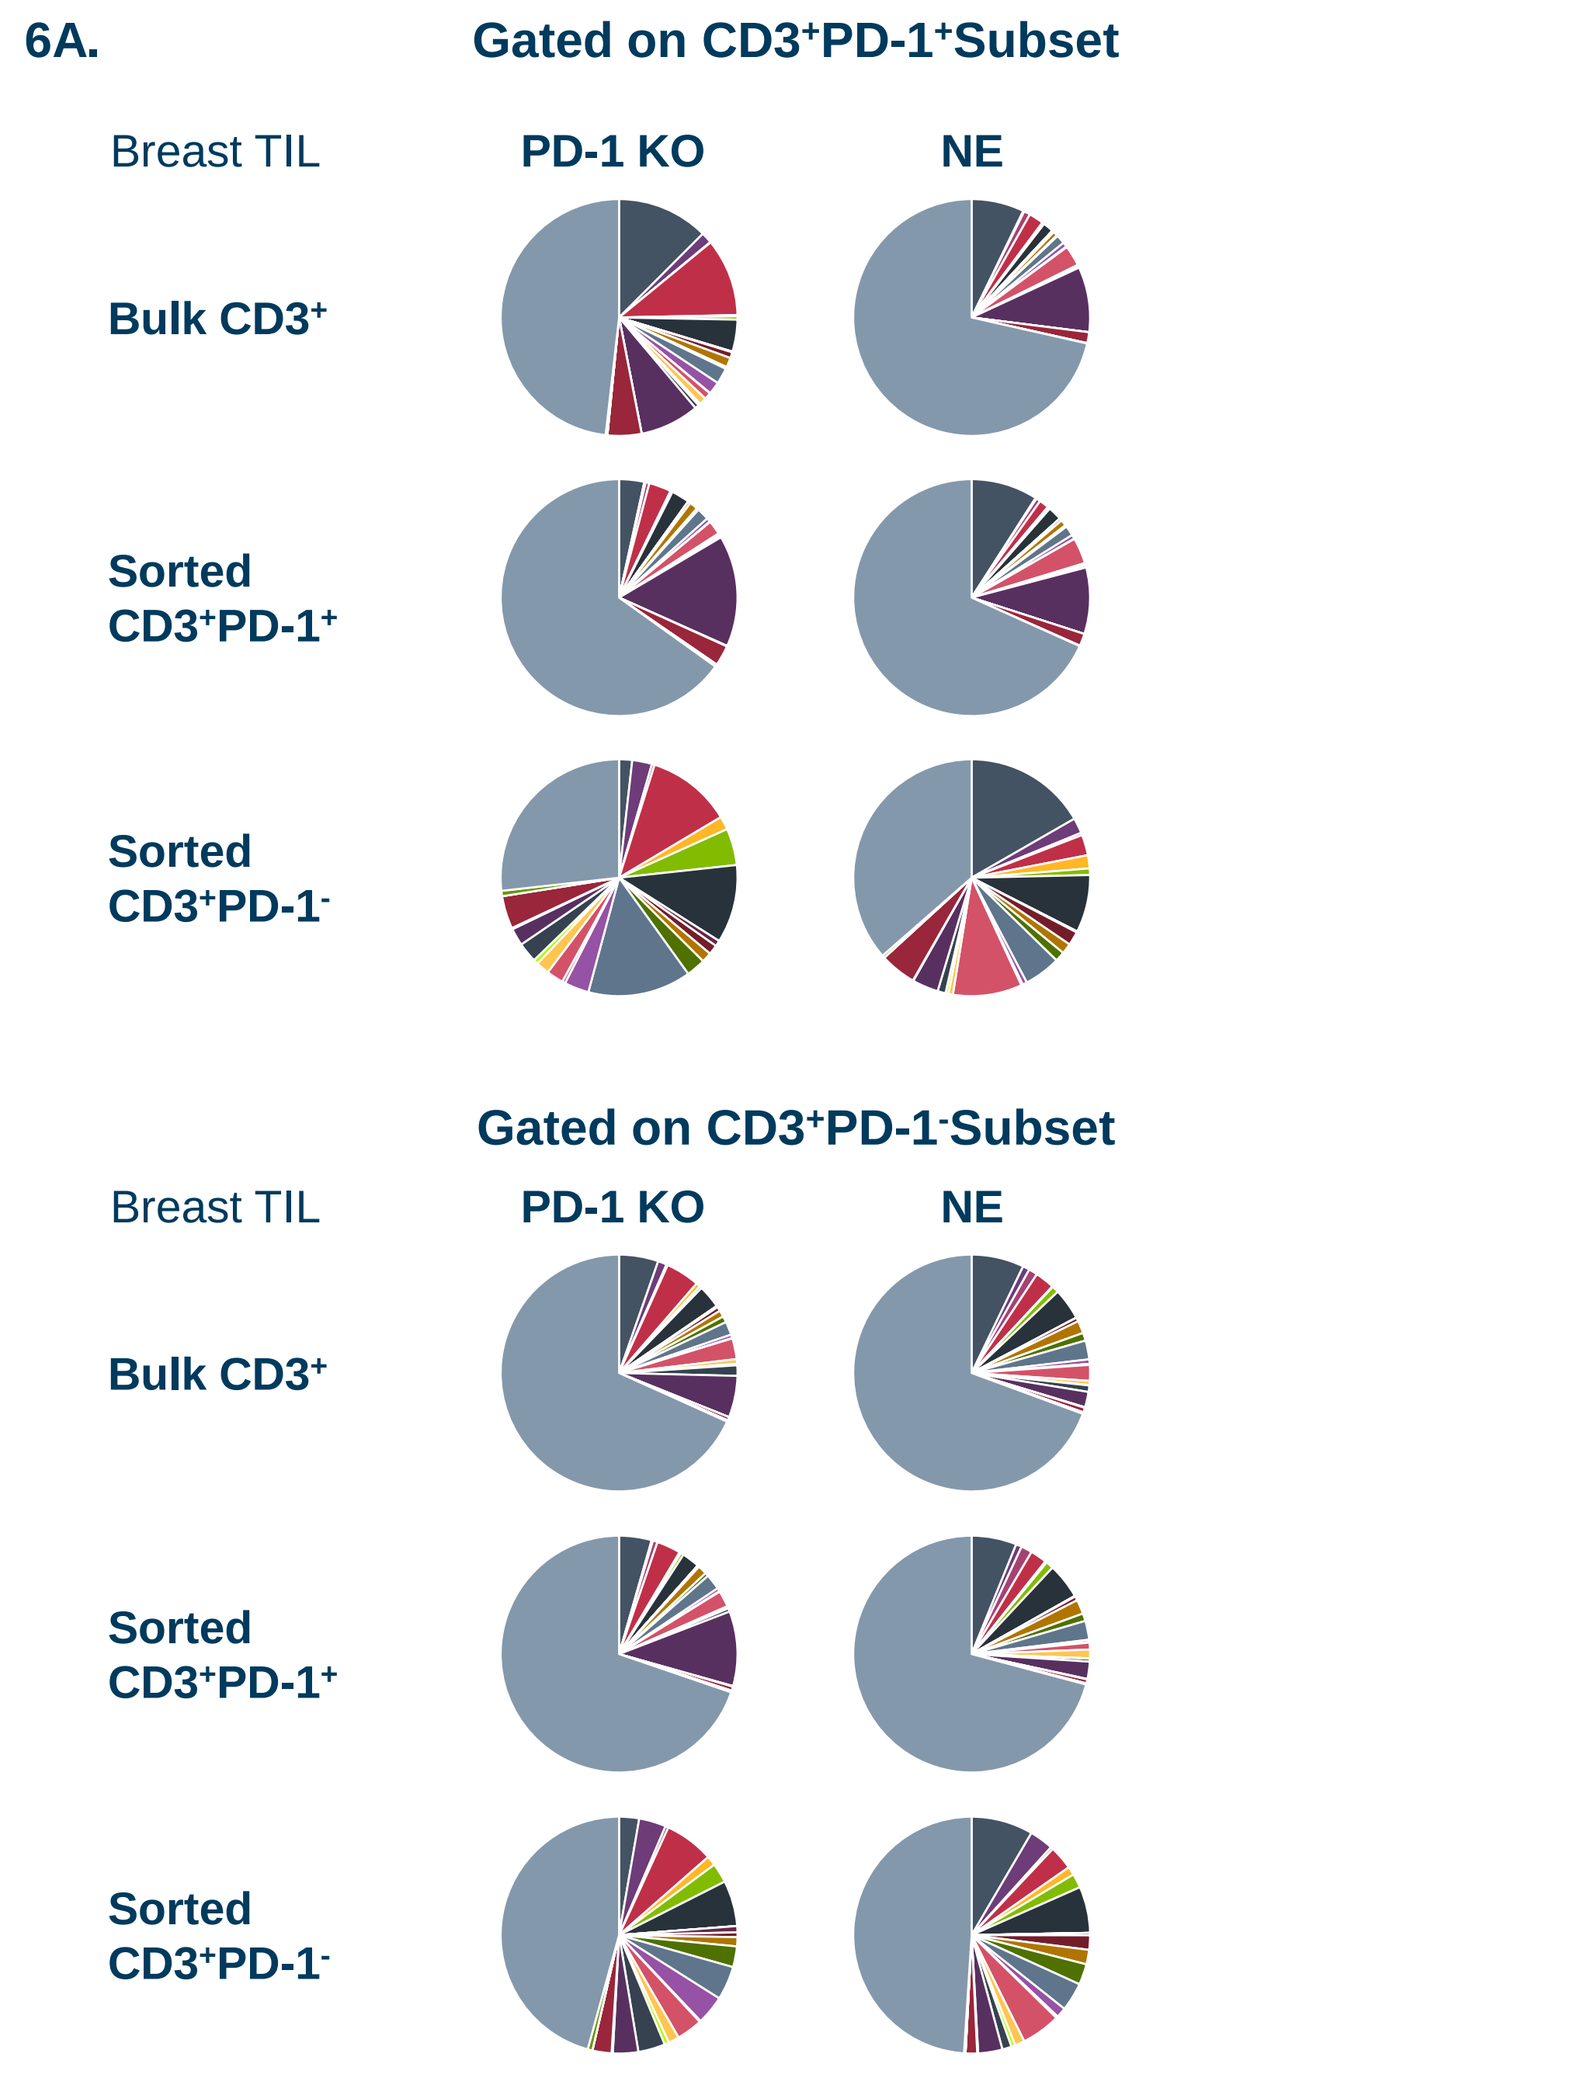
6A.
Gated on CD3+PD-1+Subset
Breast TIL
PD-1 KO
NE
Bulk CD3+
Sorted CD3+PD-1+
Sorted CD3+PD-1-
Gated on CD3+PD-1-Subset

Figure 6. Assessment of TCR repertoire & *PDCD1* gene editing.
TIL from one lung and one breast cancers were sorted into $CD3^+PD\text{-}1^+$ and $CD3^+PD\text{-}1^-$ subsets. The cells were electroporated with 4 ug of PD-1 TALEN® mRNAs and expanded TIL were re-stimulated with anti-CD3 and assessed for PD-1 and TCR Vβ repertoire. **(A)** A flow cytometry-based assay was used to identify the TCR Vβ subtypes present in the various experimental populations. The data were analyzed using the FlowJo software. The percentage of TCR repertoires are plotted in representative pie charts from breast TIL. Colors represent each TCR Vβ repertoire.

The relatively unbiased distribution of PD-1 KO across the T cell repertoire that constitutes the polyclonal TIL product suggests that the modification will affect the broad spectrum of TIL.

Conclusion

- A robust process for the generation of TALEN®-mediated PD-1 KO TIL and their expansion to therapeutically relevant numbers was successfully established.
- PD-1 KO TIL displayed the combination of memory, differentiation, and activation markers characteristic of the highly effective unmodified TIL product. No compensatory upregulation of T cell inhibitory markers was observed.
- PD-1 KO may confer increased potency to TIL, as suggested by the robust effector function displayed in all in vitro assays
- Based on clonal distribution studies, PD-1 KO is expected to affect the full spectrum of diverse and polyclonal TIL population.
- Overall, the data support the development of PD-1 KO TIL to possibly replace the combination of TIL and anti-PD-1 antibody in enhancing the potency of TIL immunotherapy

Disclosures

- This study and poster are sponsored by Iovance Biotherapeutics, Inc.
- Authors are employees of Iovance Biotherapeutics, Inc. or Cellectis, Inc. and may have stock options.

Acknowledgement

- All listed authors meet the criteria for authorship set forth by the International Committee for Medical Journal Editors.
- The authors would like to thank the participating patients and their families for donation of material used in this study.