422a ASCO ANNUAL MEETING, JUNE 1-5, 2018 | McCORMICK PLACE, CHICAGO, IL, USA

A Phase 2, Multicenter Study to Assess the Efficacy and Safety of Autologous Tumor Infiltrating Lymphocytes (LN-144) for Treatment of Patients with Metastatic Melanoma

Amod Sarnaik[1], Brendan Curti[2], Diwakar Davar[3], John M. Kirkwood[3], Omid Hamid[4], Jose Lutzky[5], Melissa Wilson[6], Harriet Kluger[7], Eric Whitman[8], Giao Phan[9], Sajeve Samuel Thomas[10], Karl Lewis[11], Hendrik-Tobias Arkenau[12], Bente Larsen[13], Igor Gorbatchevsky[13], Corina Andresen[13], Sam Suzuki[13], Nancy Samberg[13], Maria Fardis[13], Jason Chesney[14]

[1]Moffitt Cancer Center, Tampa, FL; [2]Earle A. Chiles Research Institute, Providence Cancer Center, Portland, OR; [3]University of Pittsburgh Medical Center – Hillman Cancer Center, Pittsburgh, PA; [4]The Angeles Clinic and Research Institute, Los Angeles, CA; [5]Mount Sinai Comprehensive Cancer Center, Miami Beach, FL; [6]New York University – Perlmutter Cancer Center – Langone Medical Center, New York, NY; [7]Yale Cancer Center, New Haven, CT; [8]Atlantic Health System Cancer Care, Morristown, NJ; [9]Virginia Commonwealth University, Richmond, VA; [10]University of Florida Health Cancer Center, Orlando, FL; [11]University of Colorado Cancer Center, Denver CO; [12]Sarah Cannon Research Institute London UK; [13]Iovance Biotherapeutics, San Carlos, CA; [14]James Graham Brown Cancer Center, Louisville, KY

IOVANCE BIOTHERAPEUTICS
ADVANCING IMMUNO-ONCOLOGY
999 Skyway Road, STE 150, San Carlos, CA 94070
For more information, please contact:
Amod.Sarnaik@providence.org
Clinical.Inquiries@iovance.com

BACKGROUND

- There are an estimated 232,000 newly diagnosed cases of melanoma skin cancers occurring globally each year, making it the fifth most common malignancy in men and sixth most common malignancy in women.[1]
- With the approval of ipilimumab in 2011, followed by approvals for pembrolizumab and nivolumab in 2014, there has been rapid implementation of these immunotherapeutic agents with variable impact on patient outcomes, while requiring management of significant toxicities.[2]
- Adoptive cell therapy (ACT) utilizing tumor infiltrating lymphocytes (TIL) for patients with metastatic melanoma and other solid tumors, demonstrates potentially durable and complete responses, even in heavily pretreated patients in studies conducted by the National Cancer Institute.[3]
- The C-144-01 study will enroll metastatic melanoma patients who have progressed following anti-PD-1 therapy, and BRAF inhibitor, if BRAF mutation positive.
- This phase 2 multicenter trial utilizes a central GMP facility for the manufacture of LN-144 in either a non-cryopreserved generation 1 (Gen 1), or cryopreserved generation 2 (Gen 2) investigational TIL infusion product.

Figure 1. Iovance cryopreserved LN-144 manufacturing process (22 days)

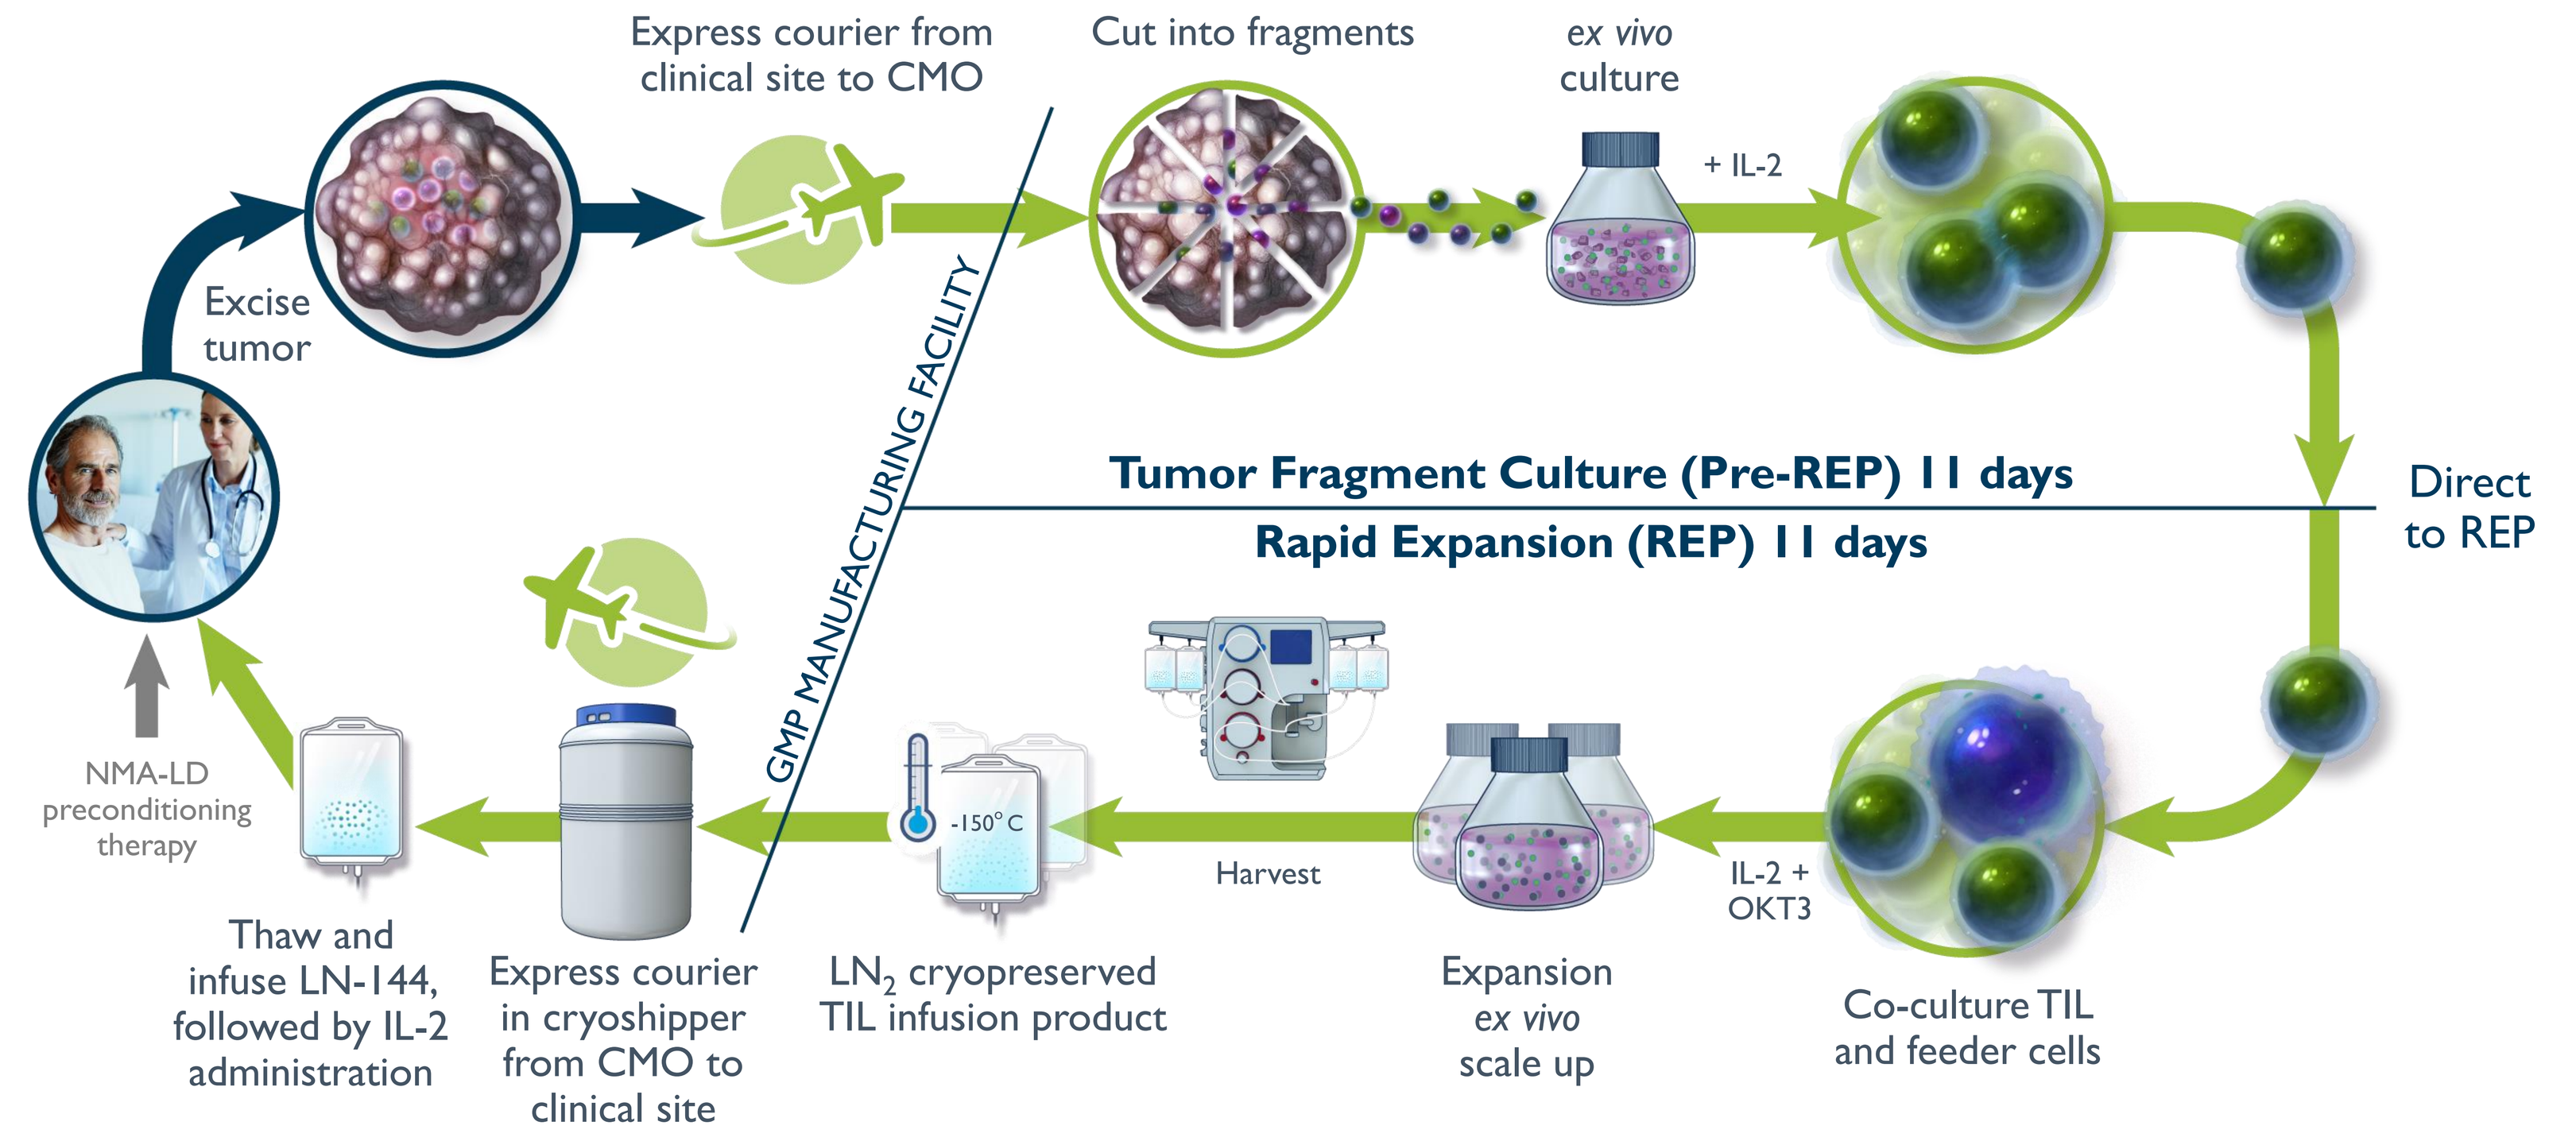
Express courier from clinical site to CMO
Cut into fragments
ex vivo culture
+ IL-2
Excise tumor
GMP MANUFACTURING FACILITY
Tumor Fragment Culture (Pre-REP) 11 days
Rapid Expansion (REP) 11 days
Direct to REP
NMA-LD preconditioning therapy
-150° C
Harvest
IL-2 + OKT3
Thaw and infuse LN-144, followed by IL-2 administration
Express courier in cryoshipper from CMO to clinical site
LN2 cryopreserved TIL infusion product
Expansion ex vivo scale up
Co-culture TIL and feeder cells

STUDY OVERVIEW

- Patients must have confirmed diagnosis of Stage IIIC or Stage IV Metastatic Melanoma with progression on/after prior immune checkpoint (anti-PD-1) therapy and, if BRAF mutation positive, after BRAF inhibitor systemic therapy.
- Up to 60 investigational centers in the US & Europe, to enroll approximately 85 patients.
- The study consists of 3 treatment cohorts:
 - Cohort 1: patients to receive Gen 1/non-cryopreserved LN-144; enrollment closed
 - Cohort 2: patients to receive Gen 2/cryopreserved LN-144
 - Cohort 3: patients from Cohort1/Cohort 2 to receive a 2nd treatment with LN-144 therapy
- Patients receive a nonmyeloablative lymphodepletion (NMA-LD) preparative regimen (cyclophosphamide 60 mg/kg x 2 days, followed by fludarabine 25 mg/m^2 x 5 days), preceding the autologous TIL infusion (LN-144), after which patients receive up to six doses of intravenous IL-2 (600,000 IU/kg).
- Patients will be evaluated for tumor response every 6 weeks following LN-144 infusion up till Month 6, after which, patients will be assessed every 12 weeks at 9, 12, 15, 18, 21, and 24 months.

STUDY DESIGN

Figure 2. Study Design

Now enrolling at sites in US and Europe

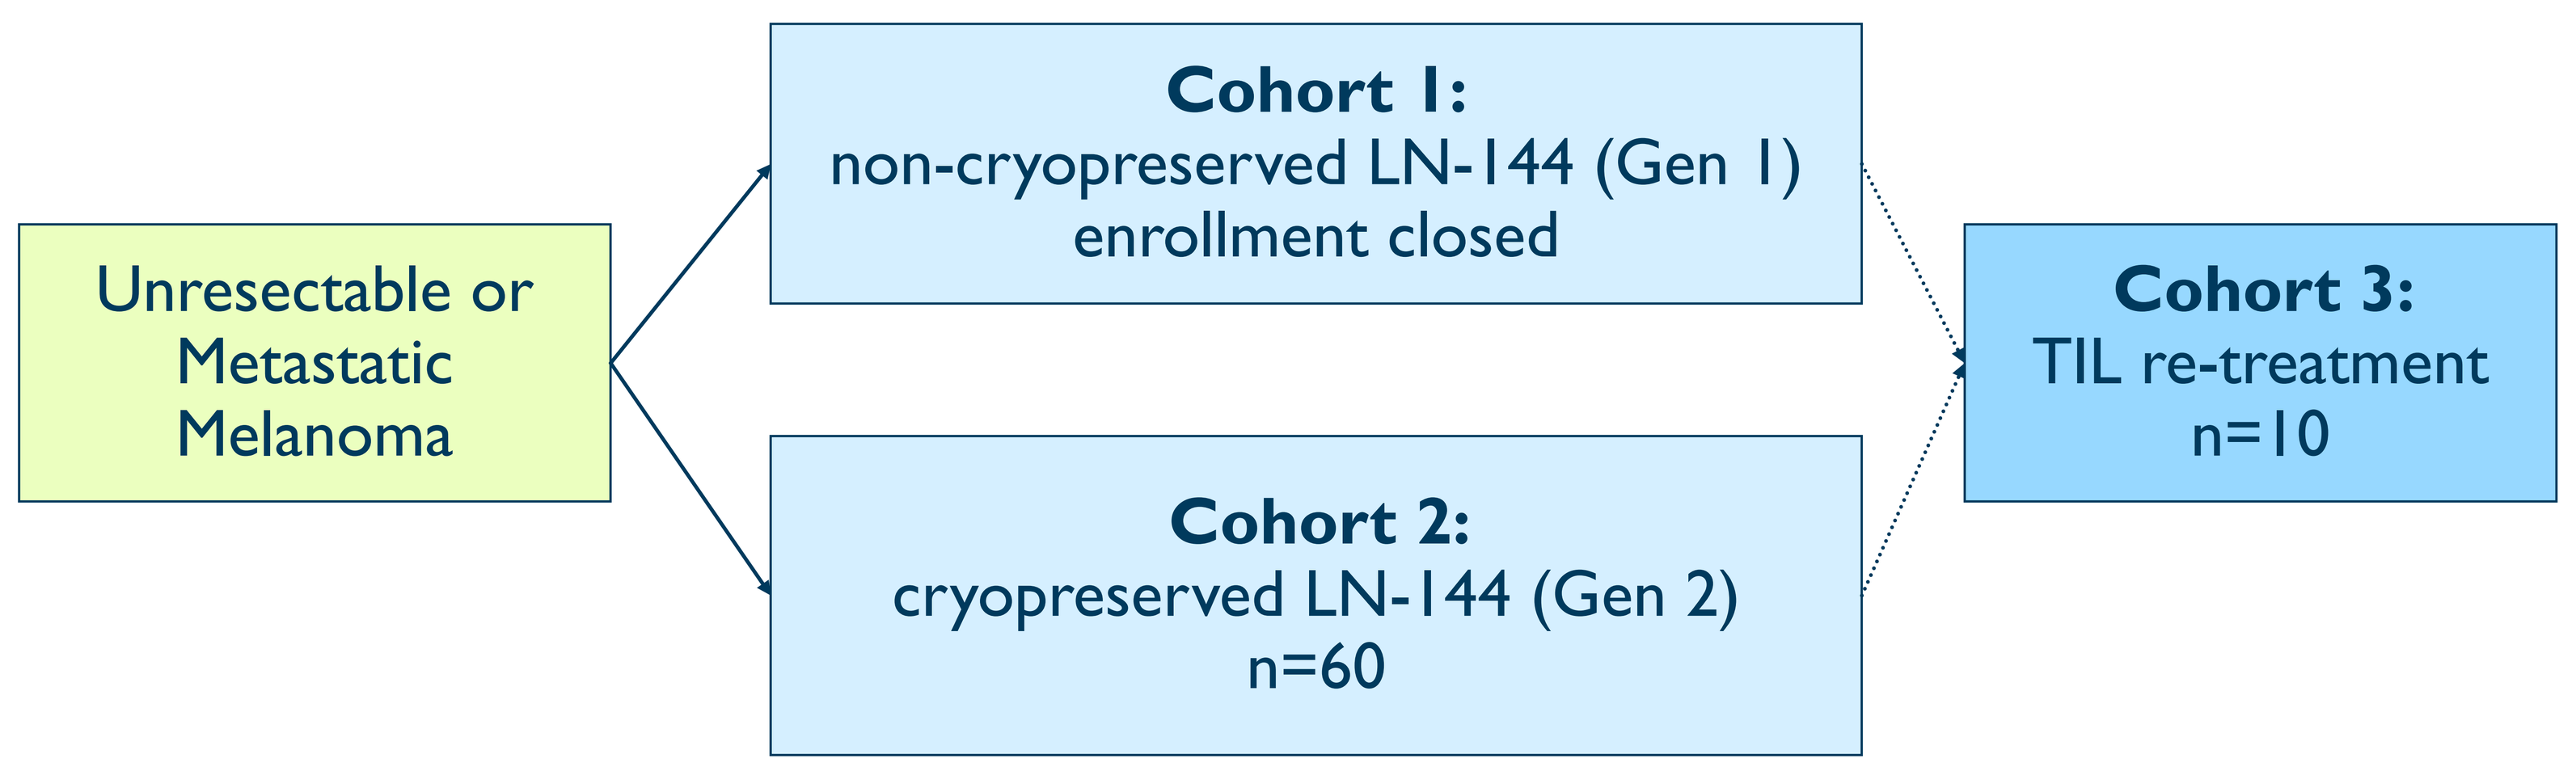
Unresectable or Metastatic Melanoma
Cohort 1: non-cryopreserved LN-144 (Gen 1) enrollment closed
Cohort 2: cryopreserved LN-144 (Gen 2) n=60
Cohort 3: TIL re-treatment n=10

STUDY FLOWCHART

Study Flowchart

Screening Period
≤ 28 days

Enrollment
Tumor harvest

Treatment Period
- NMA-LD (Day -7 to Day -1)
- TIL infusions (Day 0)
- IL-2 therapy (Day 0 to Day 4)

Response Assessment Period
- Every 6 weeks for the first 6 months
- Every 3 months thereafter

Overall Survival FU Period
Up to 5 years

Patients enrolled in Cohort 1 (Gen 1) or Cohort 2 (Gen 2)
Cohort 3: retreatment for patients who are eligible to receive 2nd treatment with TIL

Abbreviations: NMA-LD = nonmyeloablative lymphodepletion; TIL = tumor infiltrating lymphocytes; IL-2 = interleukin-2; OS = overall survival; FU = follow up; Gen 1 = generation 1; Gen 2 = generation 2.

OUTCOME MEASURES

Primary:
- Objective response rate (ORR)

Secondary:
- Duration of response (DOR), disease control rate (DCR), and progression-free survival (PFS)
- Overall survival (OS)
- Safety evaluation

Exploratory:
- Persistence of LN-144, immune correlates of response, outcome, and toxicity
- Efficacy per irRECIST[4]
- Health-related quality of life (HRQoL)

MAJOR INCLUSION & EXCLUSION CRITERIA

Major Inclusion Criteria
- Metastatic melanoma (Stage IIIC or Stage IV) with ≥1 prior line of systemic therapy, including progression on/after immune checkpoint inhibitor (eg, anti-PD-1), and BRAF inhibitor systemic therapy, if BRAF mutation-positive;
- One tumor lesion resectable for TIL generation (yielding tumor tissue ≥1.5 cm in diameter), and ≥ one tumor lesion for RECIST assessment as target;
- Age of 18 years to 70 years at the time of consent;
- ECOG performance status of 0 or 1, and estimated life expectancy of ≥3 months;
- Adequate bone marrow, liver, and renal function;
- Seronegative for the HIV Ab, hepatitis B Ag and hepatitis C Ab or Ag;
- Patients must have recovered from prior immune-mediated Grade ≥ 2 diarrhea or colitis.

Major Exclusion Criteria
- Uveal/ocular melanoma;
- Prior organ allograft or cell transfer therapy;
- Symptomatic and/or untreated brain metastases;
- Systemic steroid therapy >10 mg/day;
- Active systemic infections, coagulation disorders, or other active major medical illnesses of the cardiovascular, respiratory, or immune system;
- Primary or acquired immunodeficiency;
- History of hypersensitivity to any component of TIL therapy and other study drugs: cyclophosphamide, fludarabine, antibiotics of the aminoglycoside group (i.e., streptomycin, gentamicin), LN-144 or IL-2;
- No prior systemic BRAF-directed kinase inhibitor, if BRAF mutation positive (V600);
- LVEF < 45% or NYHA functional classification > Class 1 at Screening;
- FEV_1 ≤ 60%;
- Primary malignancy in the previous 3 years, requiring treatment in last year.

SUMMARY

- Relapsed and refractory Metastatic Melanoma presents high unmet medical need with low survival rates and limited durable treatment options.
- TIL have demonstrated antitumor efficacy including durable long-term responses in heavily pretreated patients irrespective of prior therapy, including checkpoint inhibitors.[4]
- Iovance's Gen 2 manufacturing of LN-144 takes 22 days, significantly shortening the duration of time a patient has to wait to receive their TIL therapy.
- Gen 2, cryopreserved LN-144 offers flexibility in the timing of dosing, and leads to a reduction of cost of manufacturing.

Disclosure
This study and poster are sponsored by Iovance Biotherapeutics, Inc.

References

1. World Health Organization: Skin Cancer; 2017 *http://www.who.int/uv/faq/skincancer/en/index1.html.*
2. Surveillance, Epidemiology, and End Results (SEER) Program: Melanoma. *https://seer.cancer.gov/statfacts/html/melan.html..*
3. Goff, SL, Dudley, ME, Citrin, DE, et al. Randomized, Prospective Evaluation Comparing Intensity of Lymphodepletion Before Adoptive Transfer of Tumor-Infiltrating Lymphocytes for Patients With Metastatic Melanoma. *J Clin Oncol.* 2016;34(20):2389-2397.
4. Bohnsack O, Ludajic K, Hoos A. Adaptation of the Immune-related Response Criteria: irRECIST. ESMO2014.

NCT02360579

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster

© 2018, Iovance Biotherapeutics