1488 Presented at the Society for Immunotherapy of Cancer (SITC) 39th Annual Meeting | November 6–10, 2024 | Houston, TX

A multicenter phase 2 trial of lifileucel plus pembrolizumab in patients with checkpoint inhibitor-naive metastatic NSCLC: updated results

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors

For more information, please contact Benjamin C Creelan: ben.creelan@moffitt.org

Benjamin C Creelan[1]; Kai He[2]; Edward Garon[3]; Jason Chesney[4]; Sylvia Lee[5]; Jorge Nieva[6]; Adrian Sacher[7]; Friedrich Graf Finckenstein[8]; Brian Gastman[8]; Jeffrey Chou[8]; Rana Fiaz[8]; Melissa Catlett[8]; Guang Chen[8]; Adam Schoenfeld[9]

[1]H Lee Moffitt Cancer Center and Research Institute, Tampa, FL, USA; [2]James Cancer Center, The Ohio State University, Columbus, OH, USA; [3]University of California Los Angeles, Los Angeles, CA, USA; [4]James Graham Brown Cancer Center, University of Louisville, Louisville, KY, USA; [5]Fred Hutchinson Cancer Center, Seattle, WA, USA; [6]University of Southern California, Los Angeles, CA, USA; [7]Princess Margaret Cancer Centre, Toronto, Ontario, Canada; [8]Iovance Biotherapeutics Inc, San Carlos, CA, USA; [9]Memorial Sloan Kettering Cancer Center, New York, NY, USA

Background

- Resistance to frontline immune checkpoint inhibitor (ICI) ± chemotherapy presents a challenge in the treatment of metastatic non-small cell lung cancer (NSCLC)[1]
- Tumor-infiltrating lymphocyte (TIL) cell therapy with lifileucel alone demonstrated an objective response rate (ORR) of 21% in patients with refractory metastatic NSCLC previously treated with an ICI[2]
- ORR for pembrolizumab monotherapy in NSCLC is dependent on programmed cell death-1 ligand (PD-L1) expression, with ORR of ~10% reported for PD-L1 tumor proportion score (TPS) <1% and up to 45% for PD-L1 TPS ≥50%[3]
- Integration of TIL cell therapy in frontline regimens may improve long-term outcomes in NSCLC
- The PD-L1–negative, endothelial growth factor receptor gene wild-type (*EGFR*-wt) subgroup in particular has a high unmet need, as ICI does not add benefit and targeted therapy is not available
- We report updated efficacy and safety results for lifileucel combined with pembrolizumab in patients with ICI-naive metastatic NSCLC

Methods

- IOV-COM-202 (NCT03645928) is a global, phase 2, multicenter, multicohort, open-label study of lifileucel in patients with solid tumors
- Cohort 3A enrolled patients with ICI-naive advanced or metastatic NSCLC with disease progression
 - Patients were required to have ≥1 resectable lesion for lifileucel manufacturing and ≥1 evaluable lesion by Response Evaluation Criteria in Solid Tumors (RECIST v1.1)
 - Patients were allowed to have received up to 3–4 prior lines of therapy excluding ICI
- Patients received nonmyeloablative lymphodepletion (cyclophosphamide and fludarabine), followed by a single lifileucel infusion, and ≤6 doses of high-dose interleukin-2 (IL-2) (**Figure 1**)
- Pembrolizumab was administered once before lymphodepletion and continued for up to 2 years or disease progression or toxicity after lifileucel infusion
- Primary endpoints were ORR by RECIST v1.1 and safety including the incidence of grade ≥3 treatment-emergent adverse events (TEAEs)

Figure 1. Treatment Schema

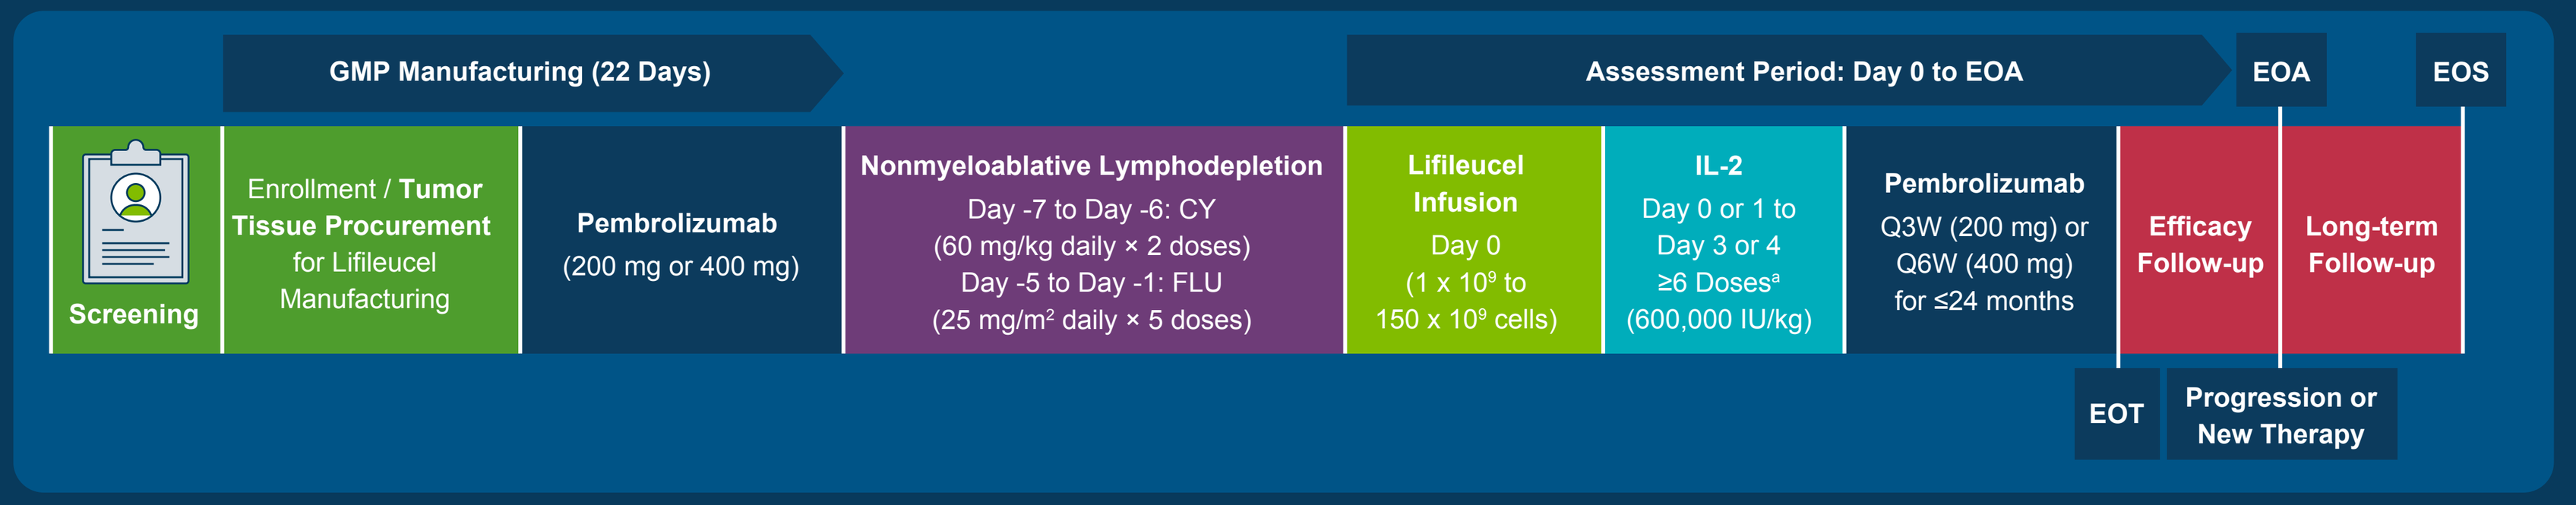
GMP Manufacturing (22 Days)
Assessment Period: Day 0 to EOA
EOA
EOS
Screening
Enrollment / Tumor Tissue Procurement for Lifileucel Manufacturing
Pembrolizumab (200 mg or 400 mg)
Nonmyeloablative Lymphodepletion
Day -7 to Day -6: CY (60 mg/kg daily × 2 doses)
Day -5 to Day -1: FLU (25 mg/m² daily × 5 doses)
Lifileucel Infusion
Day 0 (1 x 10⁹ to 150 x 10⁹ cells)
IL-2
Day 0 or 1 to Day 3 or 4 ≥6 Doses[a] (600,000 IU/kg)
Pembrolizumab
Q3W (200 mg) or Q6W (400 mg) for ≤24 months
Efficacy Follow-up
Long-term Follow-up
EOT
Progression or New Therapy

[a]Every 8–12 hours (starting 3–24 hours after completion of lifileucel infusion).

Results

- At the data cutoff date of August 12, 2024, 22 patients had received lifileucel infusion
- Patients had a median age of 57 years (range, 30–69) and had received a median of 1 (range, 0–4) line of prior systemic therapy (**Table 1**)
- Fourteen patients had *EGFR*-wt disease, of which 11 had a PD-L1 TPS of <1%
- The most common sites of tumor resection were lung (45.5%) and lymph node (22.7%)
- The median dose of lifileucel was 22.9 × 10^9 cells (range, 1 × 10^9 – 57.6 × 10^9)

Table 1. Demographic and Baseline Characteristics

Characteristic	*EGFR*-wt (n=14)	*EGFR* Mutated Post-TKI (n=8)	All (N=22)
Median age (range), years	57.0 (30–68)	55.5 (35–69)	57.0 (30–69)
Female, n (%)	6 (42.9)	6 (75.0)	12 (54.5)
Tobacco use, n (%)			
Former use	13 (92.9)	1 (12.5)	14 (63.6)
Never used	1 (7.1)	7 (87.5)	8 (36.4)
ECOG PS, n (%)			
0	6 (42.9)	6 (75.0)	12 (54.5)
1	8 (57.1)	2 (25.0)	10 (45.5)
Histologic cell type, n (%)			
Nonsquamous cell carcinoma	13 (92.9)	8 (100)	21 (95.5)
Squamous cell carcinoma	1 (7.1)	0	1 (4.5)
Resected tumor site, n (%)			
Bone	0	1 (12.5)	1 (4.5)
Liver	1 (7.1)	0	1 (4.5)
Lung	7 (50.0)	3 (37.5)	10 (45.5)
Lymph node	3 (21.4)	2 (25.0)	5 (22.7)
Other	2 (14.3)	2 (25.0)	4 (18.2)
Skin/Subcutaneous	1 (7.1)	0	1 (4.5)
Median target lesion SOD (range), mm	56.5 (13–218)	51.5 (16–103)	56.5 (13–218)
Median number of baseline target and nontarget lesions (range)	4 (2–10)	5 (2–10)	4 (2–10)
Number of baseline target and nontarget lesions, n (%)			
<3	5 (35.7)	3 (37.5)	8 (36.4)
≥3	9 (64.3)	5 (62.5)	14 (63.6)
PD-L1 tumor proportion score, n (%)			
<1	11 (78.6)	6 (75)	17 (77.3)
1–49	0	2 (25)	2 (9.1)
≥50	3 (21.4)	0	3 (13.6)
Median number of prior lines of systemic therapy (range)	1 (0–3)	2 (1–4)	1 (0–4)
Number of prior lines of systemic therapy, n (%)			
0	6 (42.9)	0	6 (27.3)
1	7 (50.0)	3 (37.5)	10 (45.5)
≥2	1 (7.1)	5 (62.5)	6 (27.3)
Prior lines of systemic therapy by agent, n (%)			
Chemotherapy	8 (57.1)	3 (37.5)	11 (50.0)
Monoclonal antibody	0	3 (37.5)	3 (13.6)
Targeted therapy	0	8 (100)	8 (36.4)

- At a median follow-up of 25.6 months, the ORR in the *EGFR*-wt subgroup was 64.3% (95% CI: 35.1–87.2) (**Table 2**)
 - Median duration of response was not reached in the EGFR-wt subgroup (95% CI: 3.7 months–NR)

Table 2. Response Outcomes by *EGFR* Mutation Status

Response Parameter	*EGFR*-wt (n=14)	*EGFR* Mutated Post-TKI (n=8)
ORR, n (%)	9 (64.3)	1 (12.5)
DCR, n (%)	11 (78.6)	7 (87.5)
BOR, n (%)		
CR	1 (7.1)	1 (12.5)
PR	8 (57.1)	0
SD	2 (14.3)	6 (75.0)
PD	2 (14.3)	0
NE	1 (7.1)	1 (12.5)
Median DOR (95% CI), months	NR (3.7, NR)	13.7 (NR–NR)

- One of the 8 patients with *EGFR* mutated post-TKI disease had a complete response

- Six responses occurred in 11 patients (54.5%) with *EGFR*-wt PD-L1–negative disease (**Figures 2** and **3**)

Figure 2. Best Percentage Change from Baseline in Target Lesion SOD in the *EGFR*-wt Subgroup

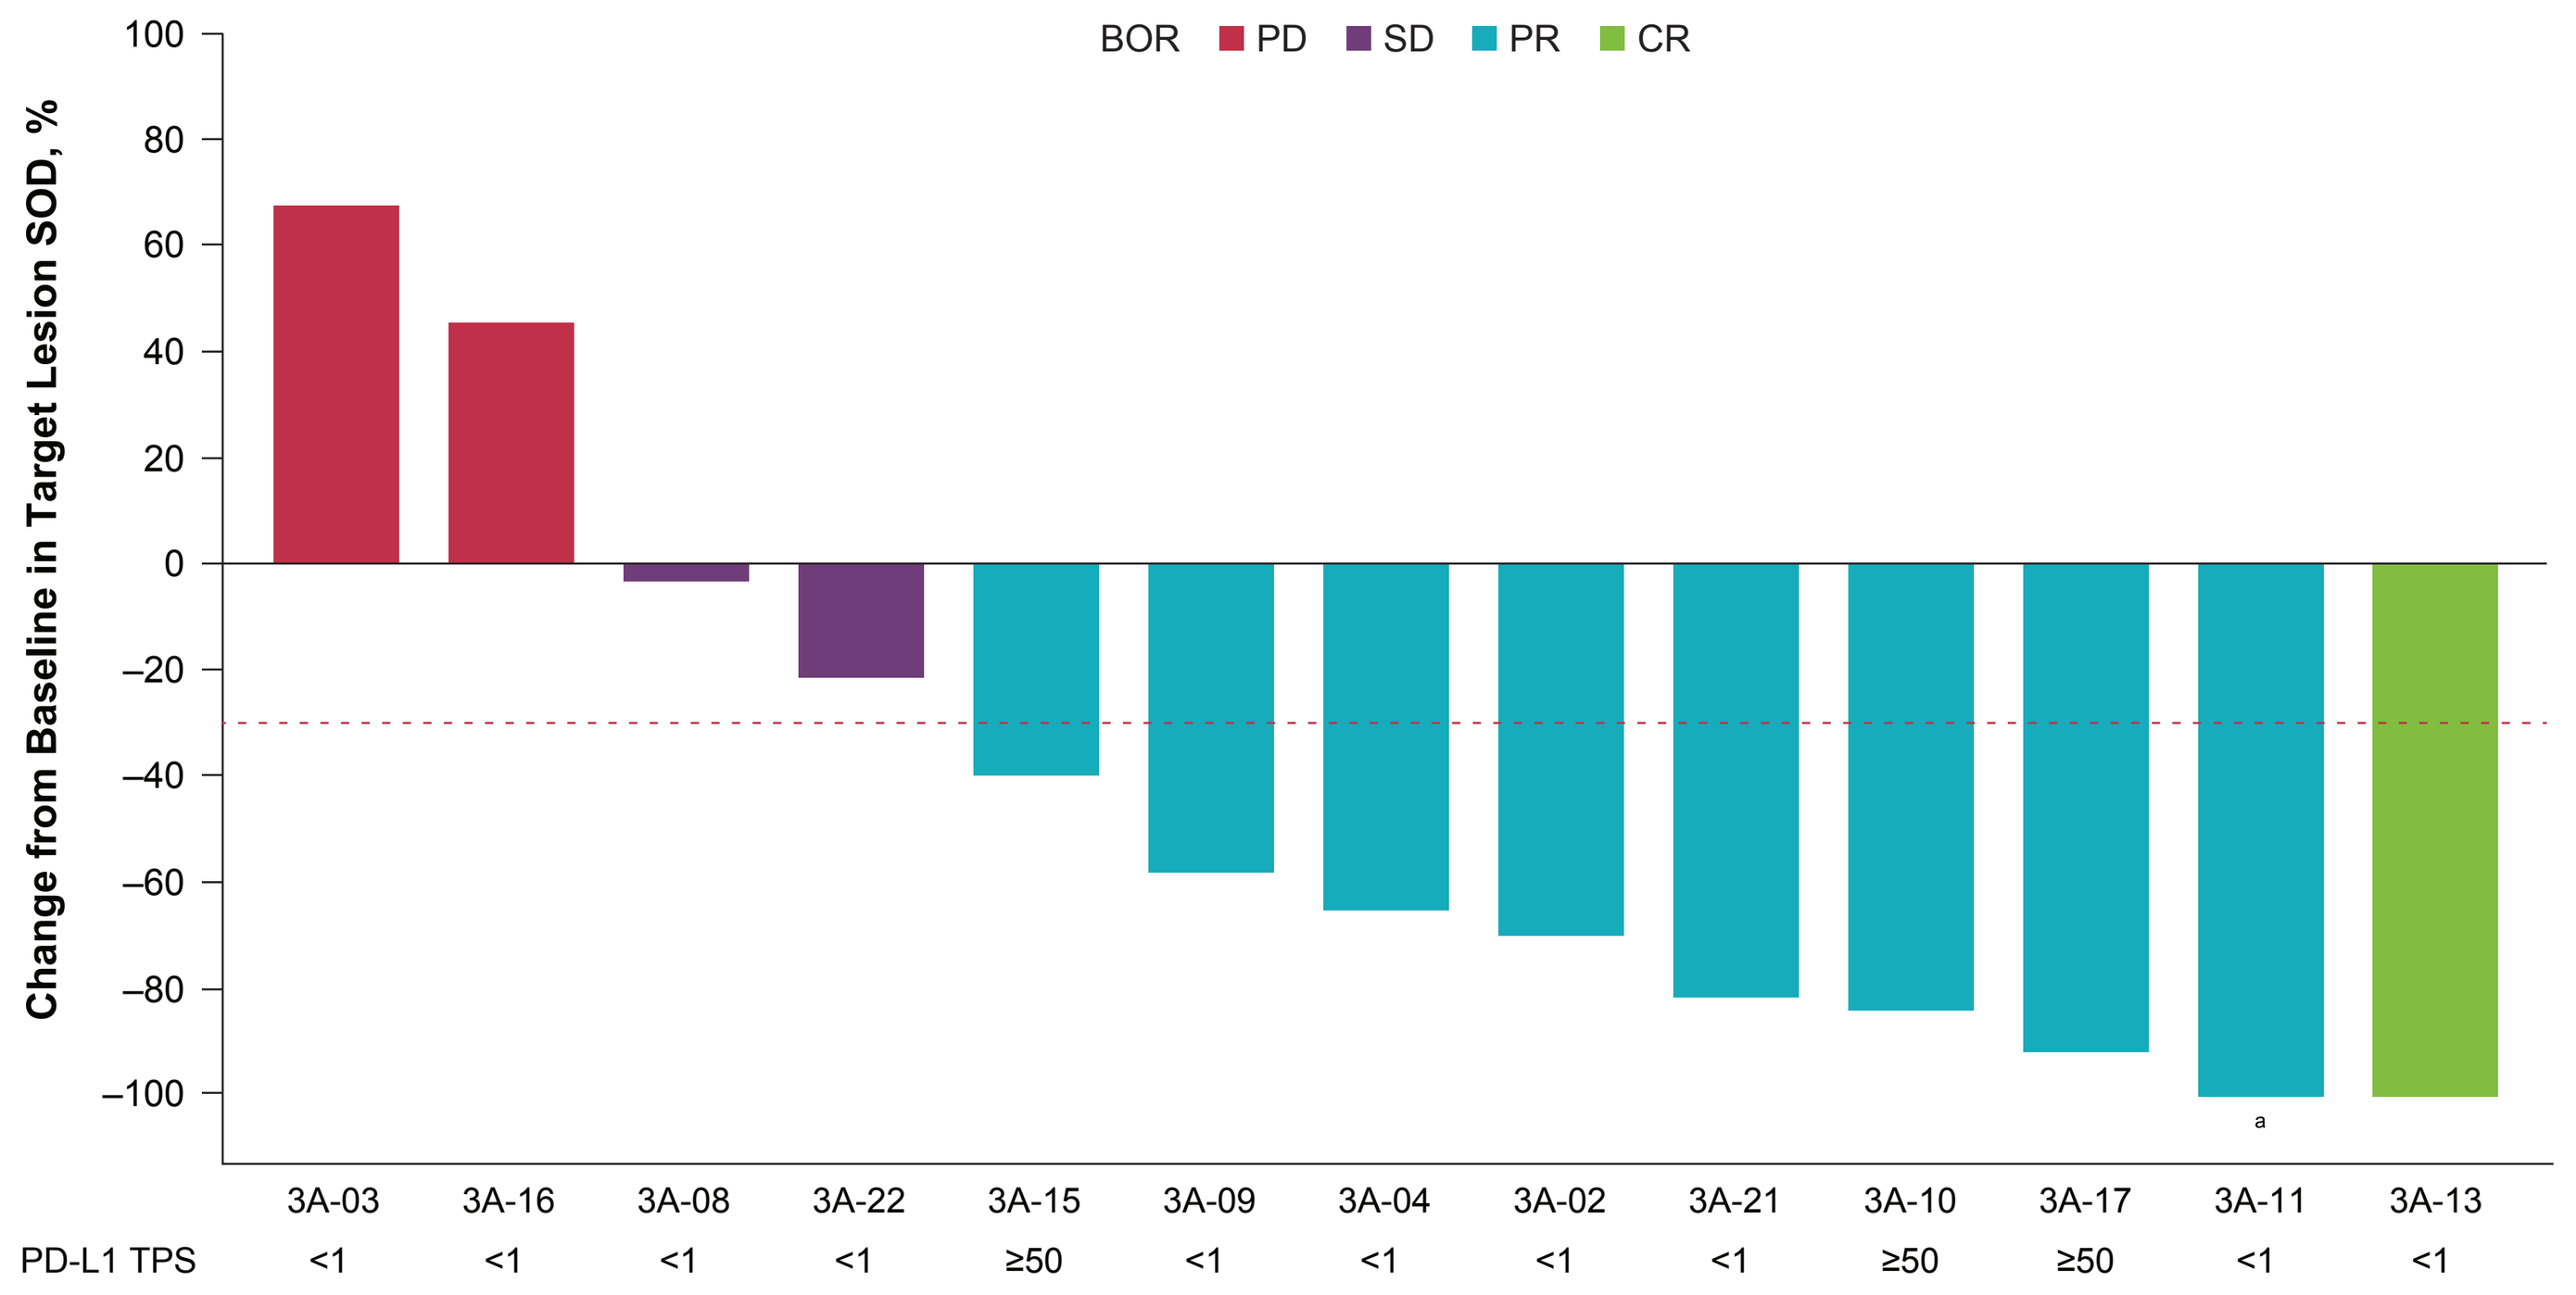
BOR PD SD PR CR
Change from Baseline in Target Lesion SOD, %
100
80
60
40
20
0
−20
−40
−60
−80
−100
3A-03 3A-16 3A-08 3A-22 3A-15 3A-09 3A-04 3A-02 3A-21 3A-10 3A-17 3A-11 3A-13
PD-L1 TPS <1 <1 <1 <1 ≥50 <1 <1 <1 <1 ≥50 ≥50 <1 <1
a

[a]A response of PR for this patient was based on a target lesion reduction of 100% with the persistence of nontarget lesions.
One patient in *EGFR*-wt PD-L1–negative group who did not have post-dose tumor response assessment was not included in the plot.

Figure 3. Percentage Change from Baseline in Target Lesion SOD Over Time in the *EGFR*-wt Subgroup

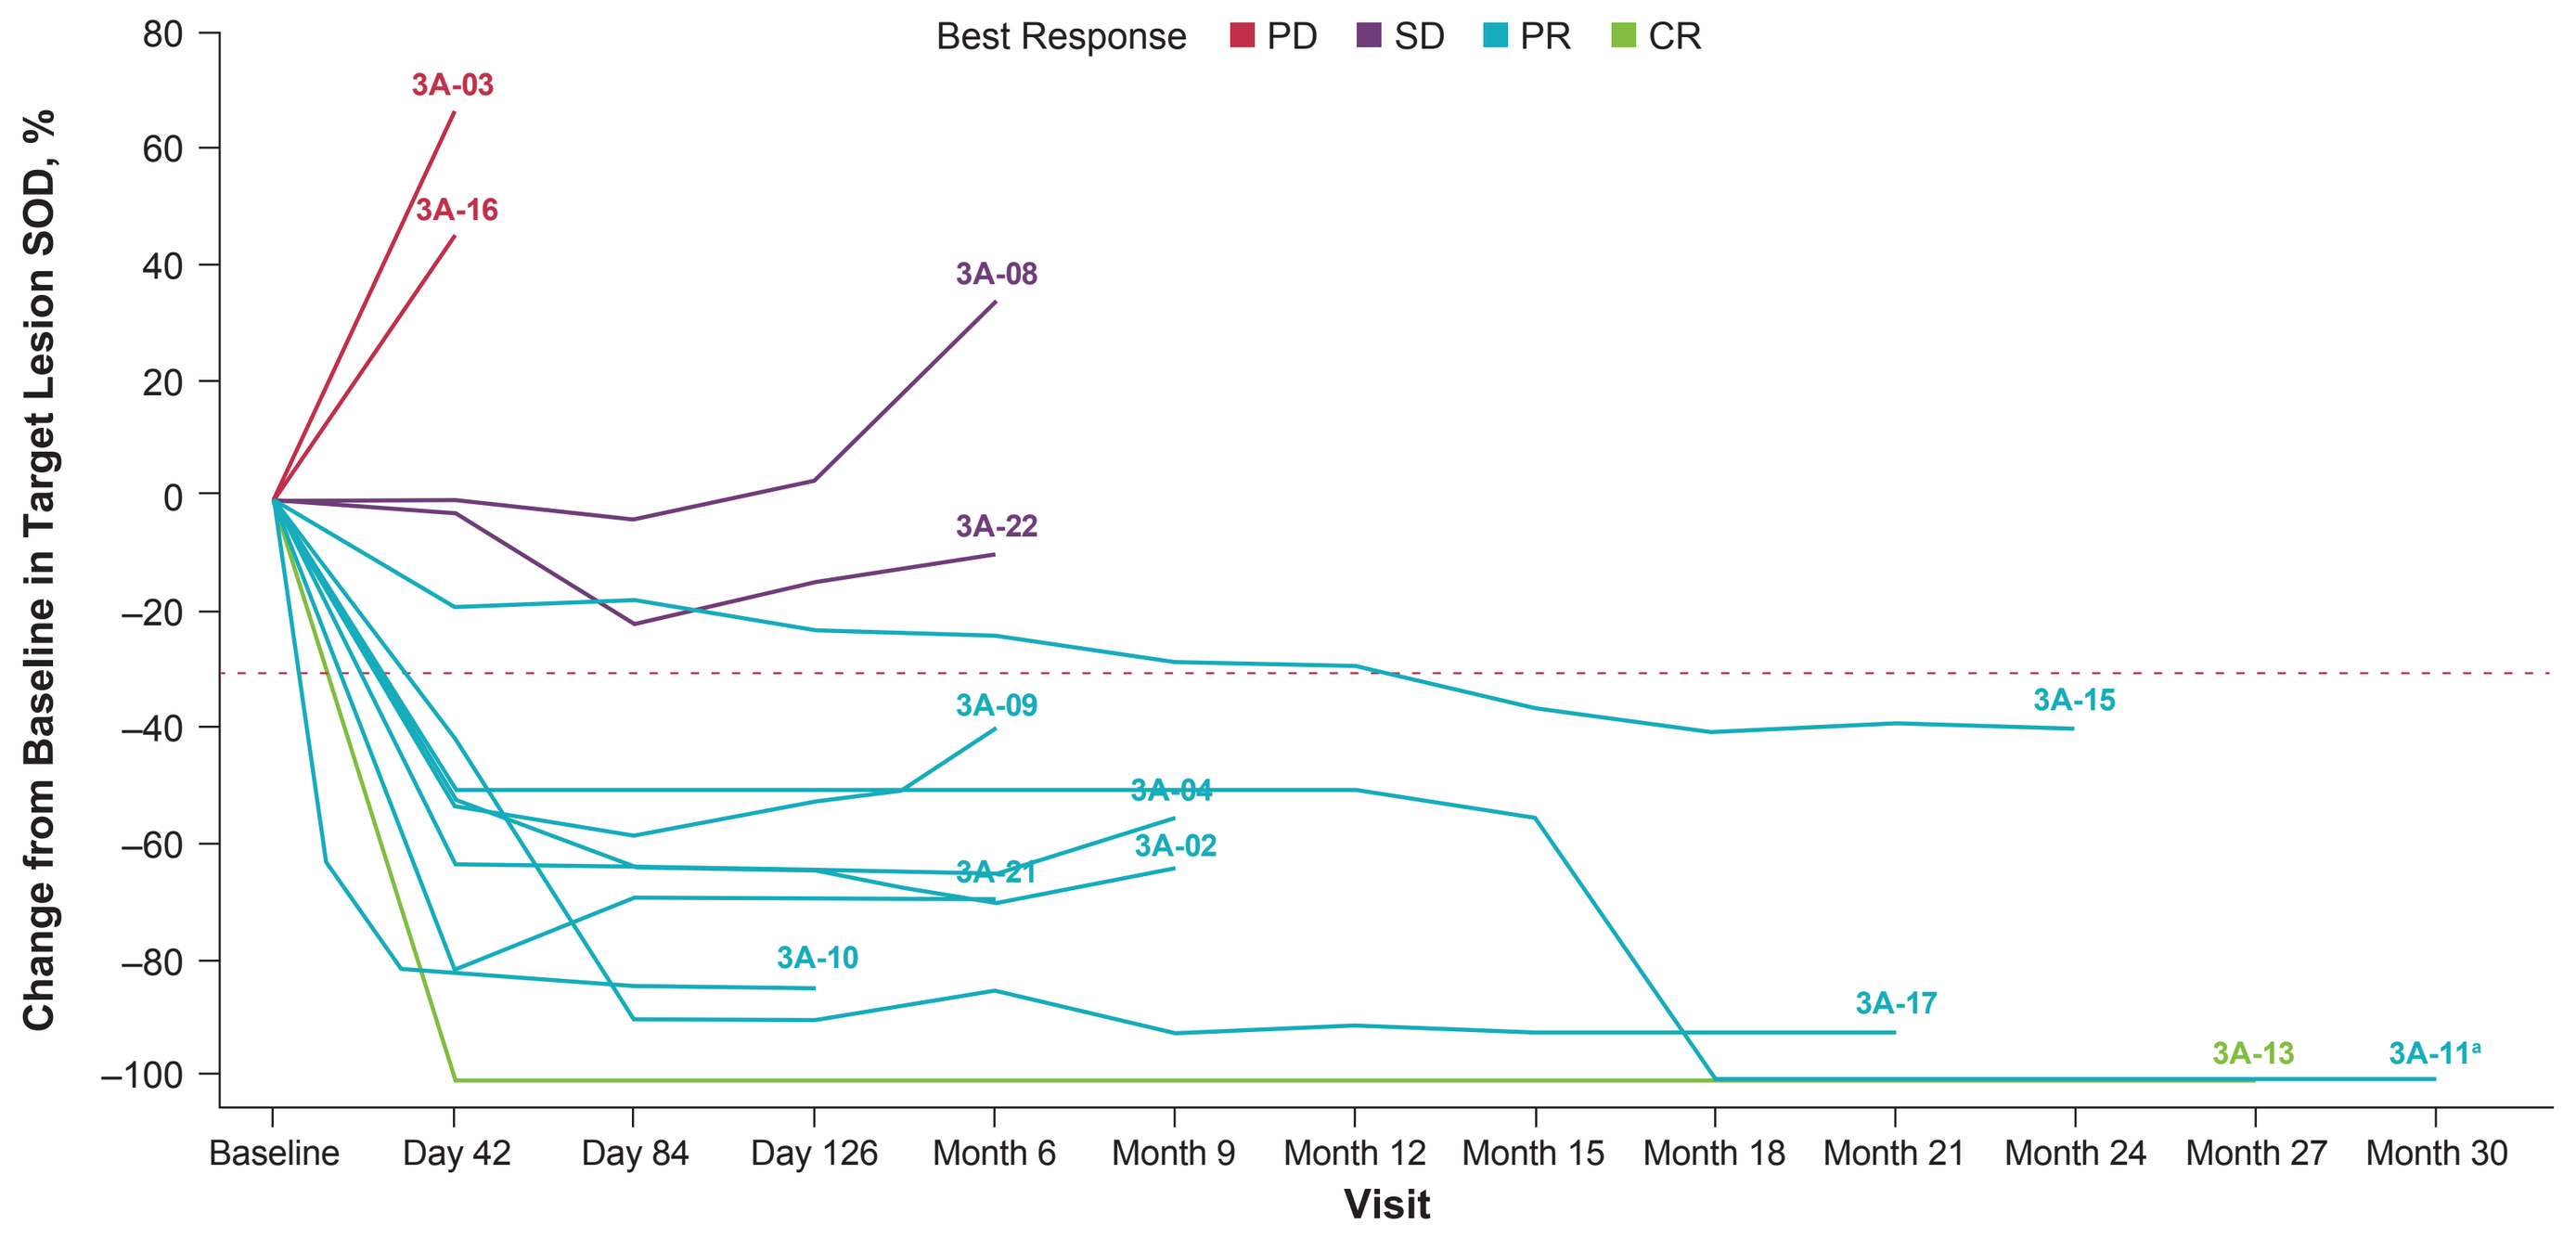
Best Response PD SD PR CR
Change from Baseline in Target Lesion SOD, %
80
60
40
20
0
−20
−40
−60
−80
−100
3A-03
3A-16
3A-08
3A-22
3A-09
3A-04
3A-21
3A-02
3A-10
3A-15
3A-17
3A-13
3A-11[a]
Baseline Day 42 Day 84 Day 126 Month 6 Month 9 Month 12 Month 15 Month 18 Month 21 Month 24 Month 27 Month 30
Visit

[a]A response of PR for this patient was based on a target lesion reduction of 100% with the persistence of nontarget lesions.

- Responses were durable, and 5 patients with *EGFR*-wt disease had ongoing responses at the last follow-up visit, which included 3 patients with PD-L1–negative disease (**Figure 4**)
 - 4 of these patients remained in response beyond 20 months from the start of TIL infusion
 - 2 of these responders had no prior treatment; 2 previously received chemotherapy
- The one complete responder with *EGFR* mutated post-TKI disease had disease recurrence after 15 months

Figure 4. Time on Study for Responders

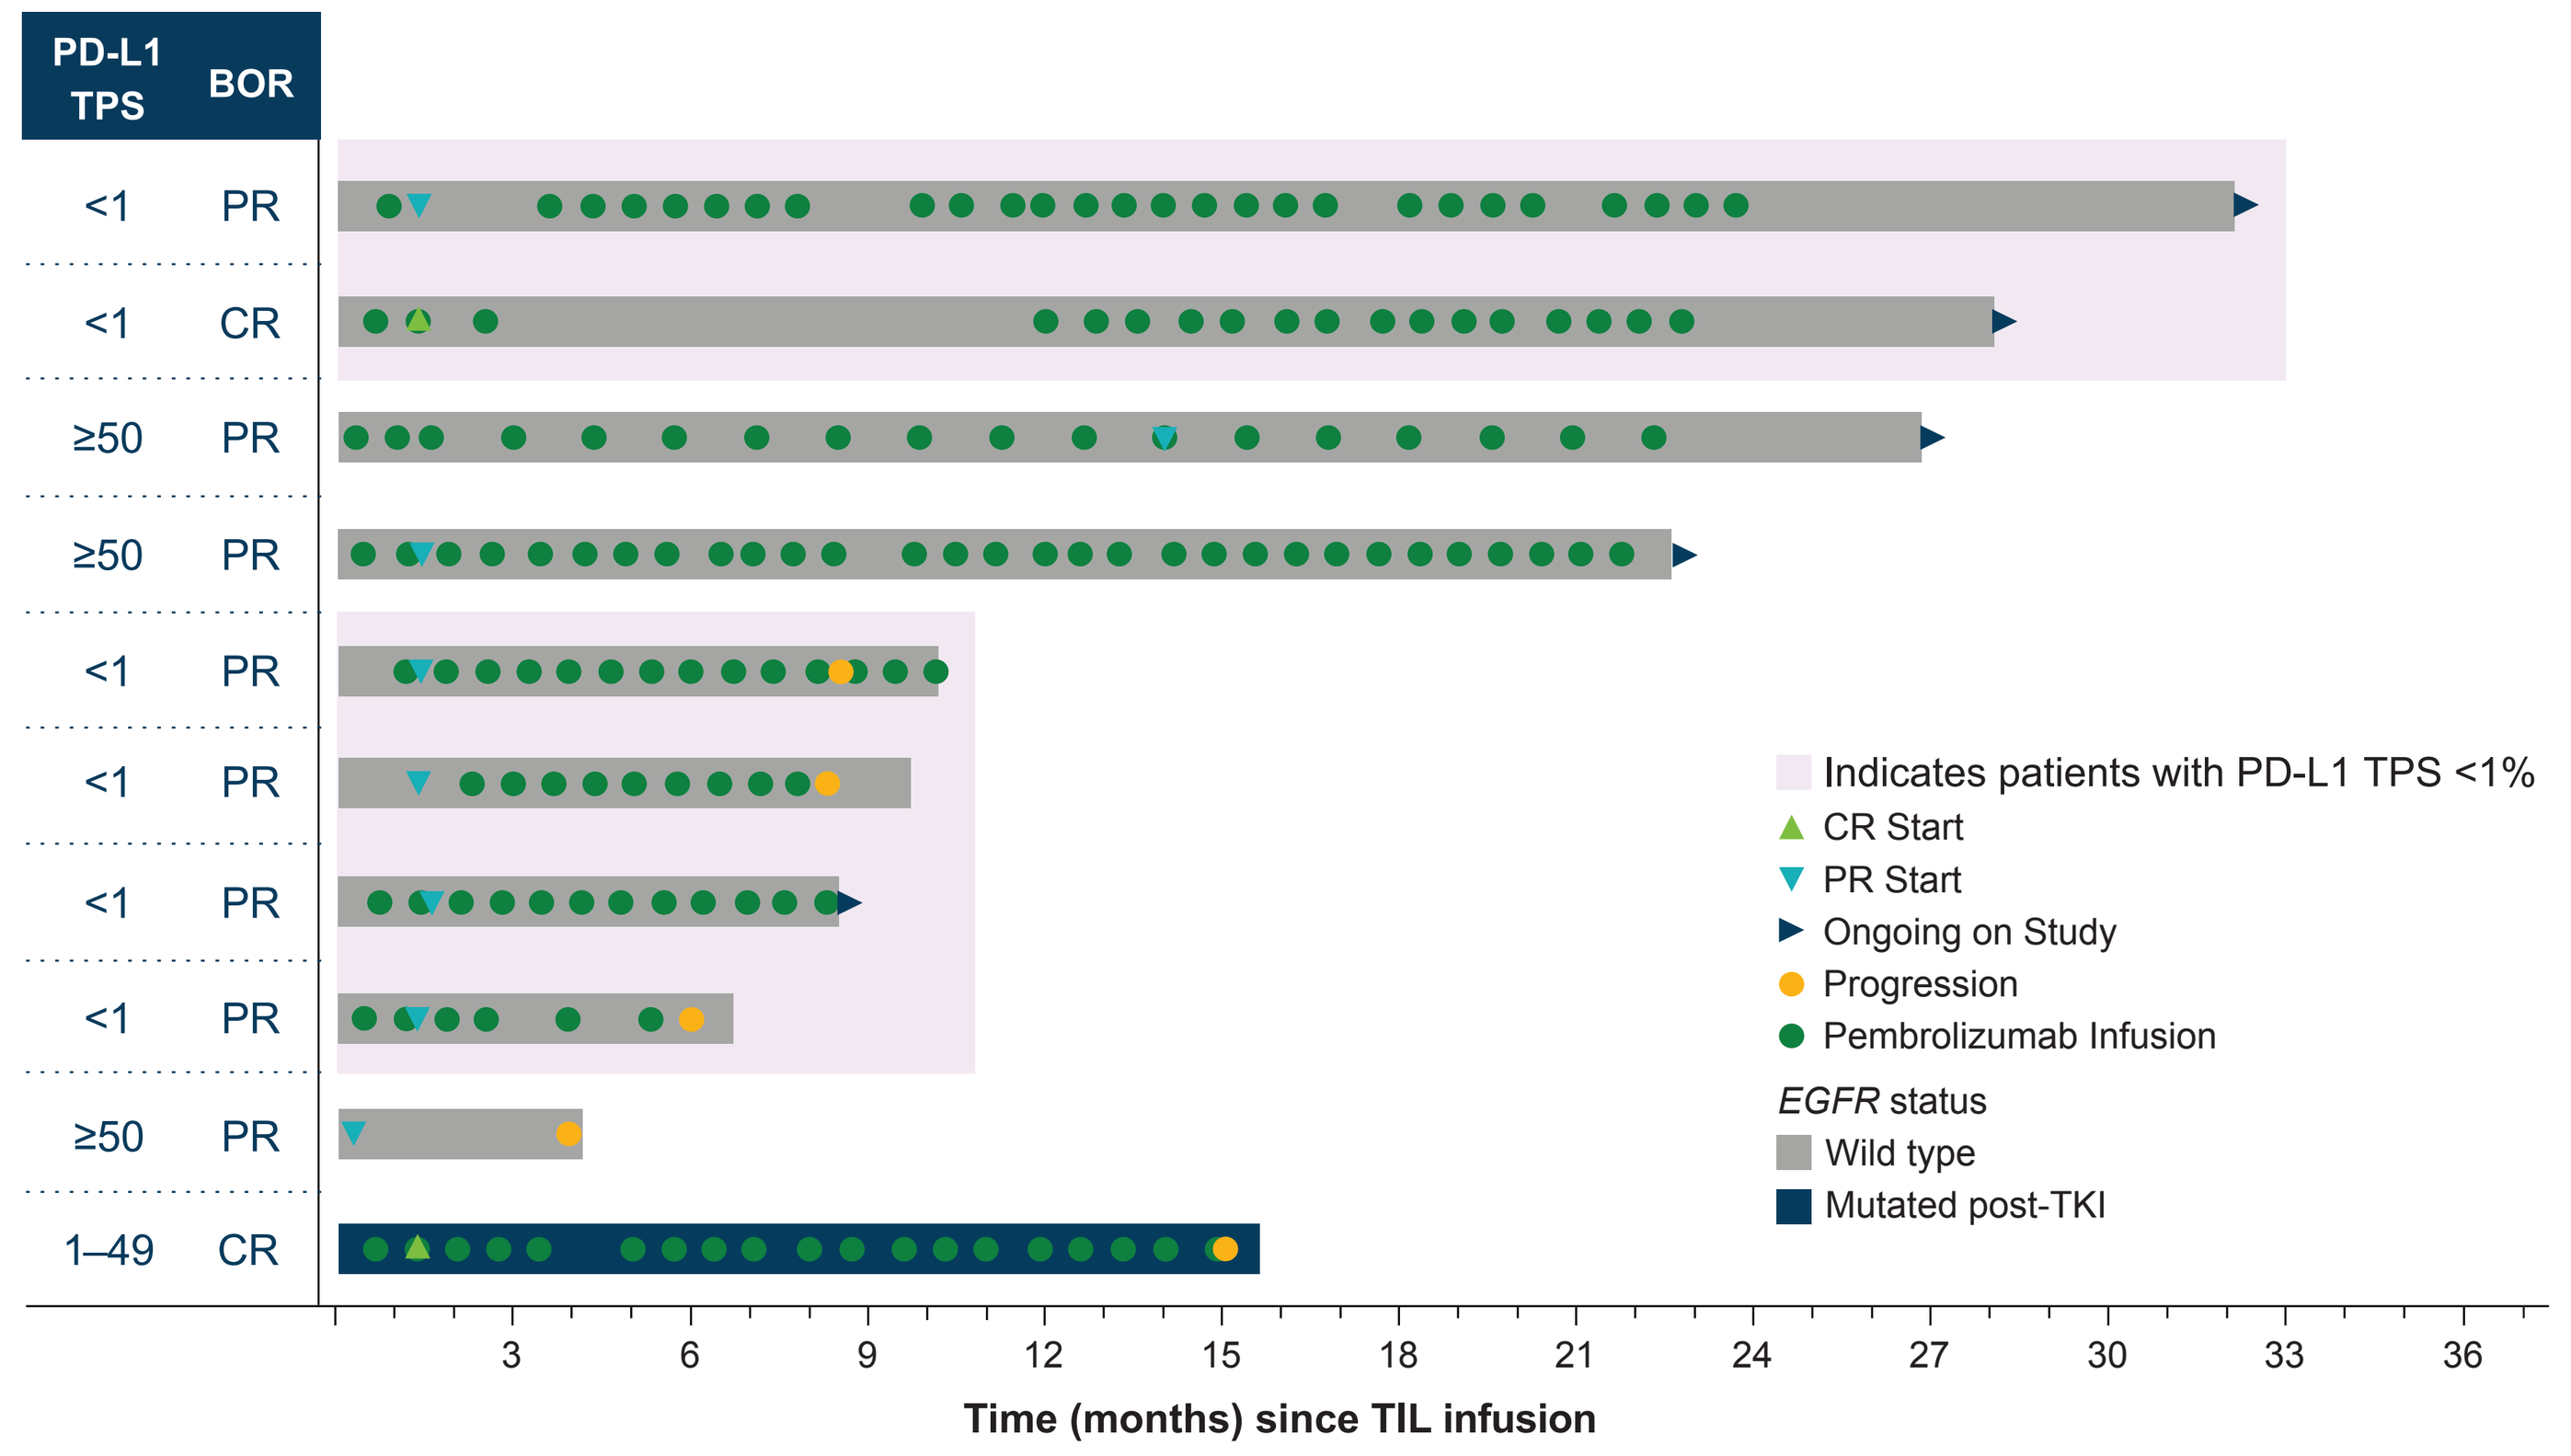
PD-L1 TPS
BOR
<1 PR
<1 CR
≥50 PR
≥50 PR
<1 PR
<1 PR
<1 PR
<1 PR
≥50 PR
1–49 CR
3 6 9 12 15 18 21 24 27 30 33 36
Time (months) since TIL infusion
Indicates patients with PD-L1 TPS <1%
CR Start
PR Start
Ongoing on Study
Progression
Pembrolizumab Infusion
EGFR status
Wild type
Mutated post-TKI

- TEAEs were consistent with underlying disease and known profiles of pembrolizumab, nonmyeloablative lymphodepletion, and IL-2
- The most common grade ≥3 nonhematologic TEAEs were hypoxia (54.5%), febrile neutropenia (45.5%), and hypophosphatemia (31.8%) (**Table 3**)

Table 3. Nonhematologic TEAEs in ≥30% of Patients[a]

TEAEs by Preferred Term, n (%)	Any Grade	Grade ≥3
Chills	17 (77.3)	0
Pyrexia	17 (77.3)	1 (4.5)
Hypoxia	17 (77.3)	12 (54.5)
Fatigue	14 (63.6)	3 (13.6)
Nausea	14 (63.6)	1 (4.5)
Dyspnea	13 (59.1)	5 (22.7)
Diarrhea	11 (50.0)	0
Hypotension	11 (50.0)	3 (13.6)
Sinus tachycardia	11 (50.0)	0
Febrile neutropenia	10 (45.5)	10 (45.5)
Arthralgia	9 (40.9)	2 (9.1)
Constipation	9 (40.9)	0
Decreased appetite	9 (40.9)	1 (4.5)
Hypoalbuminemia	9 (40.9)	1 (4.5)
Hyponatremia	9 (40.9)	3 (13.6)
Cough	9 (40.9)	0
Hypertension	9 (40.9)	3 (13.6)
Vomiting	8 (36.4)	0
Hypophosphatemia	8 (36.4)	7 (31.8)
Rash maculopapular	8 (36.4)	0
Musculoskeletal chest pain	7 (31.8)	0
Headache	7 (31.8)	0
Peripheral edema	7 (31.8)	1 (4.5)

[a]TEAEs refer to AEs regardless of causality that occur from the first dose of pembrolizumab or lifileucel infusion (whichever is earlier) and ≤30 days after the last dose of pembrolizumab or lifileucel (whichever is later) or up to start of new anticancer therapy.

- TEAEs peaked during the first 2 weeks after lifileucel infusion and rapidly declined thereafter

Figure 5. Incidence of TEAEs Over Time[a]

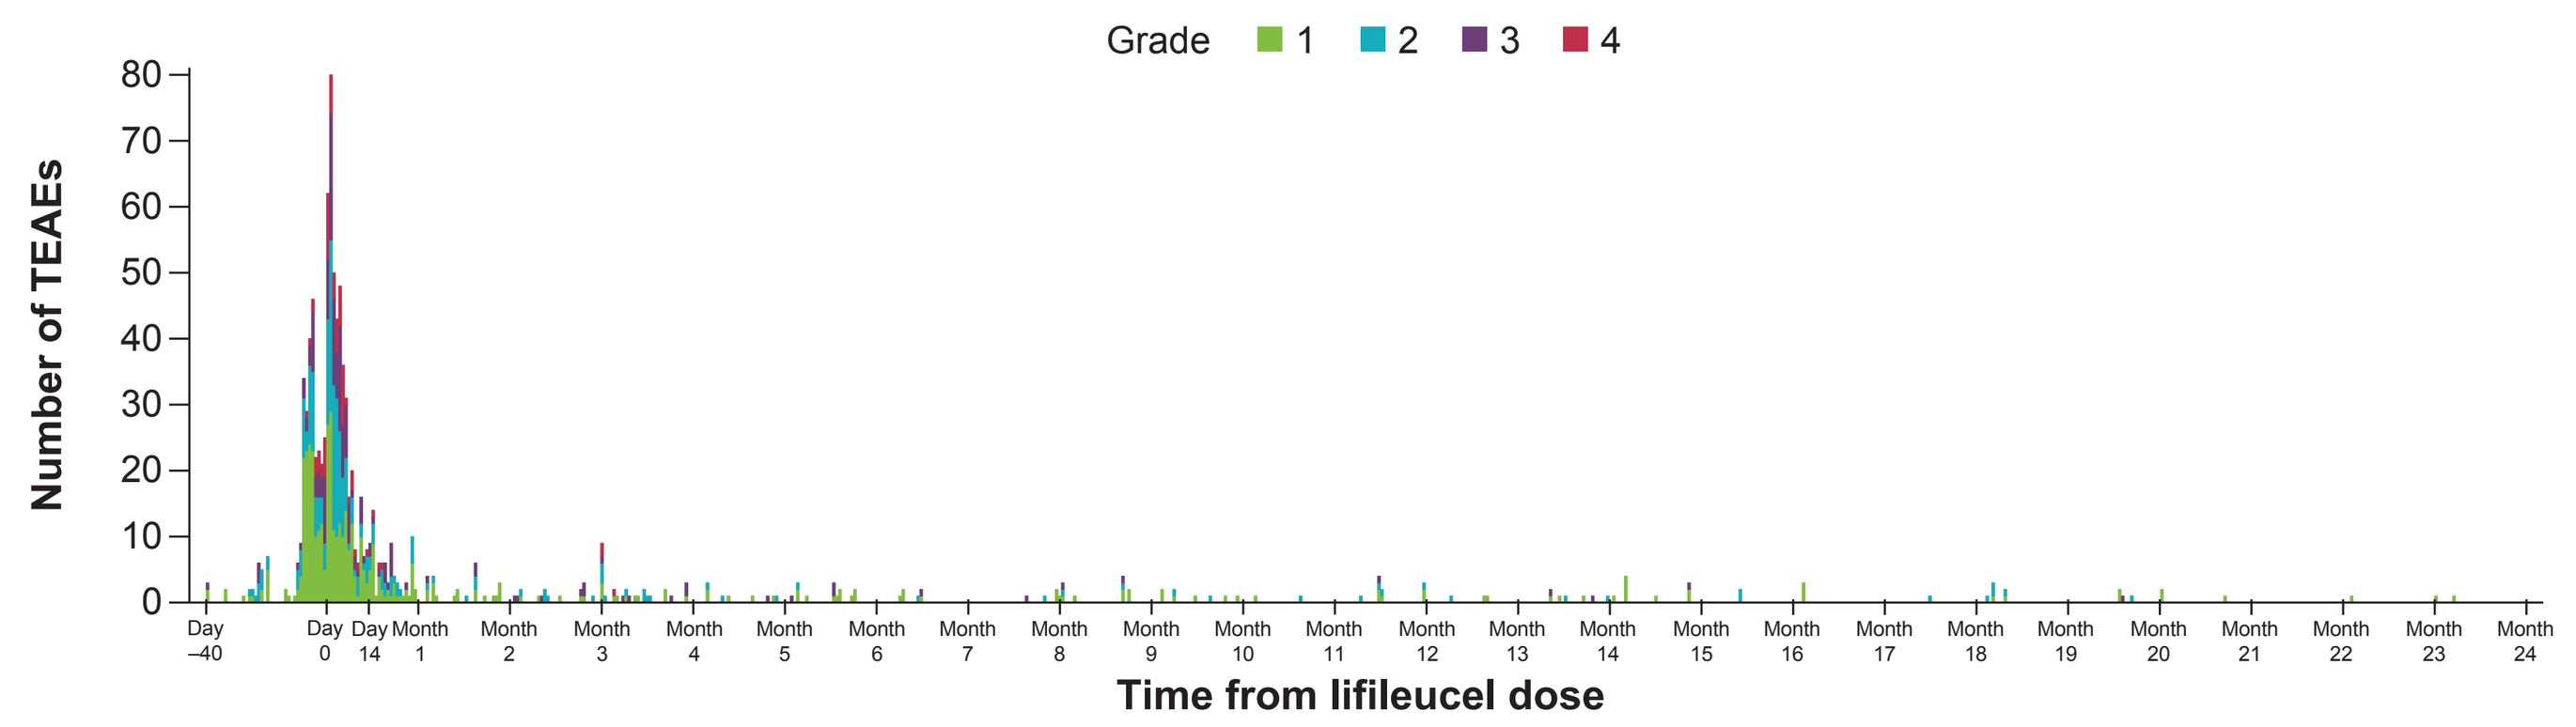
Grade 1 2 3 4
Number of TEAEs
80
70
60
50
40
30
20
10
0
Day −40
Day 0
Day 14
Month 1
Month 2
Month 3
Month 4
Month 5
Month 6
Month 7
Month 8
Month 9
Month 10
Month 11
Month 12
Month 13
Month 14
Month 15
Month 16
Month 17
Month 18
Month 19
Month 20
Month 21
Month 22
Month 23
Month 24
Time from lifileucel dose

[a]TEAEs refer to AEs regardless of causality that occur from the first dose of pembrolizumab or lifileucel infusion (whichever is earlier) and ≤30 days after the last dose of pembrolizumab or lifileucel (whichever is later) or up to start of new anticancer therapy. All occurrences are counted if a patient experiences the same TEAE, but the event is counted with the highest grade.

- All patients experienced grade 3/4 hematologic laboratory abnormalities (**Table 4**)
 - In the majority of patients, hematologic abnormalities resolved to grade ≤2 within the first 30 days after lifileucel treatment

Table 4. Grade 3/4 Hematologic Laboratory Abnormalities

Preferred Term	Events, n (%)	Events Resolved to Grade ≤2 by 30 Days After Lifileucel Infusion, n (%)
Neutropenia	22 (100)	21 (96)
Leukopenia	22 (100)	21 (96)
Lymphopenia	22 (100)	20 (91)
Thrombocytopenia	20 (91)	19 (95)
Anemia	15 (68)	15 (100)

Conclusions

- **In patients with ICI-naive metastatic *EGFR*-wt NSCLC, lifileucel plus pembrolizumab demonstrated robust antitumor activity and durable responses, including in patients with difficult to treat *EGFR*-wt PD-L1–negative tumors**
 - **ORR was 64.3% and responses included a CR and a deep PR; ORR was 54.5% in patients with *EGFR*-wt PD-L1–negative disease**
- **No new safety signals beyond what was expected with either lifileucel or pembrolizumab were observed**
- **These results support further investigation of lifileucel as part of frontline therapy in metastatic *EGFR*-wt NSCLC**

References

1. Zhou S, et al. *Front Immunol.* 2023;14:1129465. 2. Schoenfeld AJ, et al. *Cancer Discov.* 2024;14:1389–402. 3. Garon B, et al. *N Engl J Med.* 2015;37:2018–28.

Abbreviations

AE, adverse event; BOR, best overall response; CI, confidence interval; CR, complete response; CY, cyclophosphamide; DCR, disease control rate; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; EGFR, endothelial growth factor receptor; EOA, end of assessment; EOS, end of study; EOT, end of treatment; FLU, fludarabine; GMP, Good Manufacturing Practice; ICI, immune checkpoint inhibitor; IL-2, interleukin-2; NE, not evaluable; NR, not reached; NSCLC, non-small cell lung cancer; ORR, objective response rate; PD, progressive disease; PD-L1, programmed cell death-1 ligand; PR, partial response; PS, performance status; Q3W, every 3 weeks; Q6W, every 6 weeks; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease; SOD, sum of diameters; TEAE, treatment-emergent adverse event; TIL, tumor-infiltrating lymphocytes; TKI, tyrosine kinase inhibitor; TPS, tumor proportion score; wt, wild type.

Disclosures

Benjamin C Creelan: Achilles Therapeutics plc, Aptitude Health, AstraZeneca, Boehringer-Ingelheim, DAVA Oncology, ER Squibb & Sons, LLC, G1 Therapeutics, Inc, Hoffman LaRoche, Iovance Biotherapeutics, Jannsen, Johnson&Johnson, MJH Healthcare Holdings, LLC, OmniHealth Media, Regeneron, VJ HemeOnc; **Kai He:** AbbVie, Adaptimmune, Amgen, AstraZeneca, BioNTech SE, Bristol Myers Squibb, Genentech/Roche, GSK, Iovance Biotherapeutics, Lyell Immunopharma, Mirati Therapeutics, Obsidian Therapeutics, OncoC4, Perthera; **Edward Garon:** A2 Bio, AbbVie, ABL-Bio, Arcus, Arrivent, AstraZeneca, Atreca, Black Diamond Therapeutics, BridgeBio, Bristol Myers Squibb, Daiichi-Sankyo, Eli Lilly, EMD Serono, Genentech, Gilead, Hookipa, I-Mab, Iovance Biotherapeutics, iTeos, LianBio, Merck, Merus, Mirati, Novartis, Nuvalent, Pfizer, Prelude, Regeneron, Sanofi, Seagan, Sensei, Sumitomo, Strata, Summit, Synthekine, TILT Biotherapeutics; **Jason Chesney:** None to disclose; **Sylvia Lee:** Bristol Myers Squibb, Iovance Biotherapeutics, Kite Pharma, Lyell Immunopharma, Seagen; **Jorge Nieva:** Aadi Biosciences, Affyimmune, ANP Technologies, AstraZeneca, BioAtla, Cansera, Epic Sciences, G1 Therapeutics, Genentech, Indee Bio, Kalivir, Merck, MindMed, Naveris, Sanofi; **Adrian Sacher:** AdaptImmune, Amgen, AstraZeneca, BridgeBio, Bristol Myers Squibb, CRISPR Therapeutics, Genentech, Genentech-Roche, GSK, HotSpot Therapeutics, Iovance Biotherapeutics, Lilly, Merck, Pfizer, Spectrum; **Friedrich Graf Finckenstein:** Patents, Royalties, Other Intellectual Properties: Bristol Myers Squibb; Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics; **Brian Gastman, Jeffrey Chou, Rana Fiaz, Melissa Catlett, Guan Chen:** Iovance Biotherapeutics; **Adam Schoenfeld:** Achilles Therapeutics, Affini-T Therapeutics, Amgen, AstraZeneca, Bristol Myers Squibb, cTRL therapeutics, Enara Bio, GSK, Harpoon Therapeutics, Heat Biologics, Immunocore, Iovance Biotherapeutics, Johnson&Johnson, KSQ Therapeutics, Legend Biotech, Legend Therapeutics, Lyell Immunopharma, Merck, Obsidian Therapeutics, Oppenheimer and Co, PACT Pharma, Perceptive Advisors, Prelude Therapeutics, Regeneron, Synthekine, Umoja Biopharma.

Acknowledgments

- This study is sponsored by Iovance Biotherapeutics, Inc. (San Carlos, CA, USA)
- Medical writing and editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Iovance
- Special thank you to the IOV-COM-202 study patients and their families

© 2024, Iovance Biotherapeutics