1032P European Society for Medical Oncology (ESMO) | 13–17 September 2024 | Barcelona, Spain

Tumor-infiltrating lymphocytes with inducible membrane-tethered IL-12 cultured in optimized medium exhibit superior antitumor activity

Patrick Innamarato,[1] Nathan J. Gilbert,[1] Alvaro de Mingo Pulido,[1] Mohammed Alkhouli,[1] Judy Fang,[1] Gerard Sapena,[1] Jamie L. Blauvelt,[2] Shari Pilon-Thomas,[2] Sean Hall,[1] Hequn Yin,[1] Yongliang Zhang[1]

[1]Iovance Biotherapeutics, Inc., San Carlos, CA, USA; [2]Moffitt Cancer Center, Tampa, FL, USA

For more information, please contact
Hequn Yin: hequn.yin@iovance.com
Yongliang Zhang: yongliang.zhang@iovance.com

Introduction

- Lifileucel is a tumor-infiltrating lymphocyte (TIL) cell therapy approved in the United States for the treatment of unresectable or metastatic melanoma previously treated with a PD-1–blocking antibody and, if *BRAF* V600 mutation positive, a *BRAF/MEK* inhibitor.[1] In addition to lifileucel, we are diligently working on developing new assets to enhance the efficacy of the TIL product[2,3]
- Interleukin-12 (IL-12)–producing TIL has shown clinical benefit; however, circulating IL-12–related AEs limited further development[4]
- The current study aimed to increase TIL efficacy by using localized expression of tethered IL-12 and more of the following approaches:
 1. Enhancing TIL potency with inducible membrane-tethered nuclear factor of activated T cells (NFAT)–IL-12 (TeIL-12)
 2. Improving persistence of TIL with constitutive expression of an additional membrane-tethered cytokine
 3. Increasing yield and function of TIL during the expansion process with cell culture optimization (CCO)

Methods

TIL manufacturing

- Tumor tissues resected from various types of solid tumors, including non-small cell lung cancer, melanoma, and endometrial cancer, were fragmented and cultured for 11 days (pre-rapid expansion process [REP])
- Harvested pre-REP T cells were then transduced with a lentivirus vector containing TeIL-12 alone or TeIL-12 bicistronically combined with an additional cytokine (**Figure 1**)
- Cytokine-transduced TIL subsequently underwent an 11-day REP using an irradiated peripheral blood mononuclear cell feeder cell, OKT3, and IL-2 manufacturing process. In some experiments, we used a CCO growth medium to enhance TIL expansion and function

Figure 1. Bicistronic TeIL-12–cytokine construct

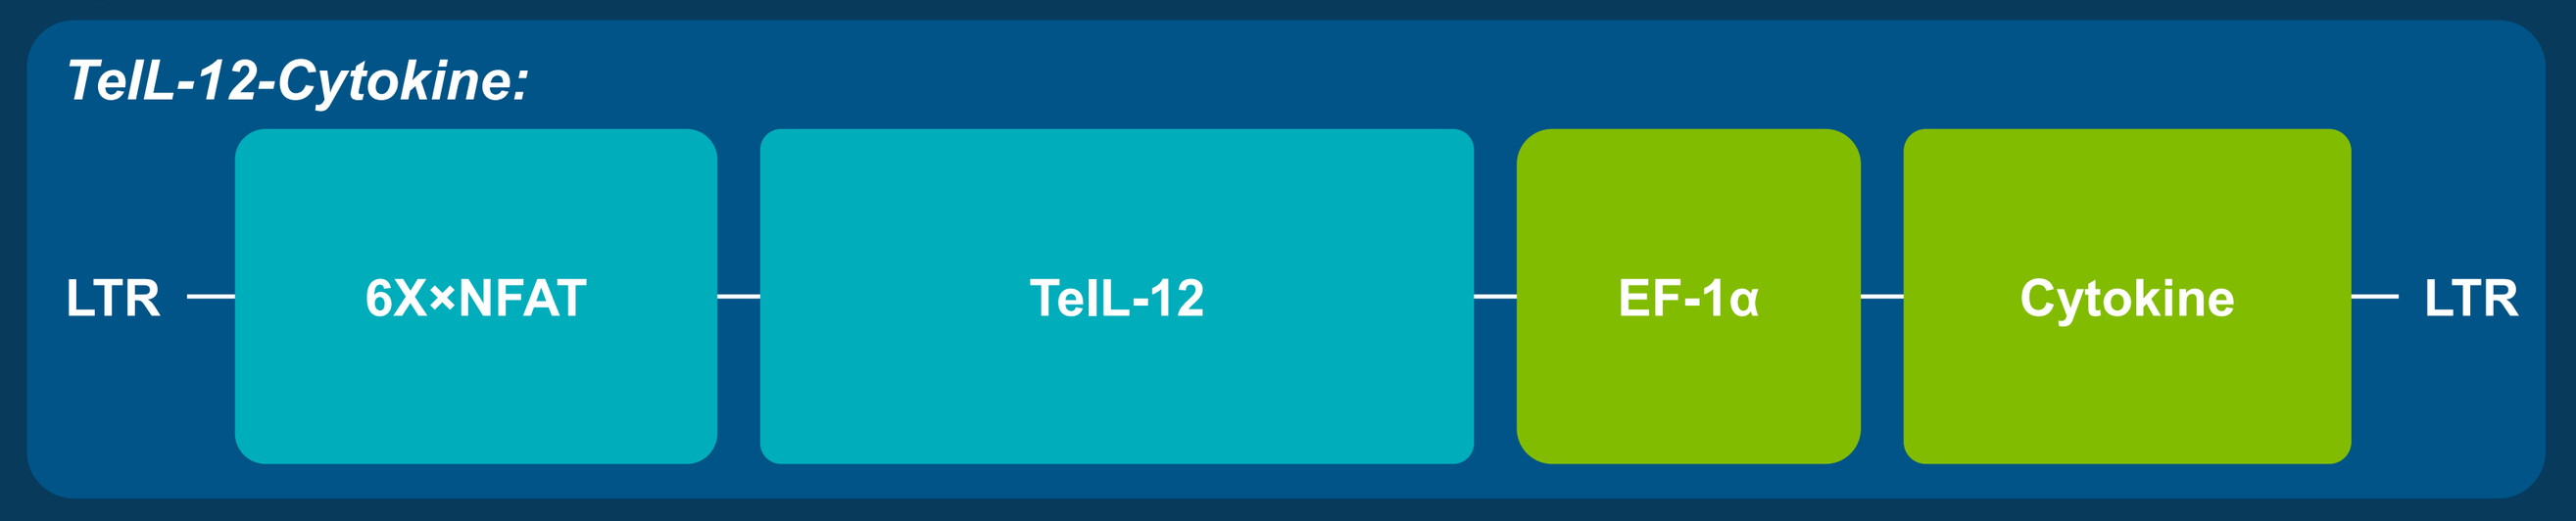
TeIL-12-Cytokine:
LTR
6X×NFAT
TeIL-12
EF-1α
Cytokine
LTR

Bicistronic construct of lentiviral vector. Inducible TeIL-12 is under 6 NFAT-driven promoters, and constitutive cytokine expression is under the EF-1α promoter.

EF-1α, elongation factor 1α; LTR, long-terminal repeat; NFAT, nuclear factor of activated T cells; TeIL-12, membrane-tethered NFAT–interleukin-12.

TIL characterization

- The impact of a single modification with TeIL-12 on efficacy was assessed *in vivo* with a patient-derived xenograft adoptive cell transfer murine model
- The impact of dual modification with dual-cytokine–engineered TIL was assessed by *in vitro* IL-2–dependent survival, autologous tumor reactivity, and antitumor reactivity
- The impact of CCO medium on dual-cytokine–engineered TIL was assessed further for yield, viability, and autologous tumor reactivity

Results

- Compared with unmodified TIL, single modification with expression of TeIL-12 alone conferred superior antitumor activity *in vivo* without detectable levels of IL-12 in circulation (**Figures 2 and 3**)

Figure 2. Single modification with TeIL-12 transduction enhanced TIL antitumor efficacy in an autologous *in vivo* melanoma PDX model

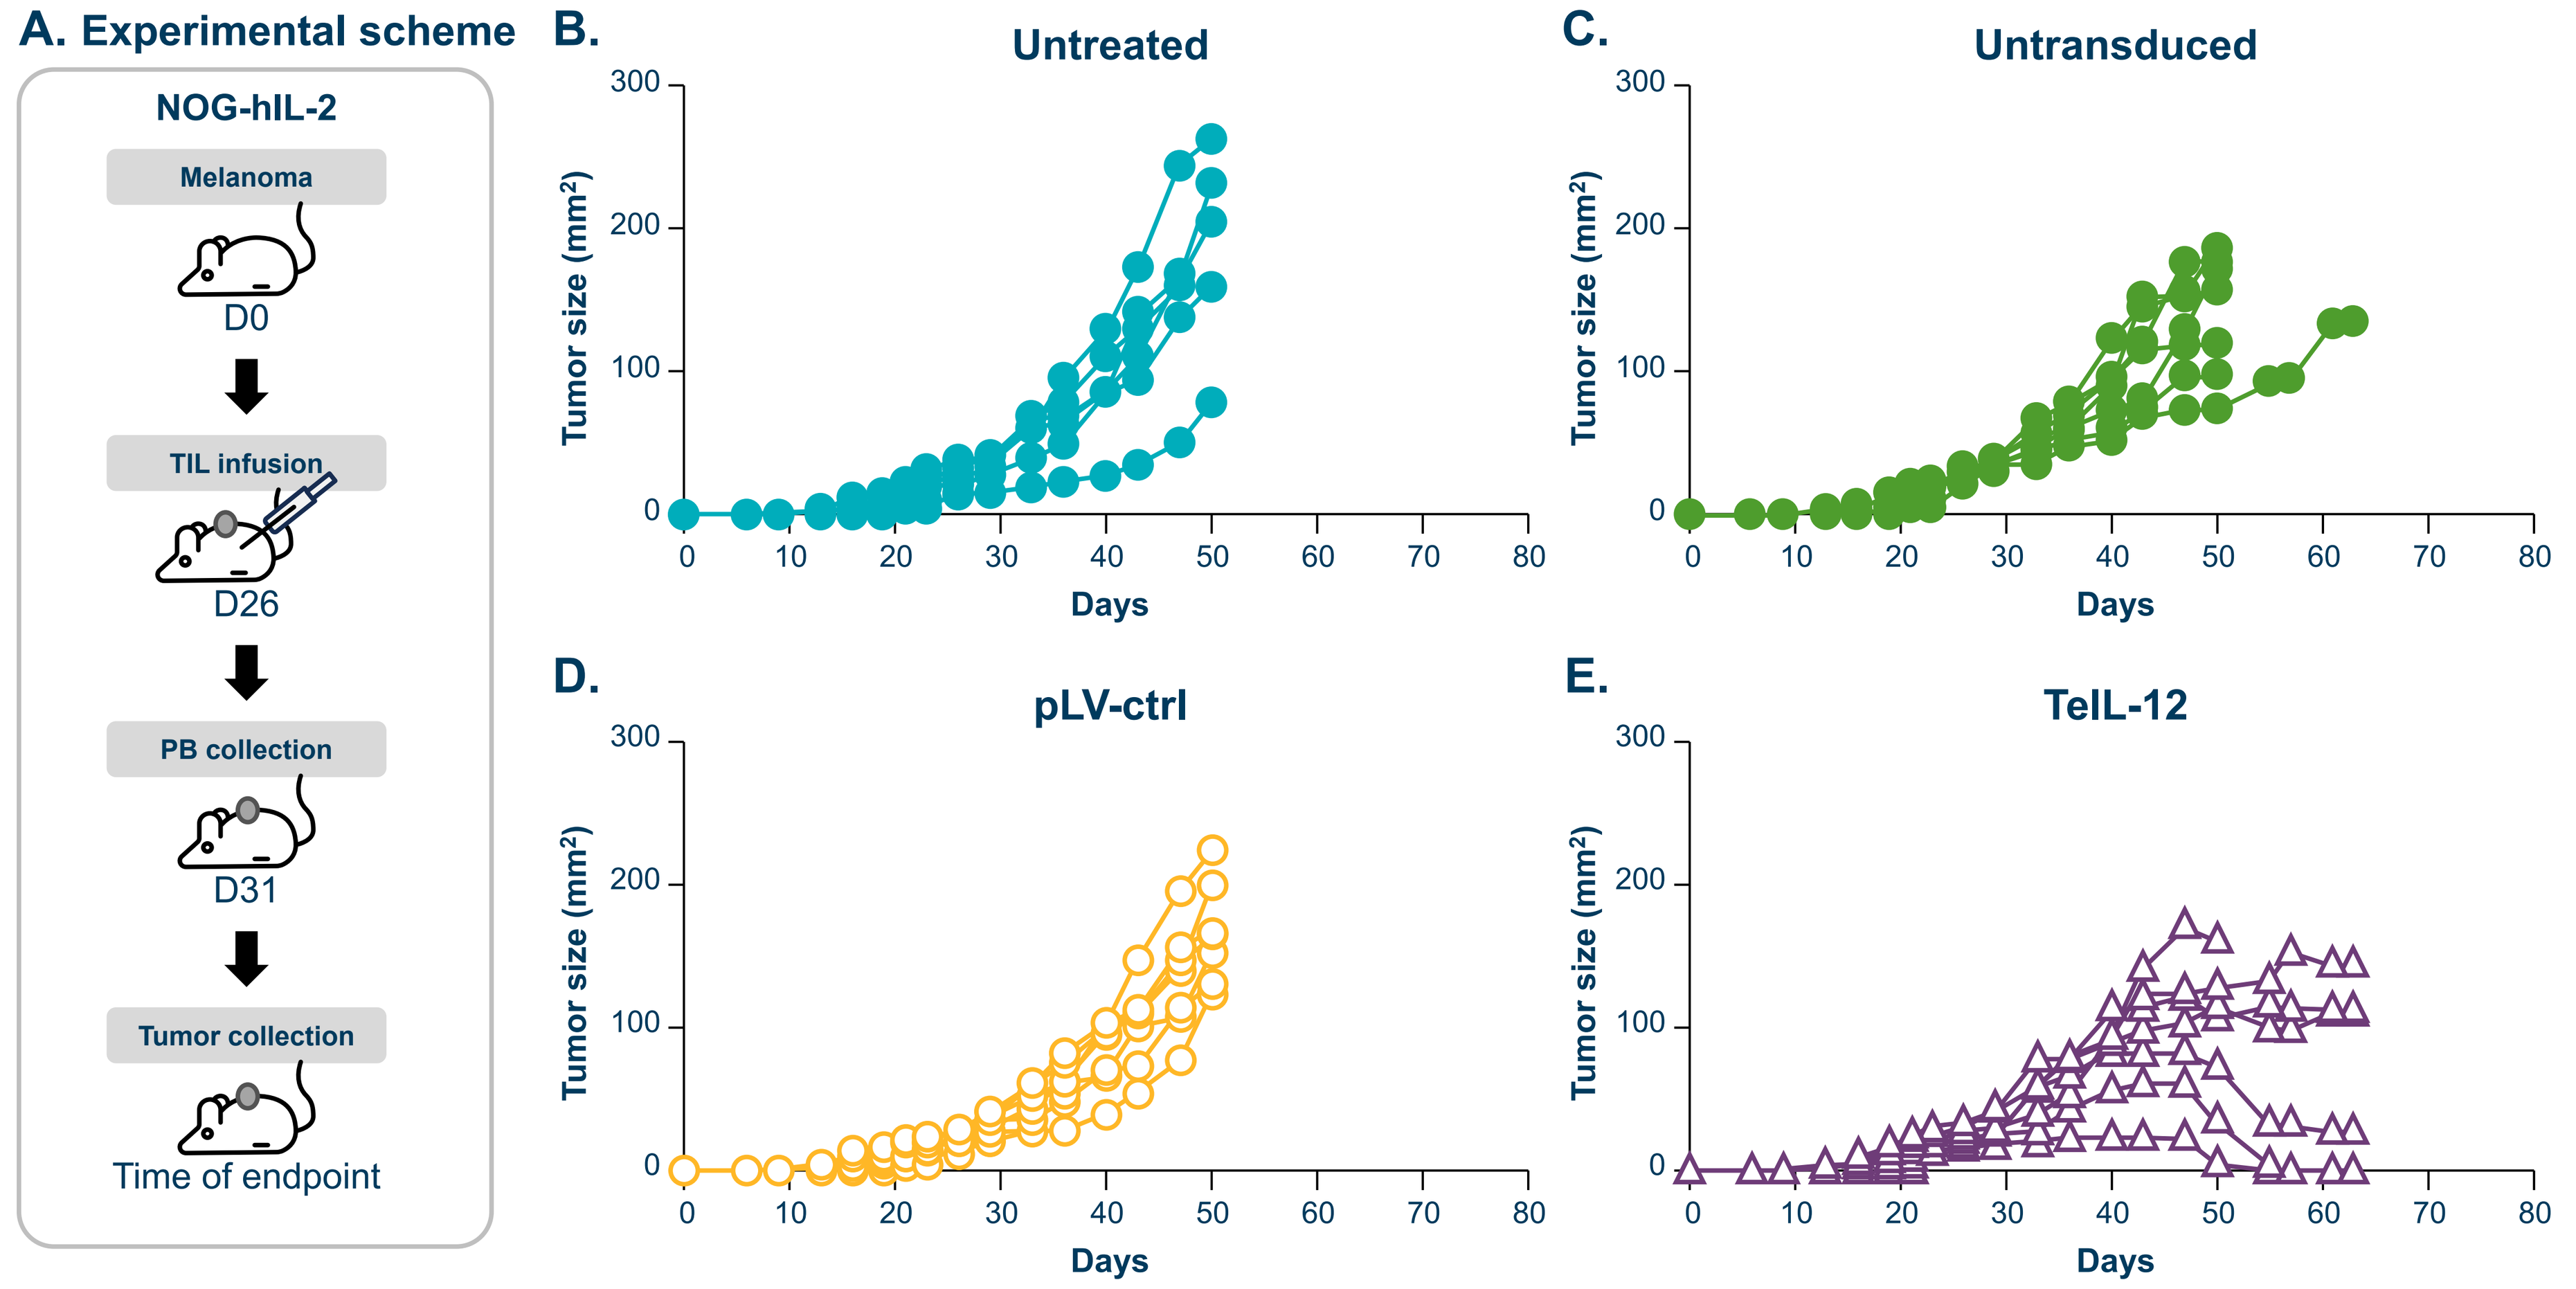
A. Experimental scheme
NOG-hIL-2
Melanoma
D0
TIL infusion
D26
PB collection
D31
Tumor collection
Time of endpoint
B. Untreated
C. Untransduced
D. pLV-ctrl
E. TeIL-12
Tumor size (mm²)
Days

Experimental scheme of *in vivo* melanoma PDX adoptive T-cell therapeutic model (A). Tumor growth curve of melanoma PDX without TIL infusion (B) or with the infusion of untransduced TIL (C), lentiviral control TIL (D), or TeIL-12 gene-engineered TIL (E). Seven mice were used in each group.

D, day; PB, peripheral blood; PDX, patient-derived xenograft; pLV-ctrl, lentiviral vector control; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes.

Figure 3. IL-12 and IFN-γ shedding was not detectable in circulation after infusion of TeIL-12–engineered TIL into mice

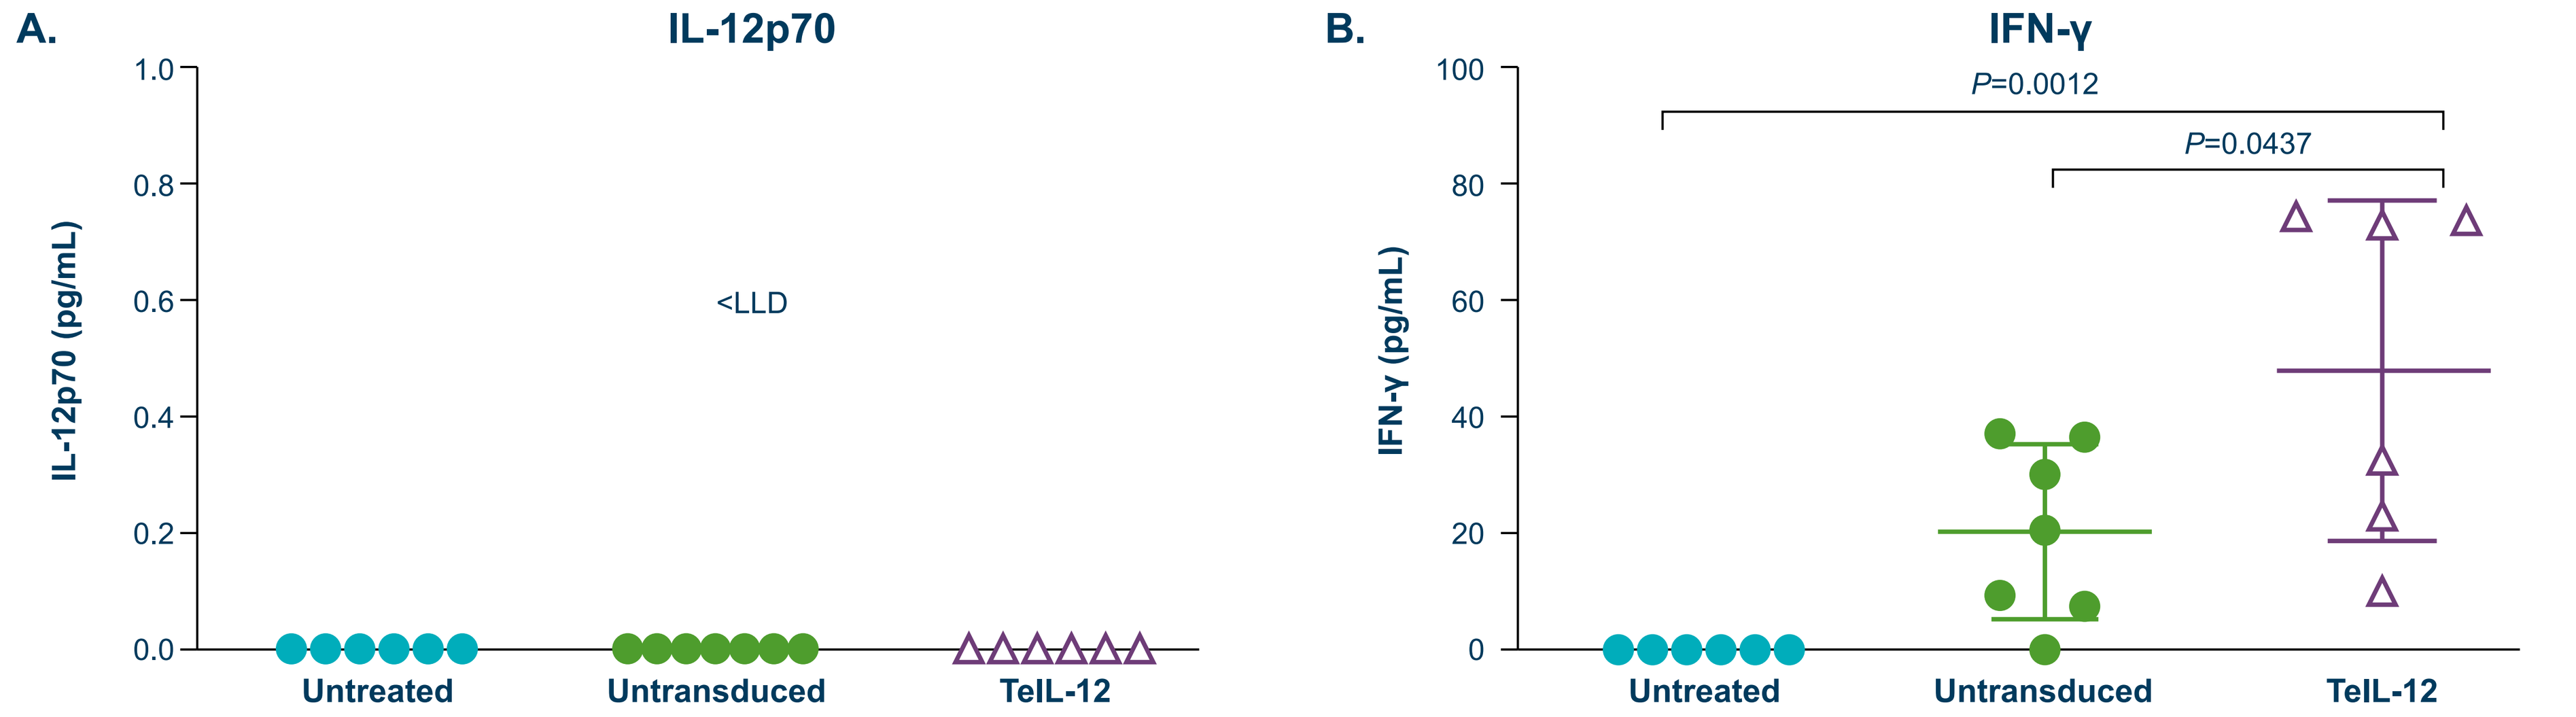
A. IL-12p70
IL-12p70 (pg/mL)
<LLD
B. IFN-γ
IFN-γ (pg/mL)
P=0.0012
P=0.0437
Untreated
Untransduced
TeIL-12

The expression levels of IL-12p70 (A) and IFN-γ (B) in peripheral blood of NOG-hIL-2 mice were examined 7 days after TIL infusion. Blood samples were collected from 6 to 7 mice. Data are displayed as mean ± SD. The LLD was >1.88 pg/mL. One-way analysis-of-variance with Tukey's test for multiple comparisons was performed by GraphPad Prism.

IFN-γ, interferon gamma; IL-12p70, interleukin-12 heterodimer; LLD, lower limit of detection; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes.

- Dual modification with TeIL-12 and an additional cytokine further enhanced the quality of the TIL product
 - The cytokine improved T-cell proliferation and survival in the absence of IL-2 (**Figure 4**)
 - Dual-cytokine–engineered TIL products showed improved *in vitro* autologous antitumor activity (**Figure 5**)

Figure 4. Tethered cytokine enhanced TIL survival/persistence *in vitro* in the absence of IL-2

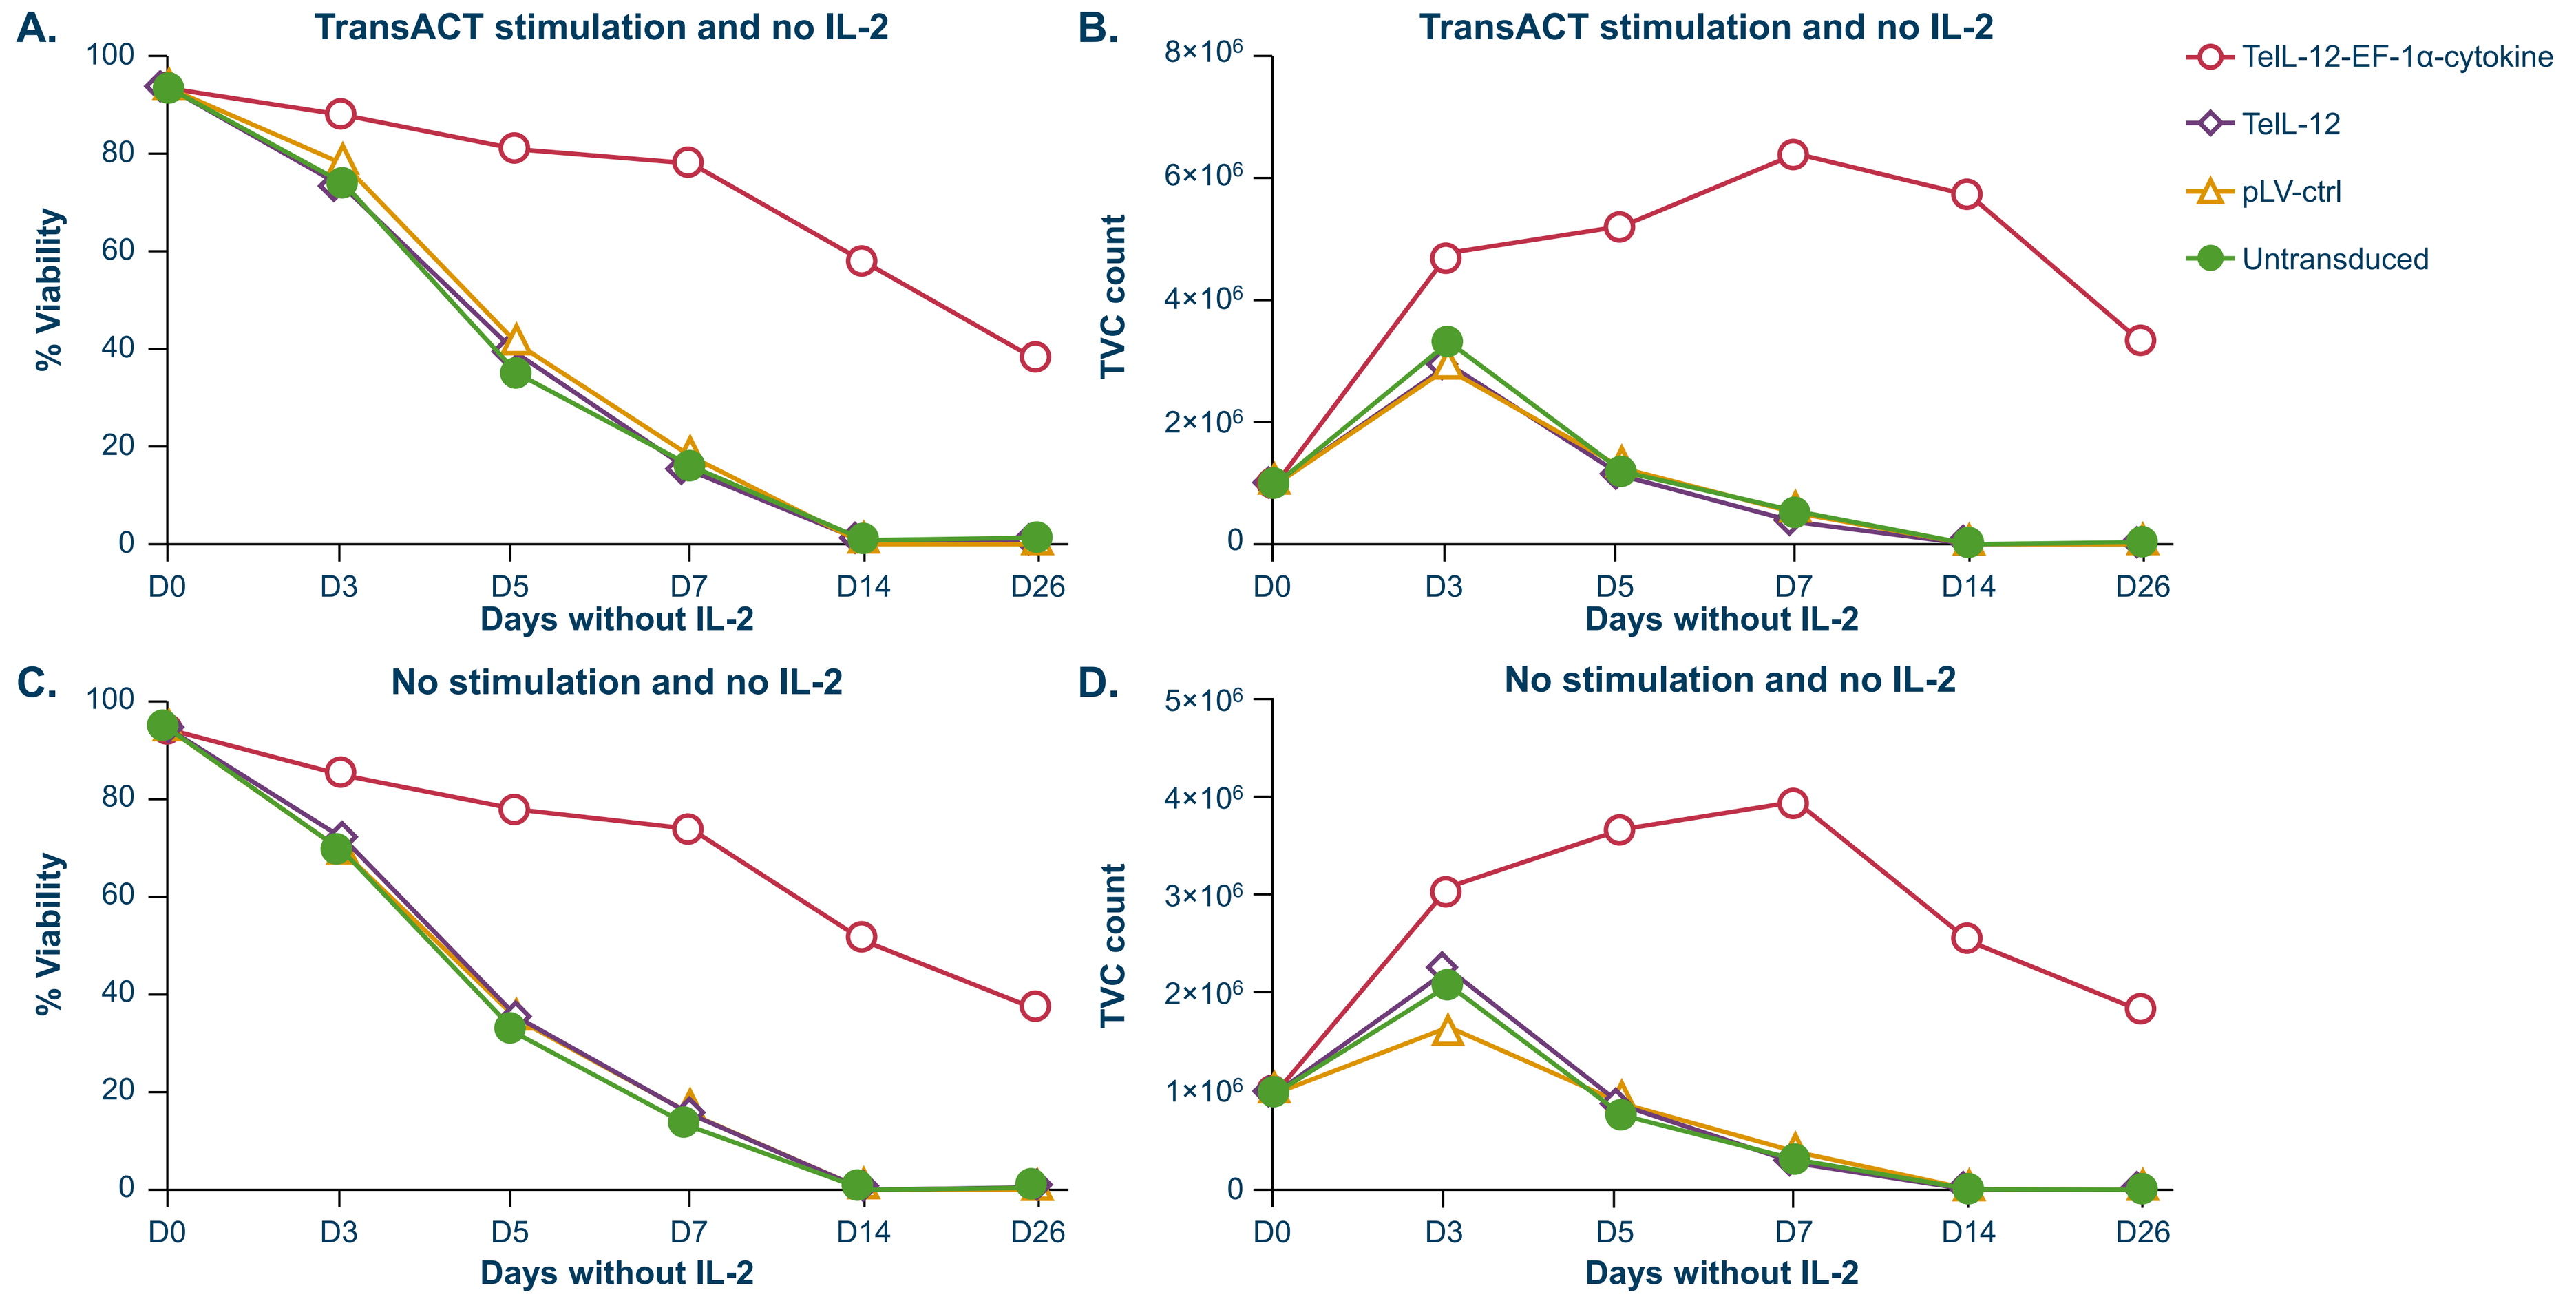
A. TransACT stimulation and no IL-2
B. TransACT stimulation and no IL-2
C. No stimulation and no IL-2
D. No stimulation and no IL-2
% Viability
TVC count
Days without IL-2
TeIL-12-EF-1α-cytokine
TeIL-12
pLV-ctrl
Untransduced

Viability (A, C) and TVC count (B, D) of the TIL product were dynamically examined during the *in vitro* culture in the IL-2–deficient medium with or without the addition of TransACT. One representative data set from more than 3 independent experiments is shown.

EF-1α, elongation factor 1α; IL-2, interleukin 2; pLV-ctrl, lentiviral vector control; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes; TVC, total viable cell.

Figure 5. Dual-cytokine–expressing TIL improved autologous antitumor activity *in vitro*

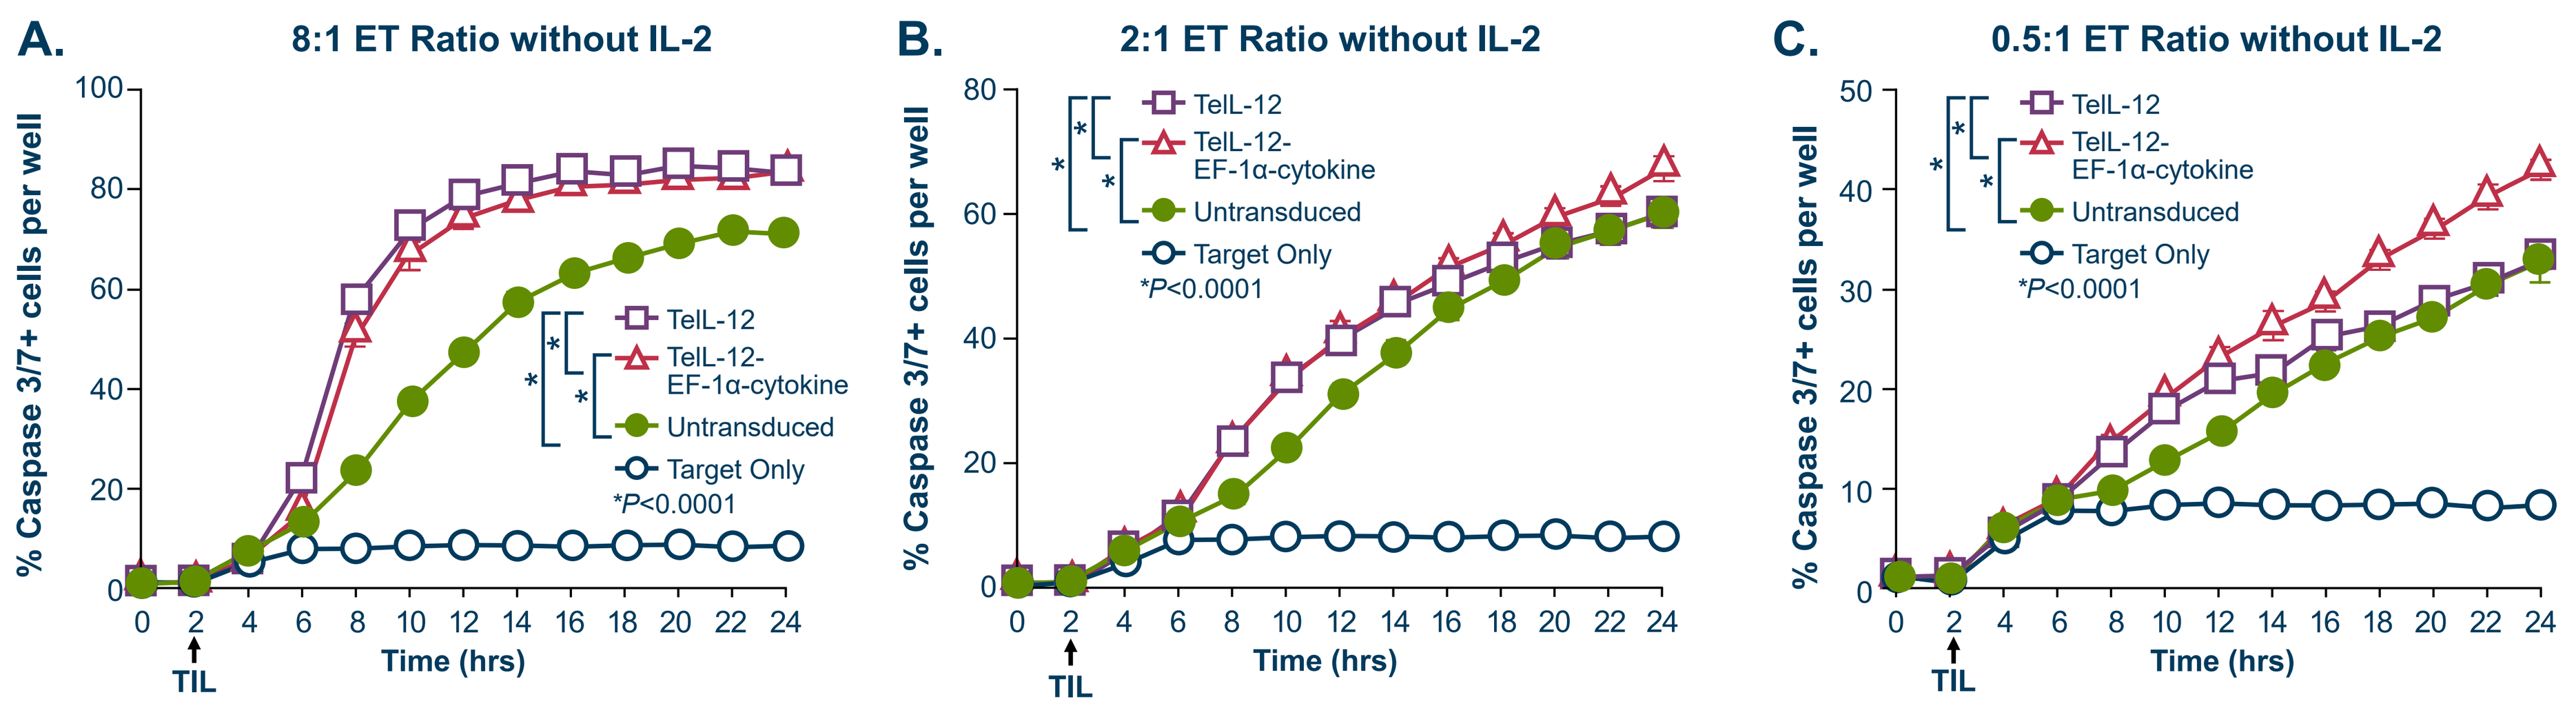
A. 8:1 ET Ratio without IL-2
B. 2:1 ET Ratio without IL-2
C. 0.5:1 ET Ratio without IL-2
% Caspase 3/7+ cells per well
Time (hrs)
TIL
TeIL-12
TeIL-12-EF-1α-cytokine
Untransduced
Target Only
*P<0.0001

Melanoma tumor cell (target) apoptosis was assessed by caspase 3/7 staining after adding the autologous TIL product (effector) with different E/Ts: 8:1 (A); 2:1 (B); 0.5:1 (C). The cytotoxicity assay was run in an IL-2–absent medium. Cell death was monitored dynamically with Incucyte detection of caspase 3/7-positive tumor cells. Two-way analysis-of-variance analysis with Dunnett's test for multiple comparisons was performed by GraphPad Prism.

EF-1α, elongation factor 1α; E/T, effector-to-target ratio; IL-2, interleukin 2; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes.

- Process modification of TIL combined with CCO medium increased the proportion of transduced cell yield by up to 50% (**Figure 6**); CCO medium increased IFN-γ production by dual-cytokine–transduced TIL when stimulated with autologous tumor digest (**Figure 7**)

Figure 6. Expansion with CCO leads to increased proportion of cytokine–positive cells in the final TIL product

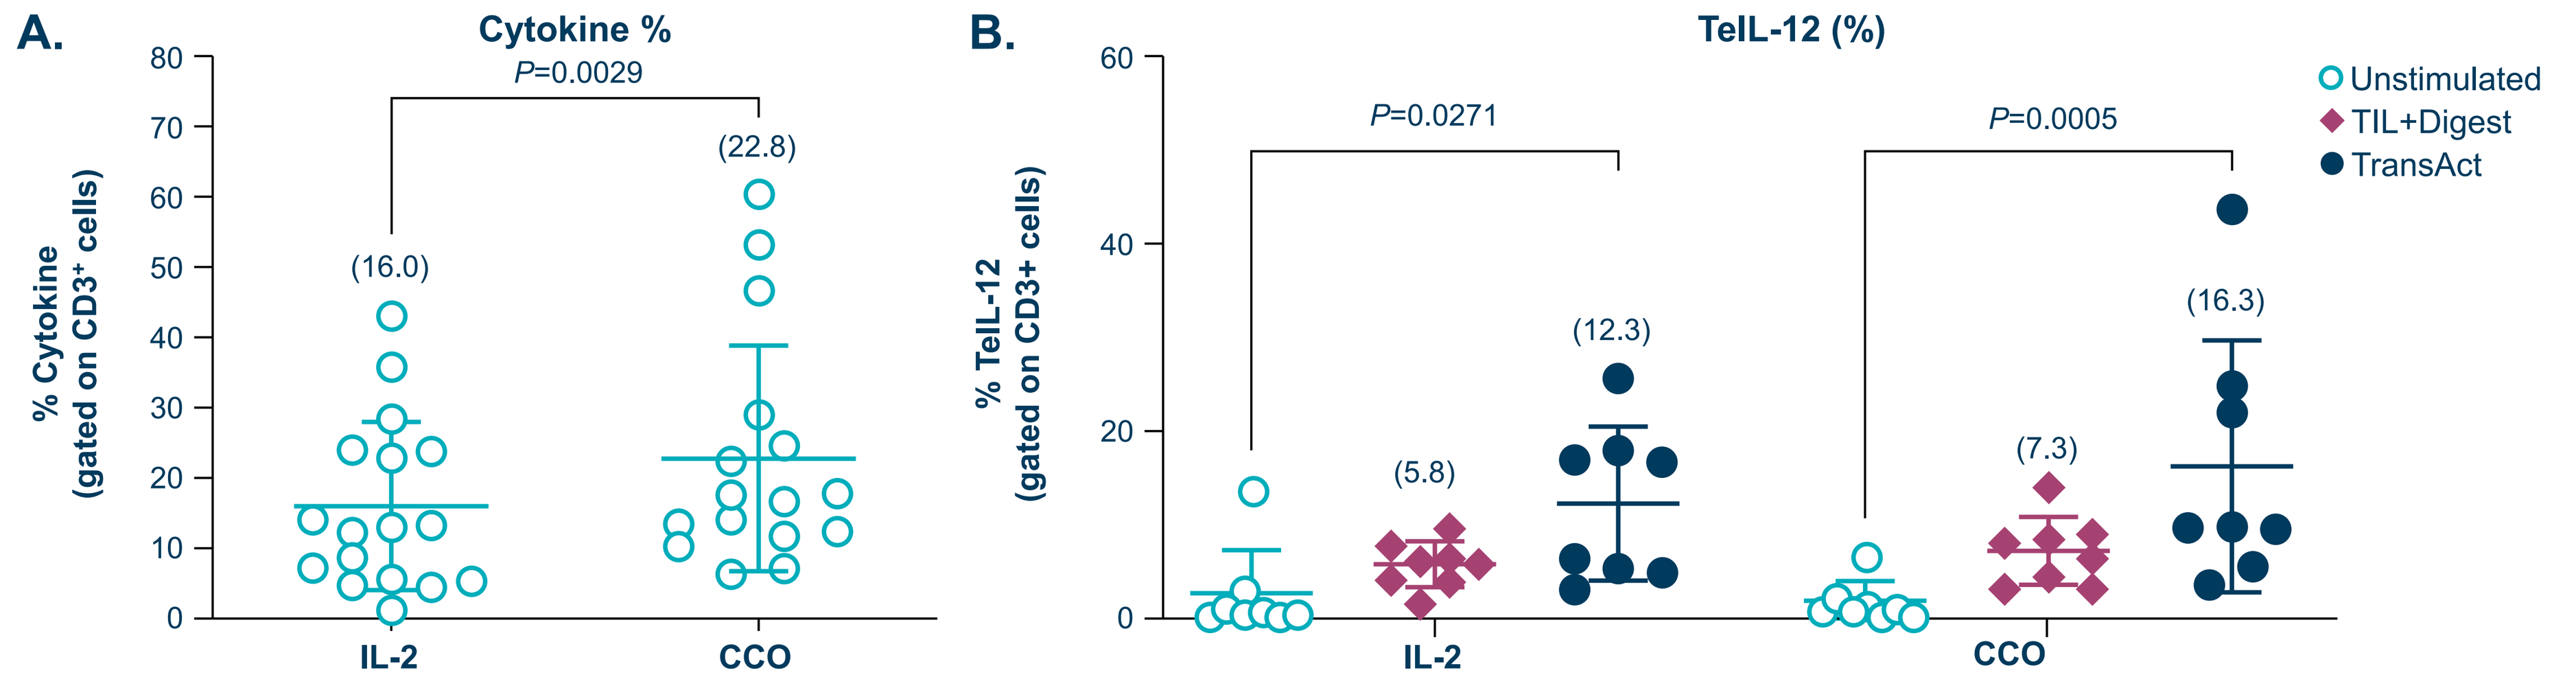
A. Cytokine %
% Cytokine (gated on CD3+ cells)
P=0.0029
(16.0)
(22.8)
IL-2
CCO
B. TeIL-12 (%)
% TeIL-12 (gated on CD3+ cells)
P=0.0271
P=0.0005
(5.8)
(12.3)
(7.3)
(16.3)
Unstimulated
TIL+Digest
TransAct

The proportion of cytokine–positive–expressing T cells was determined by flow cytometry on CD3-positive TIL within the TIL product at the end of the research scale manufacturing process (n=17, biological replicates) (A). Data are displayed as mean ± SD. Data analysis was performed by paired Student's t-test, two-tailed. The proportion of TeIL-12–positive–expressing T cells was determined by flow cytometry measurement of IL-12 before and after TCR stimulation using tumor digest or TransAct beads (1:100) (B). Data are displayed as mean ± SD. Two-way analysis-of-variance with Šídák's test for multiple comparisons was performed by GraphPad Prism.

CCO, cell culture optimization; IL-2, interleukin 2; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes.

Figure 7. Third modification with CCO further enhanced IFN-γ production upon autologous reactivity with TeIL-12 and a tethered cytokine

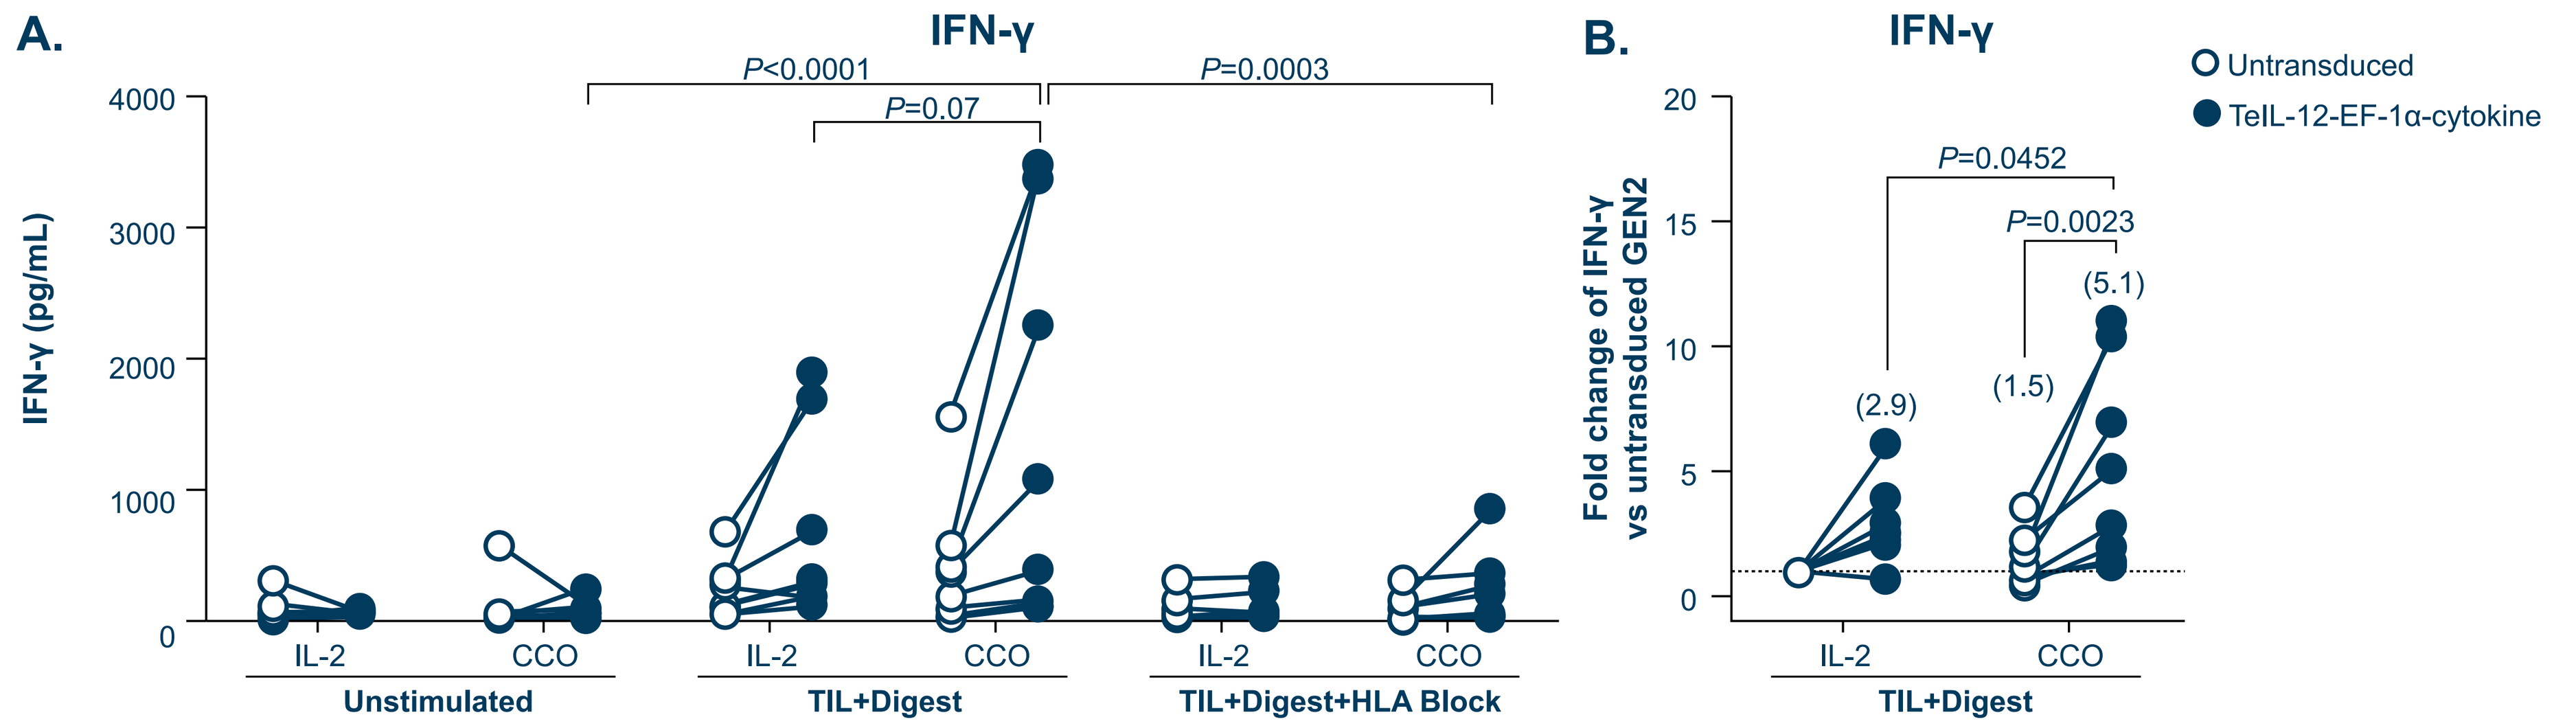
A. IFN-γ
IFN-γ (pg/mL)
P<0.0001
P=0.07
P=0.0003
Unstimulated
TIL+Digest
TIL+Digest+HLA Block
B. IFN-γ
Fold change of IFN-γ vs untransduced GEN2
P=0.0452
P=0.0023
(2.9)
(1.5)
(5.1)
Untransduced
TeIL-12-EF-1α-cytokine

IFN-γ production from TIL product in response to co-culture with autologous tumor digest from 8 biological donors (A). The experiment was set up in different groups: unstimulated, TIL+digest, and TIL+digest+HLA block. Two-way analysis-of-variance with Tukey's test for multiple comparisons was performed by GraphPad Prism. Fold change of IFN-γ against control TIL in the setting of TIL product co-culture with autologous tumor digest (B). Control TIL is defined as untransduced TIL expanded only in the presence of IL-2. Mean fold change is shown in parentheses. Two-way analysis-of-variance with uncorrected Fisher's least-significant-difference test for multiple comparisons was performed by GraphPad Prism.

CCO, cell culture optimization; EF-1α, elongation factor 1α; HLA, human leukocyte antigen; IFN-γ, interferon gamma; IL-2, interleukin 2; TeIL-12, membrane-tethered nuclear factor of activated T-cells–interleukin-12; TIL, tumor-infiltrating lymphocytes.

Conclusions

- **Expression of inducible membrane-tethered TeIL-12 on TIL showed potent *in vivo* killing efficacy**
- **Addition of another tethered cytokine further enhanced tumor killing and improved TIL persistence**
- **Combining dual-cytokine modifications with CCO enhanced TIL functionality**
- **These 3 approaches have the potential to increase TIL efficacy and may allow for modification of TIL regimens, including lymphodepletion and exogenously administered T-cell growth factors**

Disclosures

Patrick Innamarato, Nathan J. Gilbert, Alvaro de Mingo Pulido, Mohammed Alkhouli, Judy Fang, Gerard Sapena, Sean Hall, Hequn Yin, and Yongliang Zhang are employees of Iovance Biotherapeutics, Inc., San Carlos, CA, USA, and may own stock. Jamie L. Blauvelt and Shari Pilon-Thomas are employees of Moffitt Cancer Center, Tampa, FL, USA

References

1. AMTAGVI (lifileucel). www.amtagvi.com/. Accessed August 2024. 2. Natarajan A, et al. AACR 2022 Abstract #1015. 3. Zhang Y et al. SITC 2023 Abstract #403. 4. Zhang L, et al. Clin Cancer Res. 2015;21:2278–2288.

© 2024, Iovance Biotherapeutics