2552 2024 ASCO Annual Meeting | May 31–June 4, 2024 | Chicago, IL, USA

IOV-3001, a modified interleukin-2 fusion protein, for potential use in tumor-infiltrating lymphocyte cell therapy regimens

Michelle R. Simpson-Abelson,[1] Sadie Johnson,[1] Joanna Poprawski,[1] Jamie L. Blauvelt,[2] Sean Hall,[1] Hequn Yin[1]

[1]Iovance Biotherapeutics, Inc., San Carlos, CA, USA; [2]Moffitt Cancer Center, Tampa, FL, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors.

Background

- Cell therapy with tumor-infiltrating lymphocytes (TIL) has demonstrated efficacy in patients with solid tumor malignancies[1,2]
- Lifileucel, an FDA-approved one-time autologous TIL cell therapy, demonstrated clinically meaningful activity and durable responses in patients with advanced (unresectable or metastatic) melanoma[3,4]
- Interleukin-2 (IL-2) administration after TIL infusion supports the persistence and survival of infused TIL[5,6]
- Aldesleukin is a synthetic IL-2 with a short half-life and is administered every 8–12 hours to support the expansion and persistence of TIL *in vivo*
- IOV-3001 is a modified dimeric IL-2 fused to palivizumab that has a longer half-life and potentially better safety profile compared with aldesleukin
- Here, we describe the preclinical activity, pharmacokinetics (PK), pharmacodynamics (PD), and *in vivo* safety profile of IOV-3001 in cynomolgus monkeys

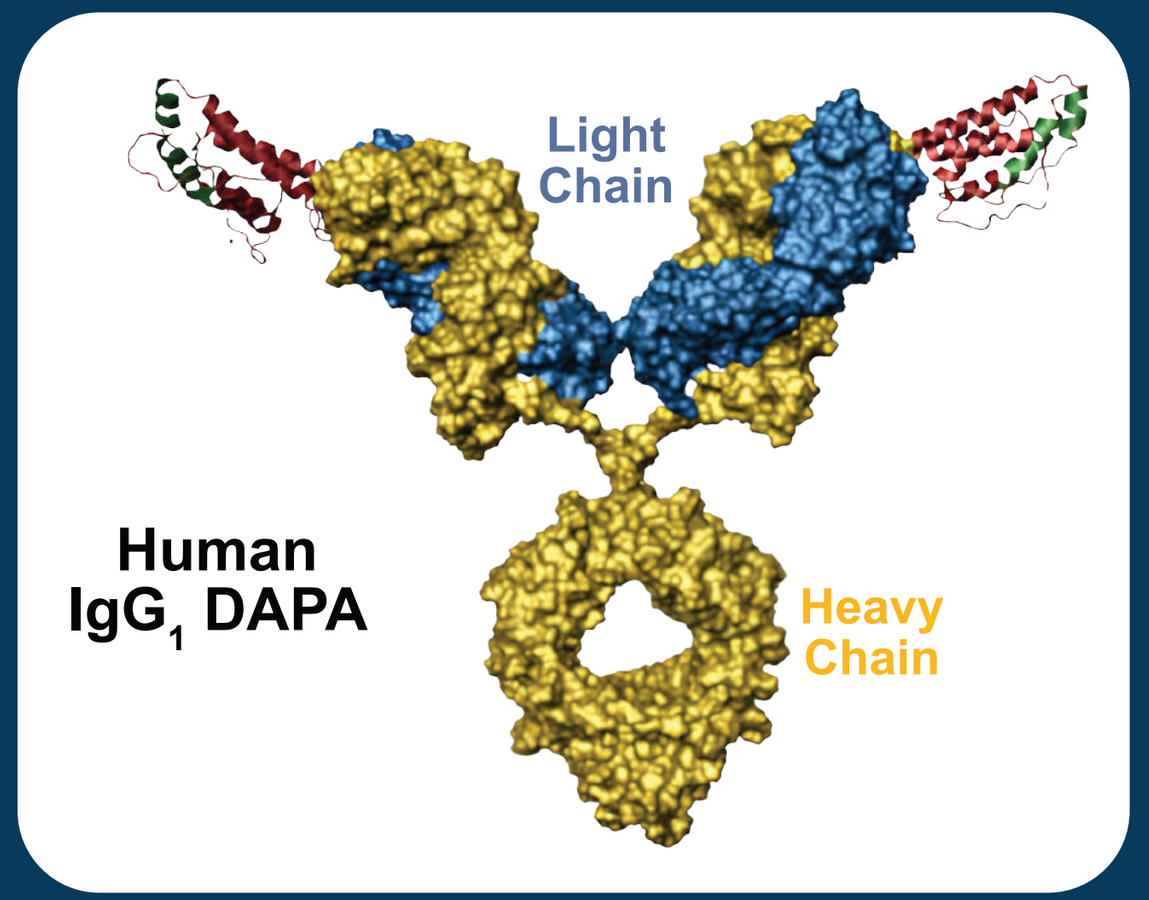
Light Chain
Human IgG$_1$ DAPA
Heavy Chain

Methods

- IOV-3001 and aldesleukin induced IL-2 receptor (IL-2R)–mediated activation of signal transducer and activator of transcription 5 (STAT5) phosphorylation was assessed in the peripheral blood mononuclear cells (PBMCs) obtained from healthy donors and TIL derived from patients with melanoma
- Anti-tumor efficacy and the pharmacological activities of IOV-3001 and aldesleukin were evaluated in C57BL/6 mice bearing established B16-D5 melanoma tumors infused with transgenic reactive splenocyte pmel-1 CD8+ T cells against the melanoma/melanocyte antigen, gp100$_{25–33}$
- In a 4-week GLP toxicology experiment, a single dose of IOV-3001 (3, 6, or 9 mg/kg) was administered to cynomolgus monkeys
- Blood samples were collected to evaluate toxicokinetic parameters, clinical pathology, and hematology
 - Pharmacodynamic parameters included serum cytokines and the cellular phenotype of PBMCs
 - Histopathological evaluation was performed in main study animals on Day 3 and in recovery animals on Day 29
- The impact of a single dose of IOV-3001 (3 or 6 mg/kg) on cardiovascular parameters was evaluated in cynomolgus monkeys in a separate GMP study

Figure 1. IOV-3001 Induced the Phosphorylation of STAT5 in PBMCs and TIL With Greater Potency Than Aldesleukin

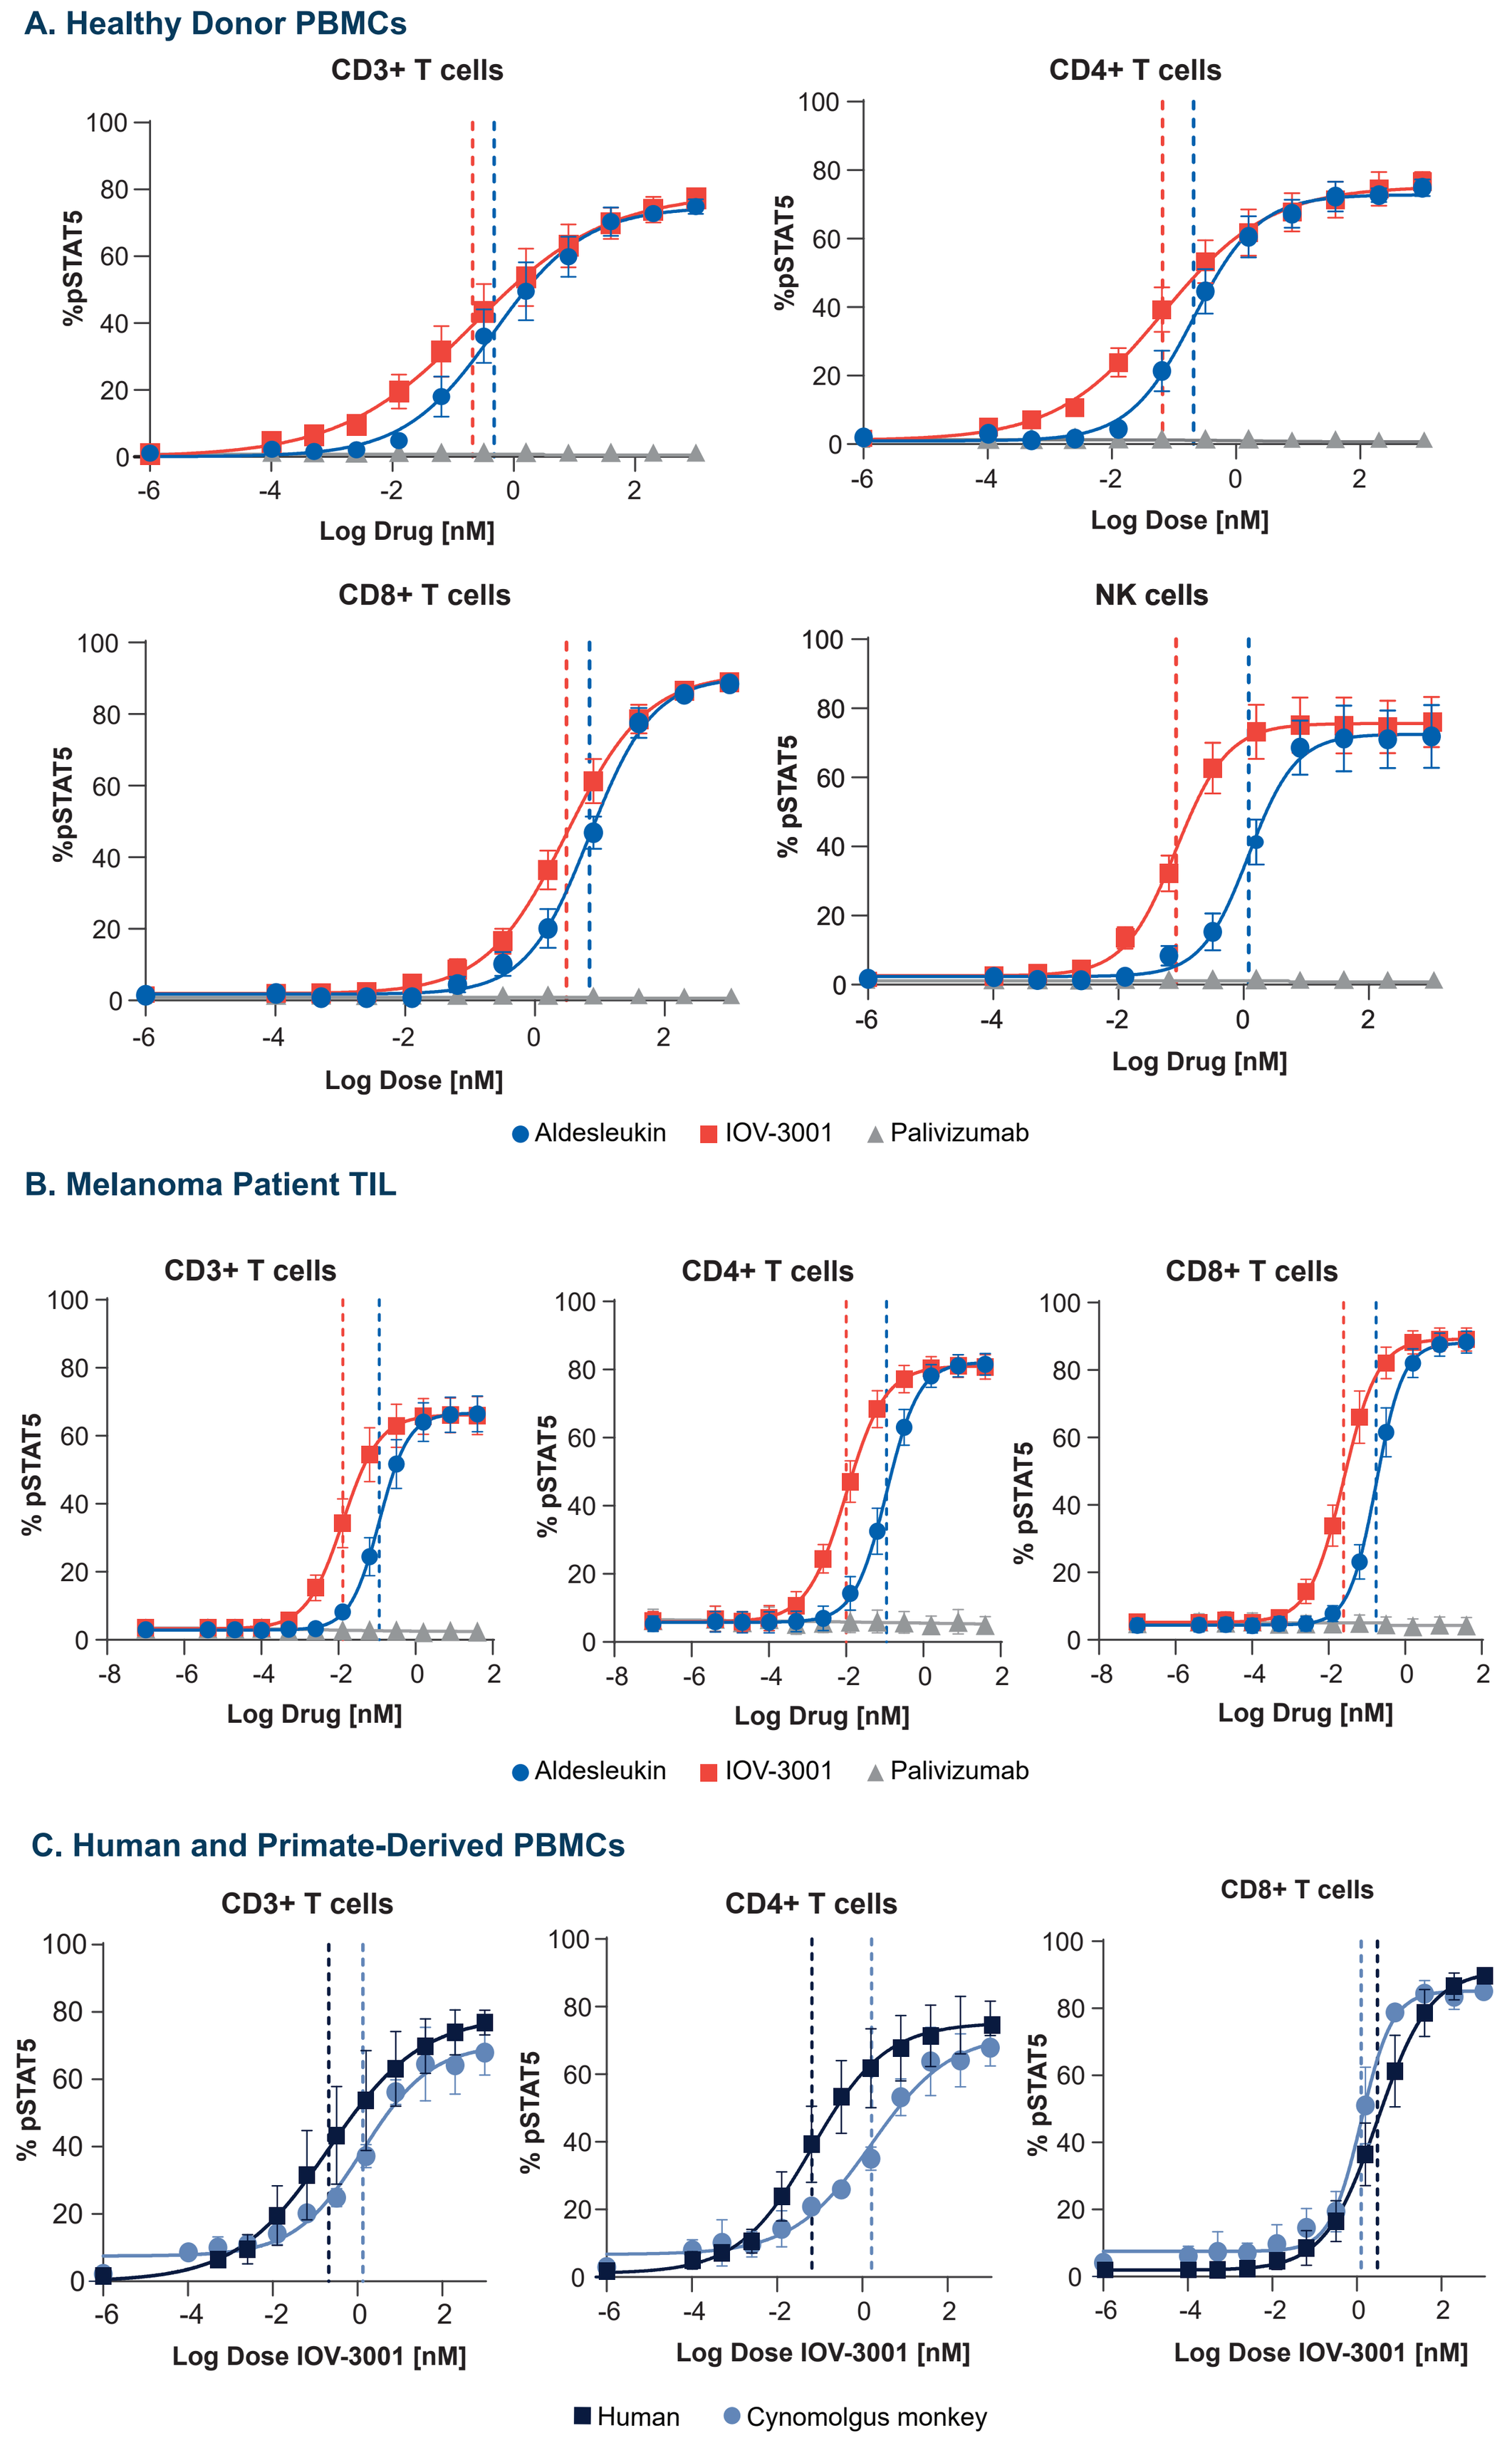
A. Healthy Donor PBMCs
CD3+ T cells
CD4+ T cells
CD8+ T cells
NK cells
%pSTAT5
Log Drug [nM]
Log Dose [nM]
Aldesleukin
IOV-3001
Palivizumab
B. Melanoma Patient TIL
CD3+ T cells
CD4+ T cells
CD8+ T cells
% pSTAT5
Log Drug [nM]
C. Human and Primate-Derived PBMCs
Log Dose IOV-3001 [nM]
Human
Cynomolgus monkey

Human PBMCs from normal healthy donors (n=3) (A), TIL from patients with melanoma (n=5) (B), and PBMCs from human and cynomolgus monkeys (n=3) (C) were assessed for phosphorylation of STAT5. Cells were incubated with IOV-3001 or aldesleukin at a dose range of 0.0001024 –1000 nM for 30 minutes and evaluated by flow cytometry to determine the percentage of pSTAT5+ T cells in the respective cell lineages. Symbols represent mean values, and vertical lines represent the standard error. Blue and red dotted vertical lines in panels A and B represent the logEC$_{50}$ for aldesleukin or IOV-3001, respectively. Black and grey dotted vertical lines in panel C represent the logEC$_{50}$ for human or cynomolgus monkey PBMCs, respectively.

© 2024, Iovance Biotherapeutics

Figure 2. Comparable Anti-tumor Activity of IOV-3001 and Aldesleukin With Similar PD Effects Posttreatment, Compared With T Cells Alone

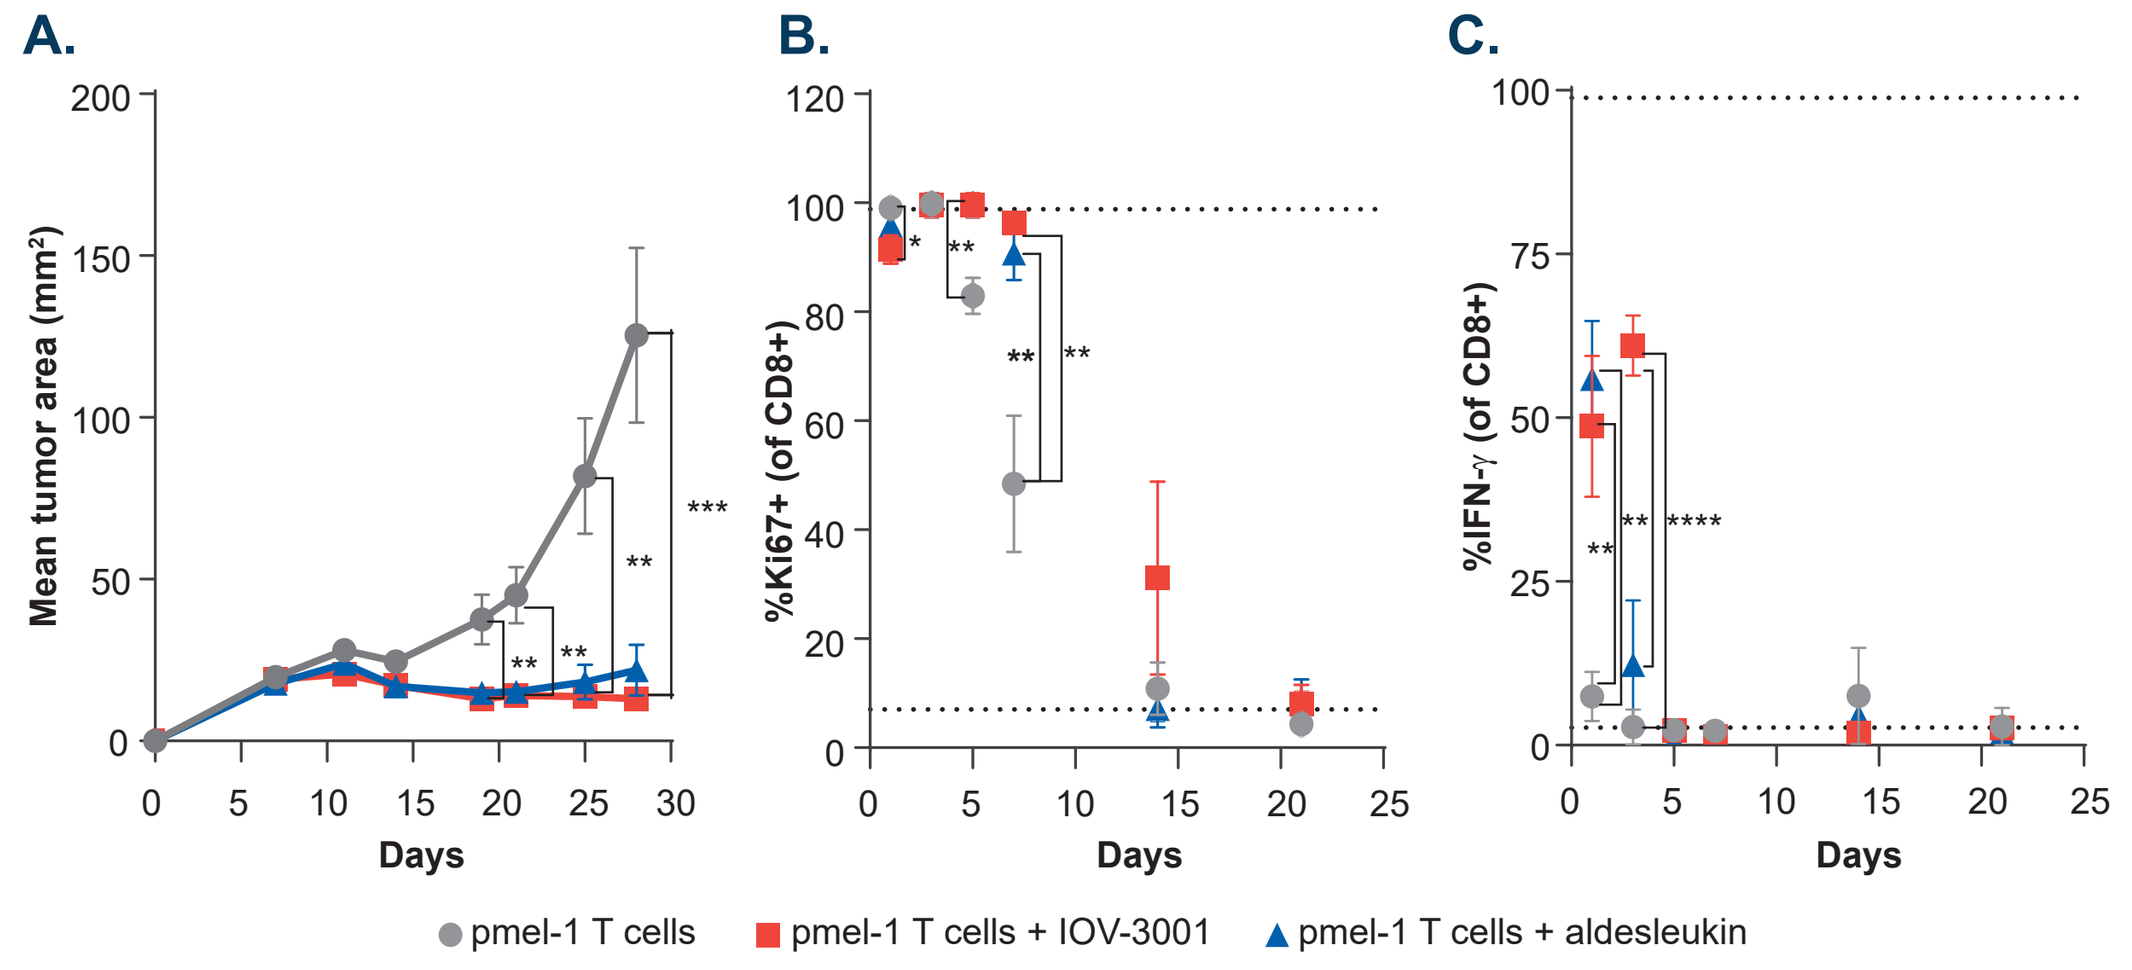
A.
B.
C.
Mean tumor area (mm²)
%Ki67+ (of CD8+)
%IFN-γ (of CD8+)
Days
pmel-1 T cells
pmel-1 T cells + IOV-3001
pmel-1 T cells + aldesleukin

B16-D5 syngeneic murine melanoma cells were subcutaneously implanted in C57BL/6 mice on Day -7; mice then underwent lymphodepletion with cyclophosphamide (200 mg/kg, Day -2) and fludarabine (100 mg/kg, Day -1). At Day 0, mice received an intravenous infusion of 5e6 cultured pmel-1 CD8+ splenocyte T cells that were pretreated with gp10025-33 peptide (5 µg/mL) and IL-2 (10 IU/mL) for 3 to 5 days. A single dose of IOV-3001 (3 mg/kg) or 6 doses of aldesleukin (250,000 IU every 8–12 hours), via intraperitoneal injection was administered within 1 hour of pmel T cell infusion. Tumors were measured using a caliper to determine mean tumor area (length × width) (A). In an independent experiment, groups of 4 mice treated with either cultured pmel-1 T cells alone or in combination with aldesleukin, or IOV-3001 were sacrificed on Days 1, 3, 5, 7, 15, and 21 post–adoptive pmel-1 T cell transfer. Spleens were harvested and stained, and flow cytometry was used to evaluate the expression of phenotypic markers within the CD8+ T-cell population including the percentage of Ki67+ cells (B) and the percentage of cells expressing IFN-γ (C). Symbols represent mean values and vertical lines represent the standard deviation. An analysis of repeated measures with the animal as the subject over time was used to compare the treatment groups. Tukey adjusted post-hoc comparisons were performed for all two-way comparisons between treatment groups at each time point. $^*p<0.05$. $^{**}p<0.01$. $^{***}p<0.001$. $^{****}p<0.0001$.

Figure 3. IOV-3001 Displayed a Prolonged Half-life in Cynomolgus Monkeys

A.

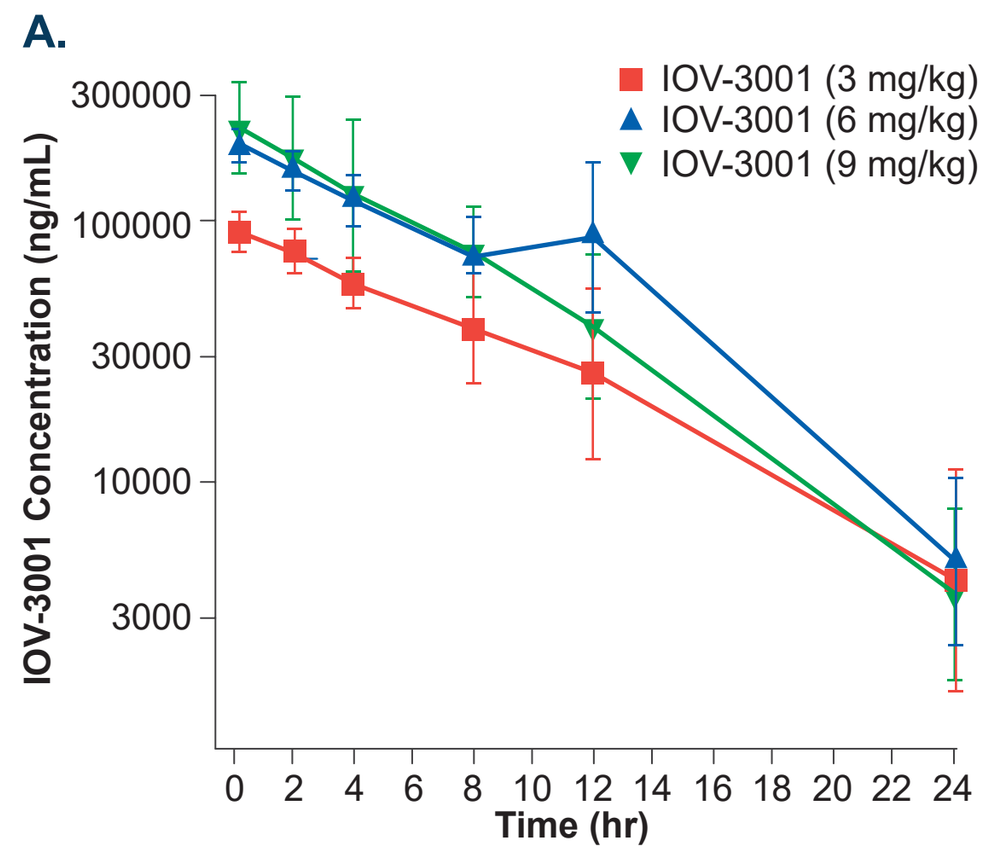
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)
IOV-3001 Concentration (ng/mL)
Time (hr)

B.

Dose Group	Sex	C_{max} (Mean ± Standard Error)		AUC_{last}		$t_{1/2}$	
Group 2: IOV-3001 (3 mg/kg)	Male	88300	± 3980	925000	± 138000	5.14	± 0.737
	Female	94500	± 9730	791000	± 119000	4.47	± 0.573
	M/F Mean	**91400**	**± 3100**	**858000**	**± 67000**	**4.81**	**± 0.337**
Group 3: IOV-3001 (6 mg/kg)	Male	201000	± 9820	1930000	± 182000	5.31	± 1.14
	Female	192000	± 15200	1730000	± 234000	4.59	± 0.555
	M/F Mean	**196000**	**± 4200**	**1830000**	**± 99400**	**4.95**	**± 0.361**
Group 4: IOV-3001 (9 mg/kg)	Male	242000	± 10600	1840000	± 133000	3.84	± 0.244
	Female	239000	± 41500	1500000	± 338000	3.31	± 0.137
	M/F Mean	**240000**	**± 1570**	**1670000**	**± 169000**	**3.58**	**± 0.264**

Cynomolgus monkeys received a single intravenous infusion of IOV-3001 (3, 6, or 9 mg/kg). Blood samples were obtained at the following post-infusion time points: 0.25, 2, 4, 8, and 12 hours, and Days 1, 2, 3, 5, and 7. The concentration of IOV-3001 was assessed using an ELISA-based PK assay. Standard PK parameters were calculated using a non-compartmental analysis. Graph shows the mean serum concentration of IOV-3001 over time (A) and table shows IOV-3001 toxicokinetic parameters by sex across dose groups (B). Symbols represent mean values and vertical lines represent the standard error.

Figure 4. Human Equivalent PK of IOV-3001 Scaled From Cynomolgus Monkey Predicts a NOAEL in Human of 3.5 mg/kg

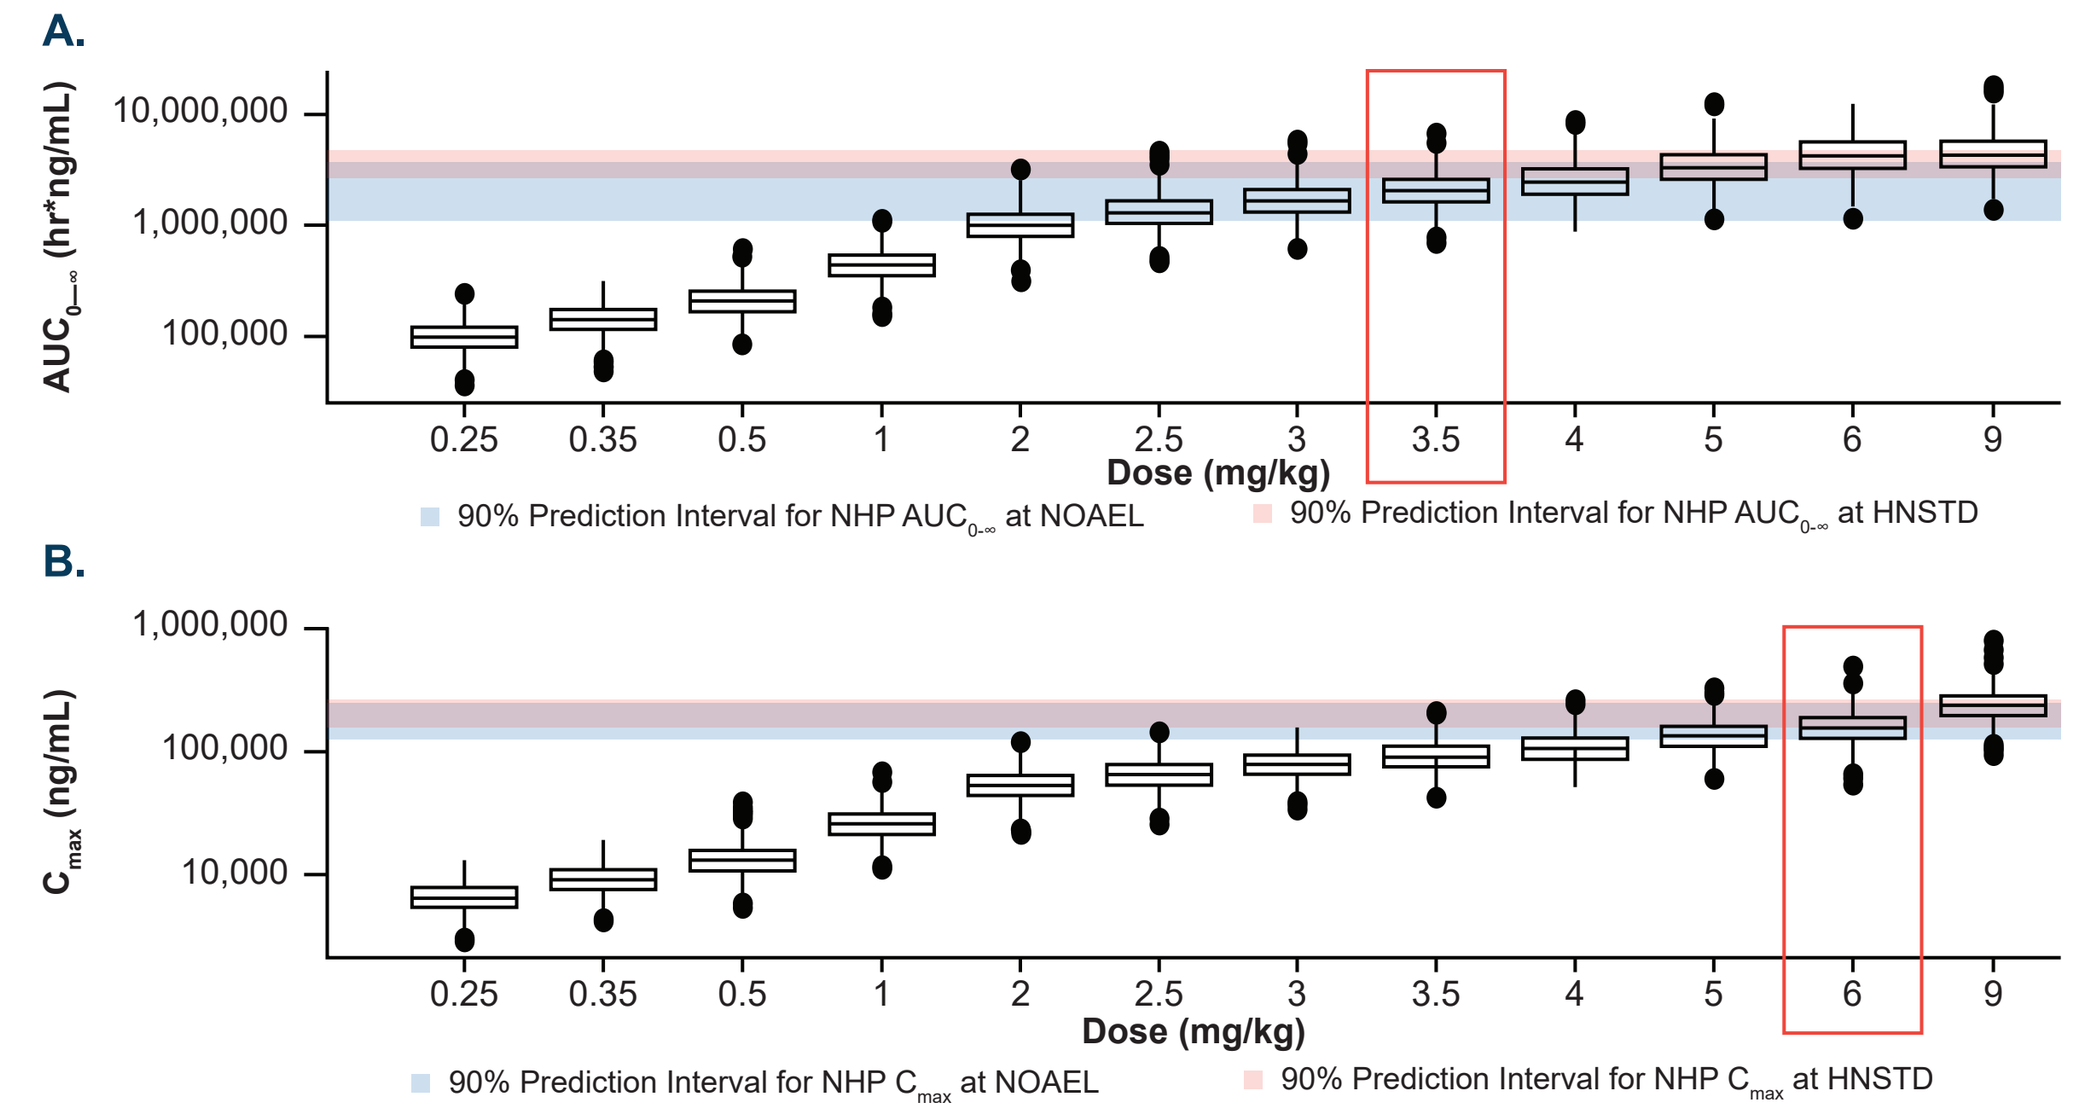
A.
AUC$_{0-\infty}$ (hr*ng/mL)
Dose (mg/kg)
90% Prediction Interval for NHP AUC$_{0-\infty}$ at NOAEL
90% Prediction Interval for NHP AUC$_{0-\infty}$ at HNSTD
B.
C$_{max}$ (ng/mL)
90% Prediction Interval for NHP C$_{max}$ at NOAEL
90% Prediction Interval for NHP C$_{max}$ at HNSTD

Cynomolgus monkeys received a single intravenous infusion of IOV-3001 (3, 6, or 9 mg/kg). Blood samples were obtained at various post-infusion time points (0.25, 2, 4, 8, and 12 hours, and Days 1, 2, 3, 5, and 7). Standard PK parameters were calculated using a non-compartmental analysis. Each PK parameter was scaled from cynomolgus monkeys to humans, resulting in simulated full concentration–time profiles. Human equivalent doses for AUC (A) and C_{max} (B) were calculated. The cynomolgus monkey NOAEL was 6 mg/kg (HED=3.5 mg/kg), and the HNSTD was 9 mg/kg (HED=5 mg/kg). Red boxes indicate human equivalent doses for AUC NOAEL and C_{max} NOAEL.

- A one-compartment model with nonlinear elimination adequately described the kinetics of IOV-3001 in non-human primates

Figure 5. IOV-3001 Displayed a Favorable Safety Profile in Cynomolgus Monkeys at Single Doses Up to 6 mg/kg

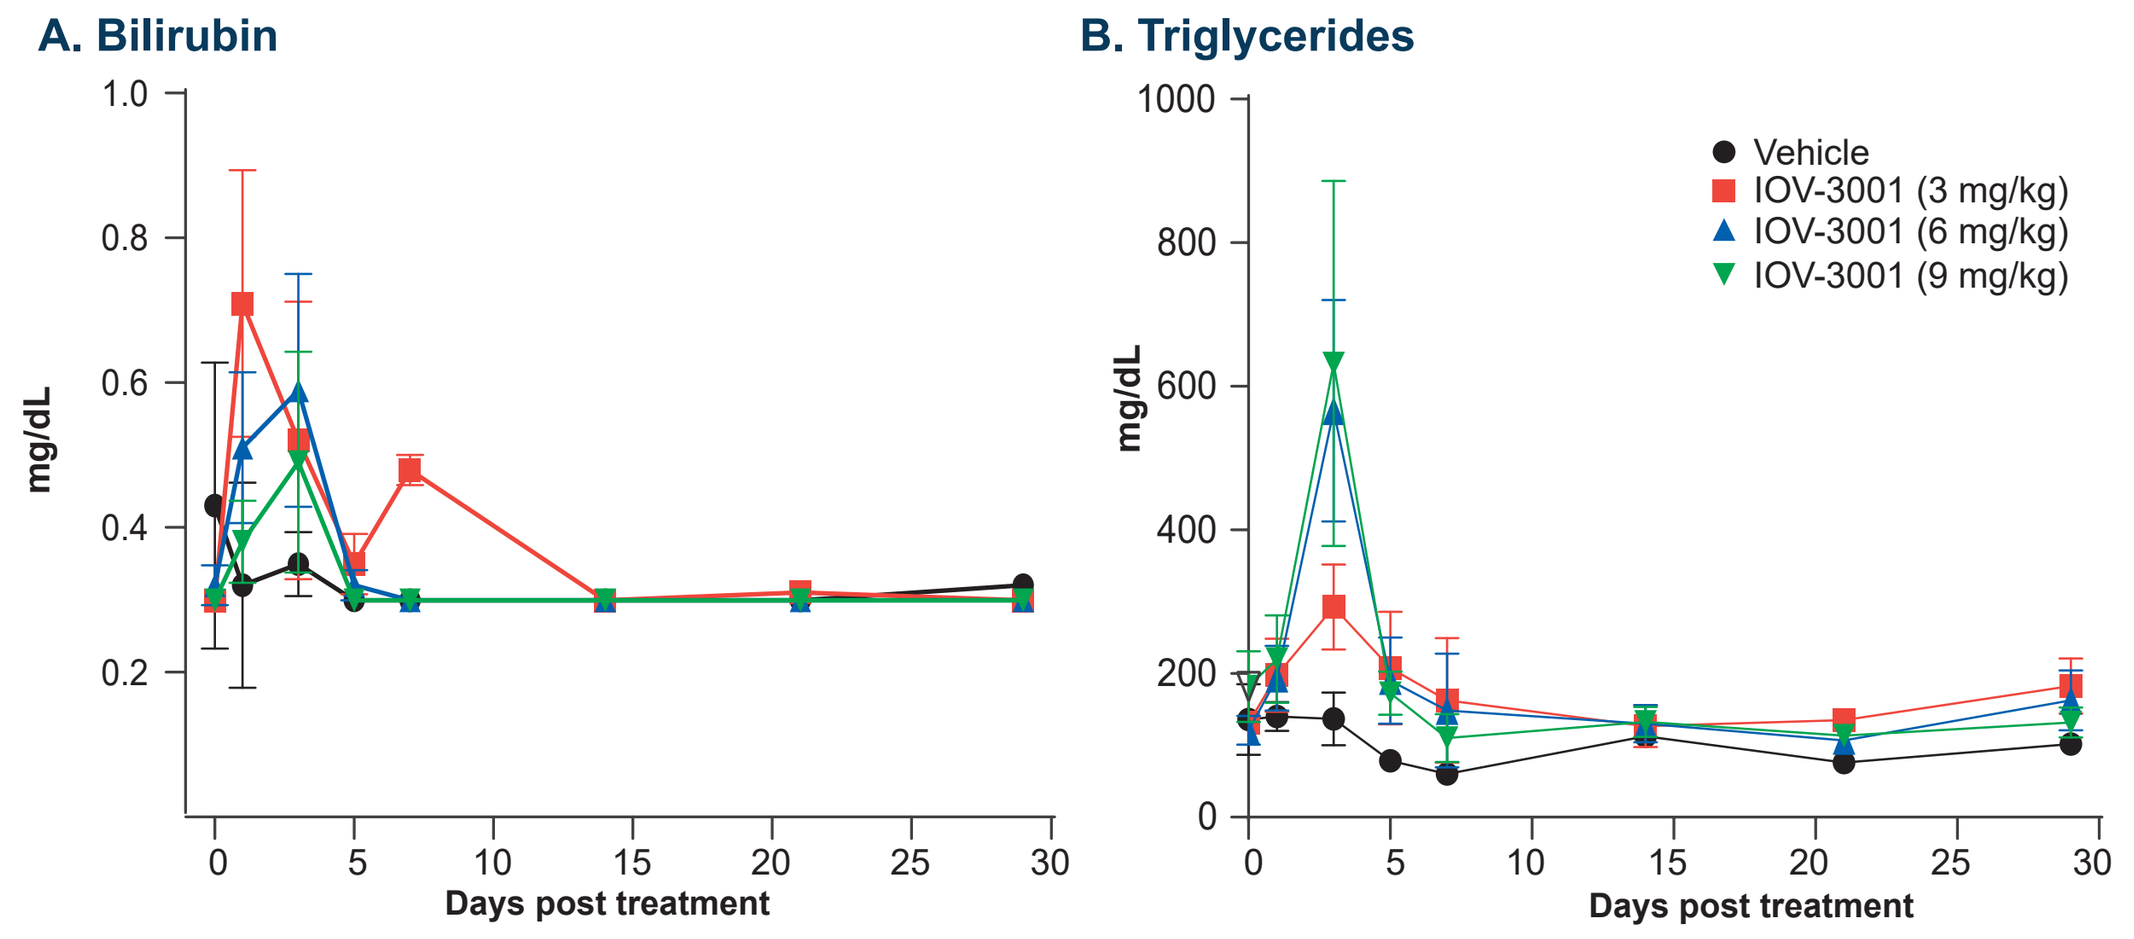
A. Bilirubin
B. Triglycerides
mg/dL
Days post treatment
Vehicle
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)

Whole blood was collected from all animals prior to IOV-3001 infusion and at Days 1, 3, 5, 7, 14, 21- and 29 post-infusion. Serum samples were analyzed on a Hitachi Cobas c501Chemistry Analyzer (Roche Diagnostics, Indianapolis, IN). Symbols represent mean values and vertical lines represent the standard deviation.

- The IOV-3001 9 mg/kg dose was the HNSTD based on clinical observations in one (out of 10) animals
- Observed symptoms included lethargy, loose stool, dehydration and hunched posture
 - Symptoms started on Day 1 with recovery of all symptoms by Day 14 post-infusion
- A trend toward increasing bilirubin and elevated triglycerides at Day 3 were observed, followed by a return to baseline levels by Day 7 (**Figure 5**)
- IOV-3001 induced increases in the number and infiltration of leukocytes in the bone marrow, spleen, lymph nodes, liver, and heart at Day 3; these changes were less prominent or had resolved by Day 29
- No effects related to IOV-3001 were observed on heart rate, RR interval, PR interval, QRS duration, QT interval, or QTc interval, across the 96-hour duration of the study
- No effects related to IOV-3001 on mean diastolic or systolic blood pressure were observed

Figure 6. IOV-3001 Induced Increases in Immune Cell Numbers

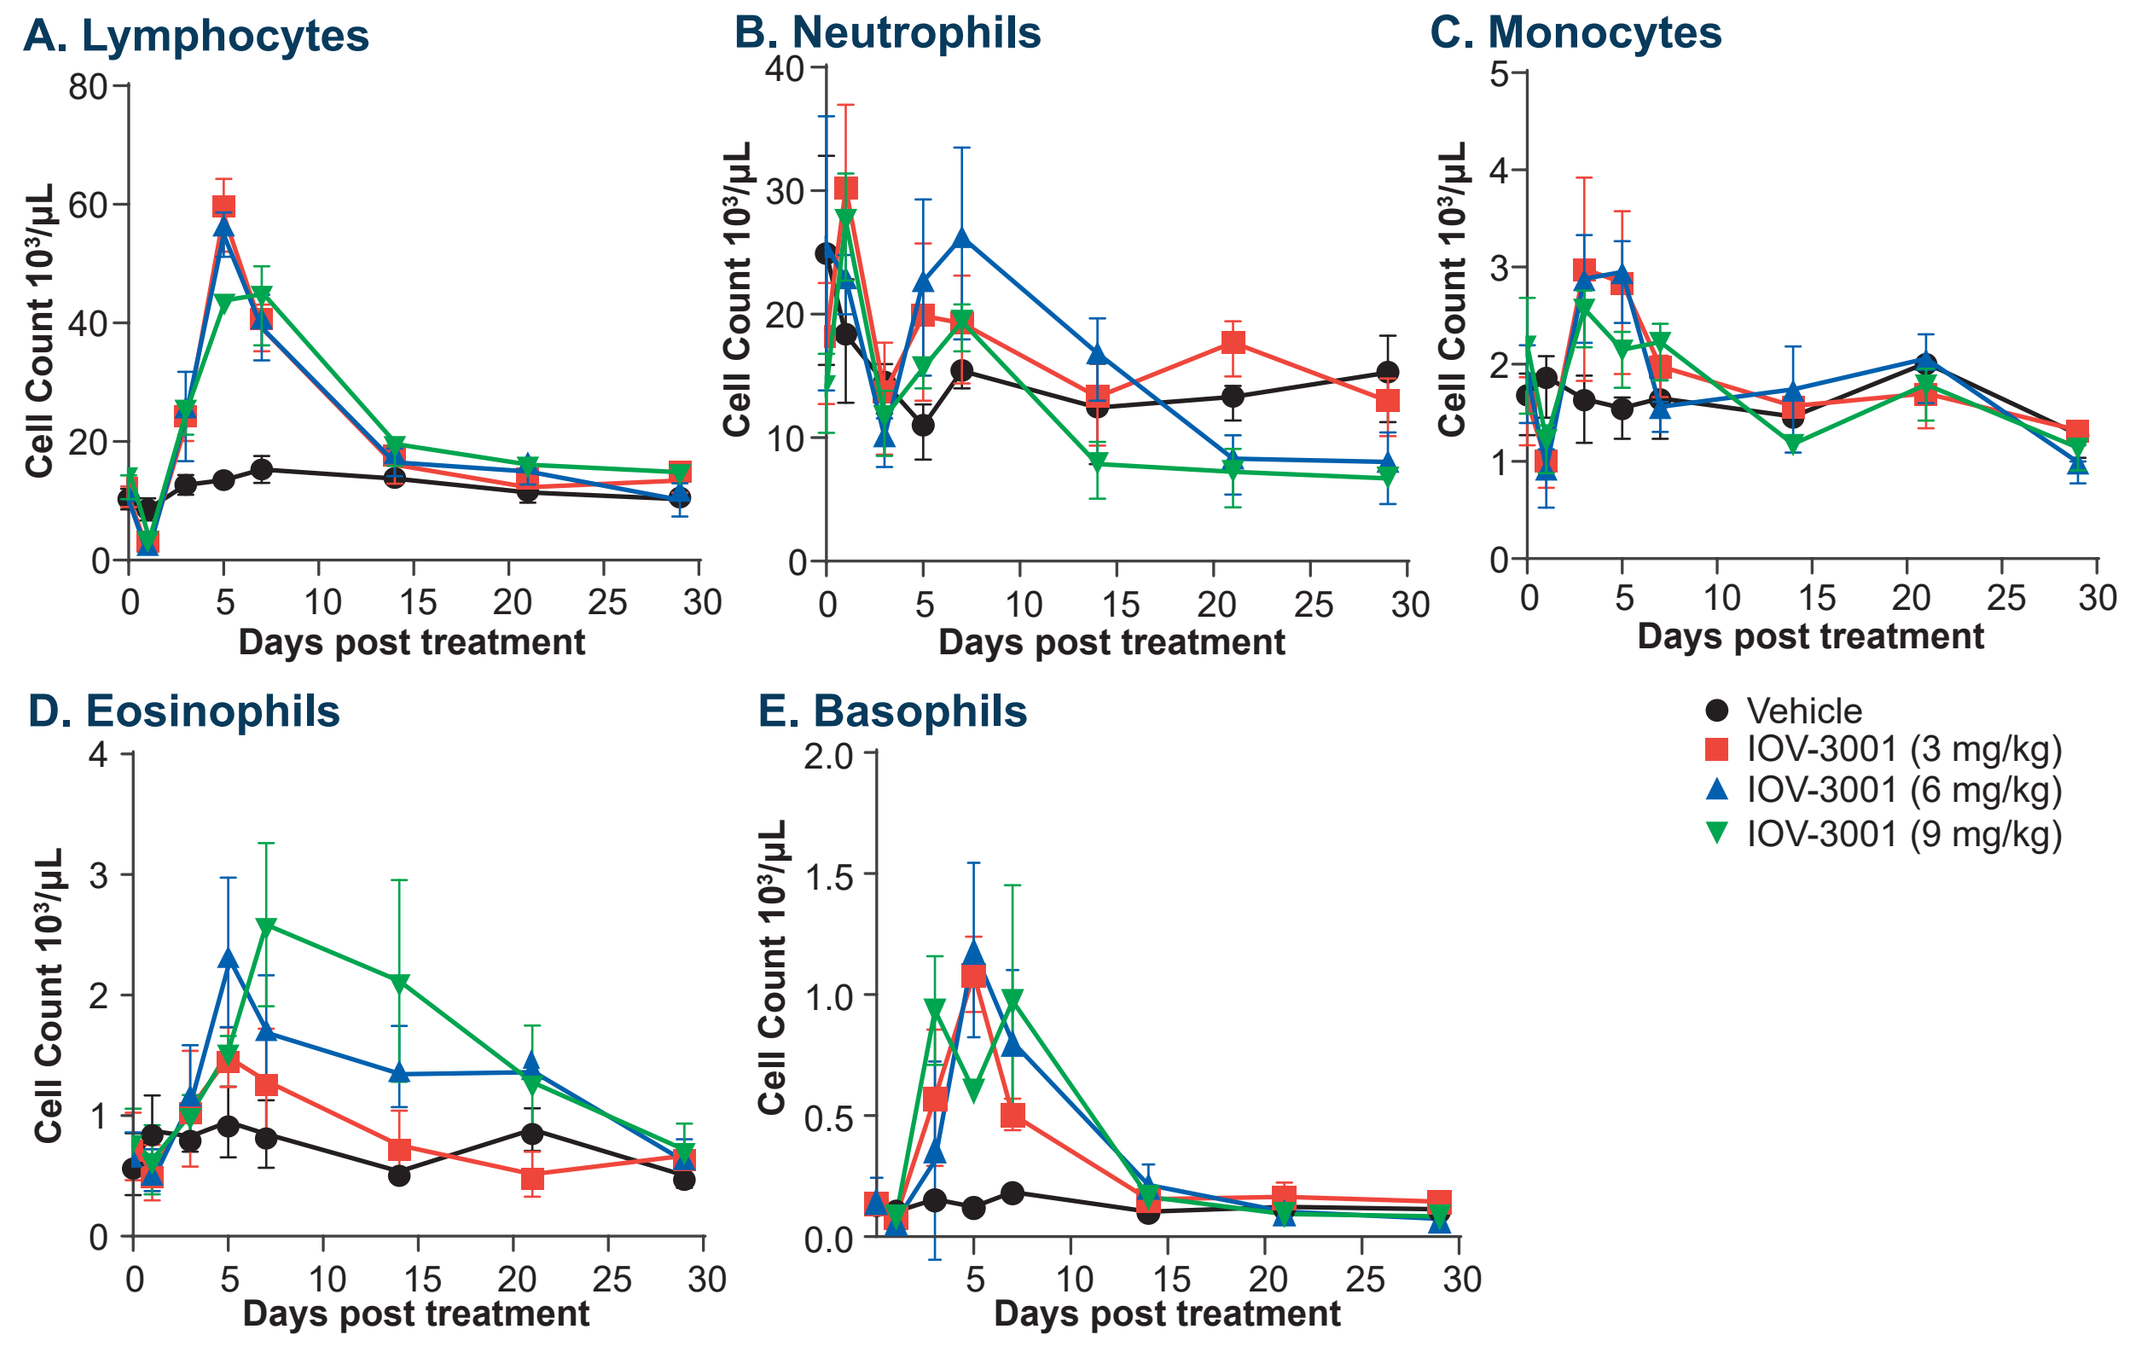
A. Lymphocytes
B. Neutrophils
C. Monocytes
D. Eosinophils
E. Basophils
Cell Count 10³/µL
Days post treatment
Vehicle
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)

Blood samples for cytokine analysis were collected prior to IOV-3001 infusion, on Day 1 (4 and 24 hours) and Days 3, 5, 7, 14, 21 and 29, and analyzed using automated technology (ADVIA™2120 Hematology System). Symbols represent mean values and vertical lines represent the standard deviation.

Figure 7. Inflammatory Cytokine Levels Increased on Day 1 After IOV-3001 Infusion and Returned to Baseline by Day 7

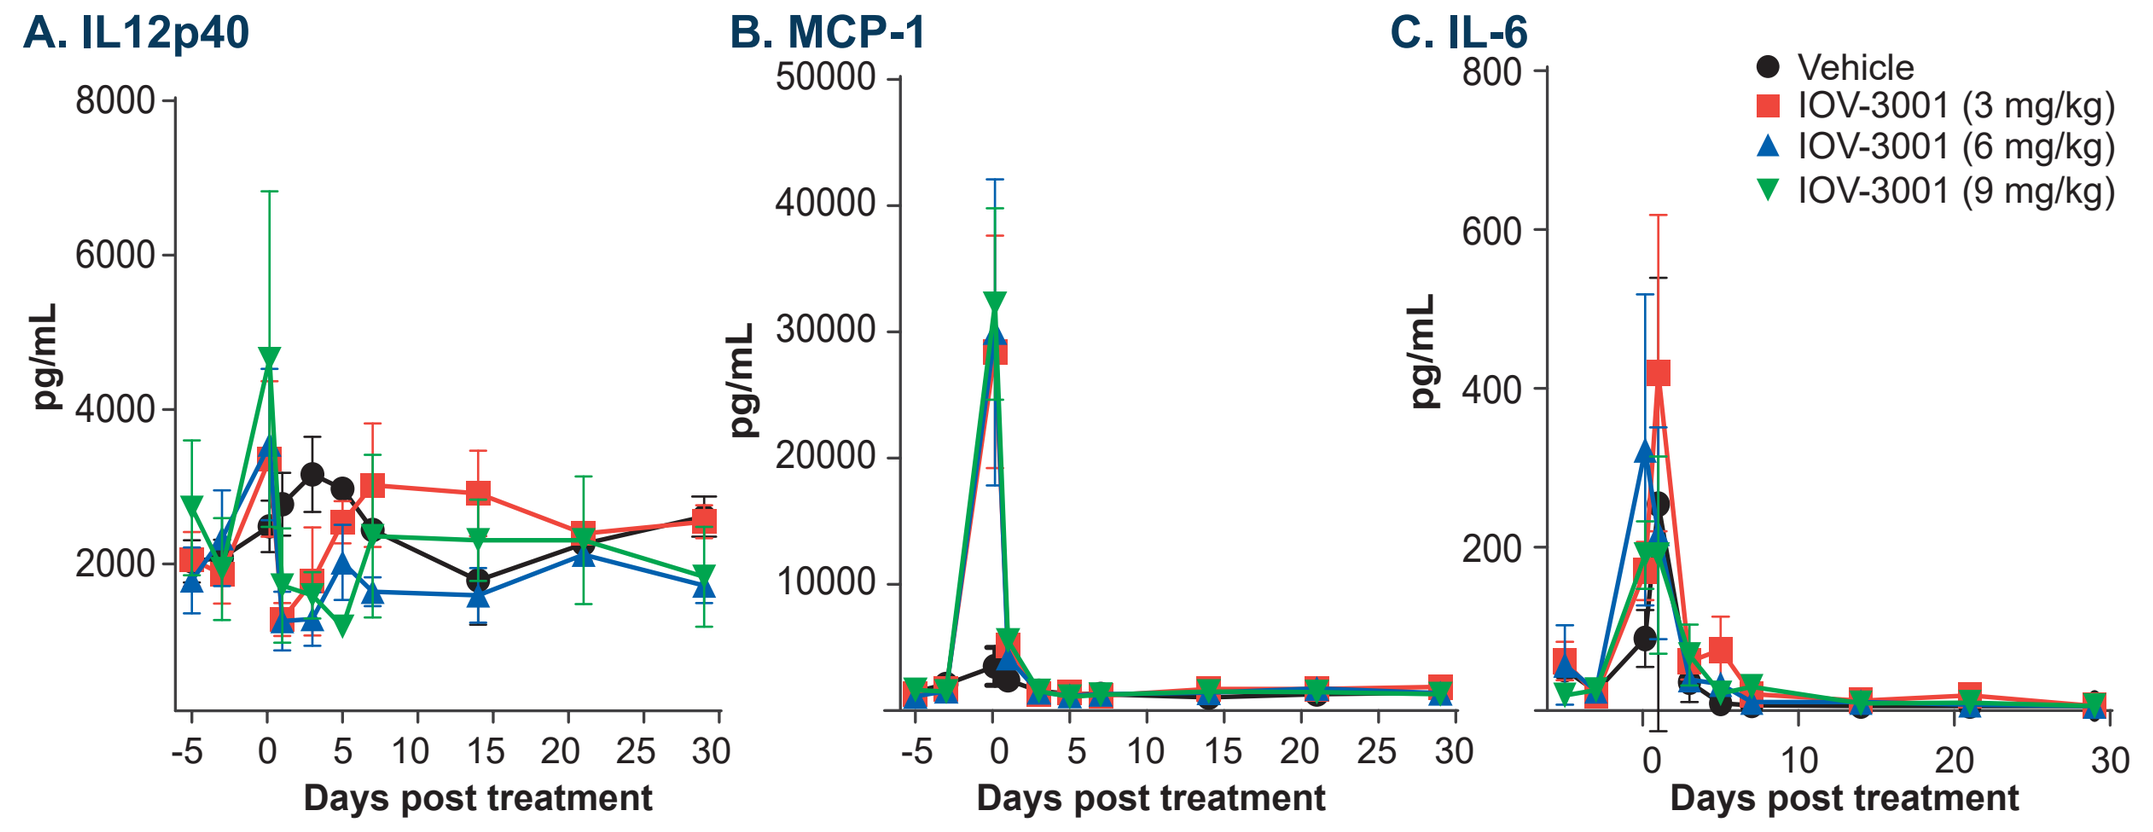
A. IL12p40
B. MCP-1
C. IL-6
pg/mL
Days post treatment
Vehicle
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)

Blood samples were collected prior to IOV-3001 infusion, on Day 1 (4 and 24 hours), and Days 3, 5, 7, 14, 21 and 29 post-infusion. Cytokine levels in serum samples were analyzed at Lovelace Biomedical using a customized Non-Human Primate Cytokine/ Chemokine assay kit (Mesoscale Discovery, Rockville, MD). Symbols represent mean values and vertical lines represent the standard deviation.

- IFN-γ, TNF-α, IL-1β, IL-2, IL-4, IL-8 and IL-10 were not detected in the serum at the assessed time points

Figure 8. Single Dose IOV-3001 Administration Increased Expansion of T Cells and NK Cells With Peak Levels Between Days 3 and 5

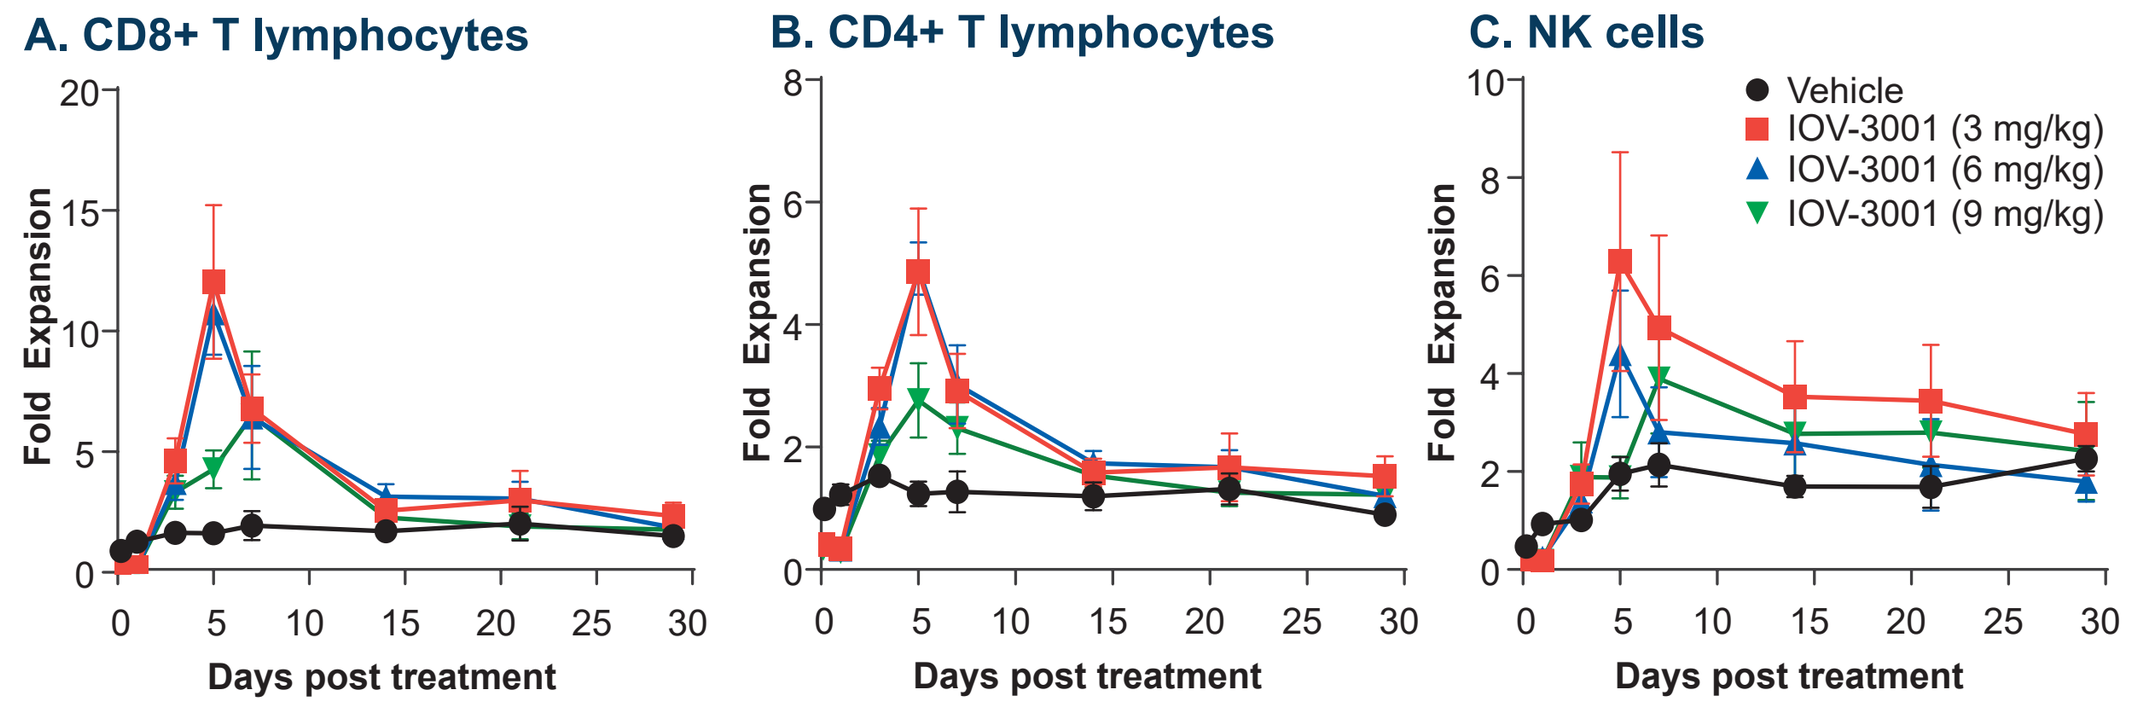
A. CD8+ T lymphocytes
B. CD4+ T lymphocytes
C. NK cells
Fold Expansion
Days post treatment
Vehicle
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)

Following a single dose of IOV-3001 (3, 6, or 9 mg/kg) or vehicle, blood samples from 10 cynomolgus monkeys for each dose group (5 male and 5 female) were assessed on Days 0 to 3. Blood samples from 4 cynomolgus monkeys (2 male and 2 female) in each dose group were assessed on Days 5 through 29. PBMCs were assessed by flow cytometry to identify and quantify T-cell and NK cell lineages at various time points post-infusion. Fold expansion for each specific cell lineage was calculated by dividing the absolute cell count at each post-infusion time point by the absolute cell count for the same cell lineage at preacclimation (average of two preacclimation time points). Figure shows the fold expansion of cytotoxic T cells (ie, CD8+T cells) (A), helper T cells (ie, CD4+ T cells) (B), and NK cells (C). Symbols represent mean values and vertical lines represent the standard error.

Figure 9. Preferential Expansion of CD8+ T Cells Over CD4+ T Regulatory Cells Following a Single Dose of IOV-3001

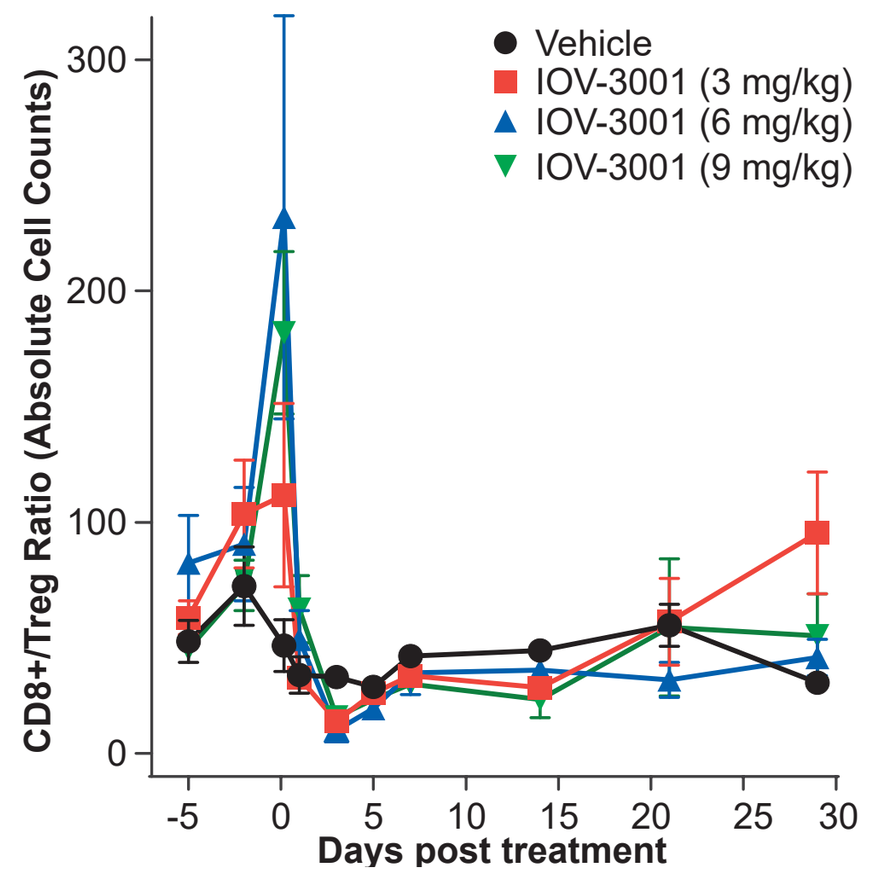
Vehicle
IOV-3001 (3 mg/kg)
IOV-3001 (6 mg/kg)
IOV-3001 (9 mg/kg)
CD8+/Treg Ratio (Absolute Cell Counts)
Days post treatment

Following a single dose of IOV-3001 (3, 6, or 9 mg/kg) or vehicle, blood samples obtained from 10 cynomolgus monkeys (5 male and 5 female) were assessed on Days −5 to 3; blood samples from 4 cynomolgus monkeys (2 male and 2 female) were assessed on Days 5 through 29. Peripheral blood mononuclear cells were assessed by flow cytometry to identify and quantify T-cell lineages. The ratio of CD8+ T cells to CD4+ T regulatory cells was calculated by dividing the absolute cell count of CD8+ T cells by the absolute cell count of CD4+ T regulatory cells. Symbols represent mean values and vertical lines represent the standard error.

Conclusions

- IOV-3001 induced IL-2R–mediated activation of PBMCs and TIL *in vitro*
- IOV-3001 exhibited a prolonged half-life in cynomolgus monkeys
- The pharmacodynamic effects of IOV-3001 reflecting the mechanism of action of IL-2 were demonstrated in B16-D5 mice implanted with pmel-1 T cells and in PBMCs from cynomolgus monkeys
- IOV-3001 demonstrated a favorable preclinical safety profile in cynomolgus monkeys, with an observed NOAEL of 6 mg/kg; PK modeling determined 3.5 mg/kg as the HED NOAEL in humans
- Overall, these data suggest that IOV-3001 may have a better safety profile and may require less frequent dosing in humans compared with aldesleukin
- The features of IOV-3001 demonstrated in this analysis support its investigation as part of TIL cell therapy regimens
- A phase 1 clinical trial is planned to evaluate the safety and efficacy of IOV-3001

Abbreviations

AUC, area under the plasma concentration–time curve; AUC$_{0-\infty}$, area under the plasma concentration–time curve from time 0 to infinity; AUC$_{last}$, area under the plasma concentration–time curve from time 0 to the last measurable concentration; C$_{max}$, maximum plasma concentration; DAPA, D265A and P329A mutations; EC$_{50}$, half maximal effective concentration; GLP, Good Laboratory Practice; GMP, Good Manufacturing Practice; HED, human equivalent dose; HNSTD, highest non-severely toxic dose; IFN- γ, interferon gamma; IL-2, interleukin-2; IL-2R, interleukin-2 receptor; IL-2Rα, interleukin-2 receptor alpha; log, logarithm; M/F, male/female; NHP, non-human primate; NK, natural killer; NOAEL, no observed adverse effect level; PBMCs, peripheral blood mononuclear cells; PD, pharmacodynamic; PK, pharmacokinetic; pSTAT5, phosphorylated signal transducer and activator of transcription 5; STAT5, signal transducer and activator of transcription 5; t$_{1/2}$, half-life; TFN-α, tumor necrosis factor alpha; TIL, tumor-infiltrating lymphocytes.

References

1. Rosenberg SA, et al. *Clin Cancer Res.* 2011;17(13):4550–4557. **2.** Rohaan MW, et al. *N Engl J Med.* 2022;387(23): 2113–2125. **3.** Amtagvi. Prescribing Information. 2024. **4.** Chesney J, et al. *J Immunother Cancer.* 2022;10:e005755. **5.** Wu R, et al. *Cancer J.* 2012;18(2):160–175. **6.** Rosenberg SA. *J Immunol.* 2014;192(12):5451–5458.

Acknowledgments

This study was sponsored by Iovance Biotherapeutics, Inc. (San Carlos, CA). Additional support for this study was provided by Jeffrey Chou and Friedrich Graf Finckenstein of Iovance Biotherapeutics. Editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Iovance.